P4080

Treatment Patterns and Outcomes Among Patients with Relapsed/Refractory Follicular Lymphoma Treated in Routine Clinical Practice in the United States with Three or More Lines of Therapy

Jamie T. Ta,[1] Stella Arndorfer,[2] Cristina Julian,[1] Mei Wu,[1] Dominic Lai,[1] Sheila Shapouri[1]

Summary

This retrospective, observational cohort study used data from a de-identified electronic health record (EHR)-derived database to examine clinical characteristics, treatment patterns, and outcomes in patients who received third- and later-line (3L+) therapy for follicular lymphoma (FL) in the United States (US)

Patients aged ≥18 years, with an initial FL diagnosis between January 2011 and May 2021, who had received ≥3 lines of therapy (LOTs) for FL were included

164 patients met the eligibility criteria

The most common 3L FL treatment regimens were rituximab (R) monotherapy (n=33 [20.1%]), R plus bendamustine (n=22 [13.4%]), R plus lenalidomide (n=22 [13.4%]), obinutuzumab (G) monotherapy (n=15 [9.1%]), idelalisib (n=13 [7.9%]), and G plus bendamustine (n=12 [7.3%])

Median time to next anti-lymphoma treatment or death (TTNTD) after 3L FL treatment was 14.1 months (95% confidence interval [CI]: 10.6–23.3)

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASH® and the author of this poster.
Download this presentation: https://bit.ly/3EAvqok

INTERACTIVE

[1]Genentech, Inc., South San Francisco, CA, USA; [2]Genesis Research, Hoboken, NJ, USA.

Background

- FL is an indolent, yet incurable disease, and patients often require several LOTs throughout their lifetime.[1]
- There are limited real-world data available regarding the treatment of a contemporary cohort of patients with relapsed/refractory (R/R) FL.
- The objective of this study was to assess real-world treatment patterns and outcomes among patients receiving 3L+ therapies for FL in the US.

Patients with an initial FL diagnosis between January 2011 and May 2021, who had received ≥3 LOTs for FL, were included in this retrospective cohort study (Figure 1)

- This study used the nationwide Flatiron Health EHR-derived de-identified database.
- The Flatiron Health database is a longitudinal database, comprising de-identified patient-level structured and unstructured data, curated via technology-enabled abstraction.[2,3]
- During the study period, de-identified data originated from approximately 280 cancer clinics (~800 sites of care) in the US.
- Patient demographic and clinical characteristics were assessed using descriptive statistics.
- The endpoints of this study were treatment patterns by LOT and TTNTD after 3L FL LOT.
- Median TTNTD was estimated using Kaplan–Meier methods.

Figure 1: Study design

Inclusion criteria
- Patients with ≥2 clinic encounters on different days in the Flatiron Health database on or after January 1, 2011
- Patients aged ≥18 years, with an initial FL diagnosis between January 2011 and May 2021, who had received ≥3 LOTs for FL (follow-up ended July 2021)

Exclusion criteria
- Patients with evidence of clinical trial participation anytime during the study
- Patients with Grade 3b FL at diagnosis
- Patients with evidence of transformation to an aggressive lymphoma anytime on or prior to 3L FL treatment
- Patients who received SCT or other anticancer therapies within the 12 months prior to first FL LOT

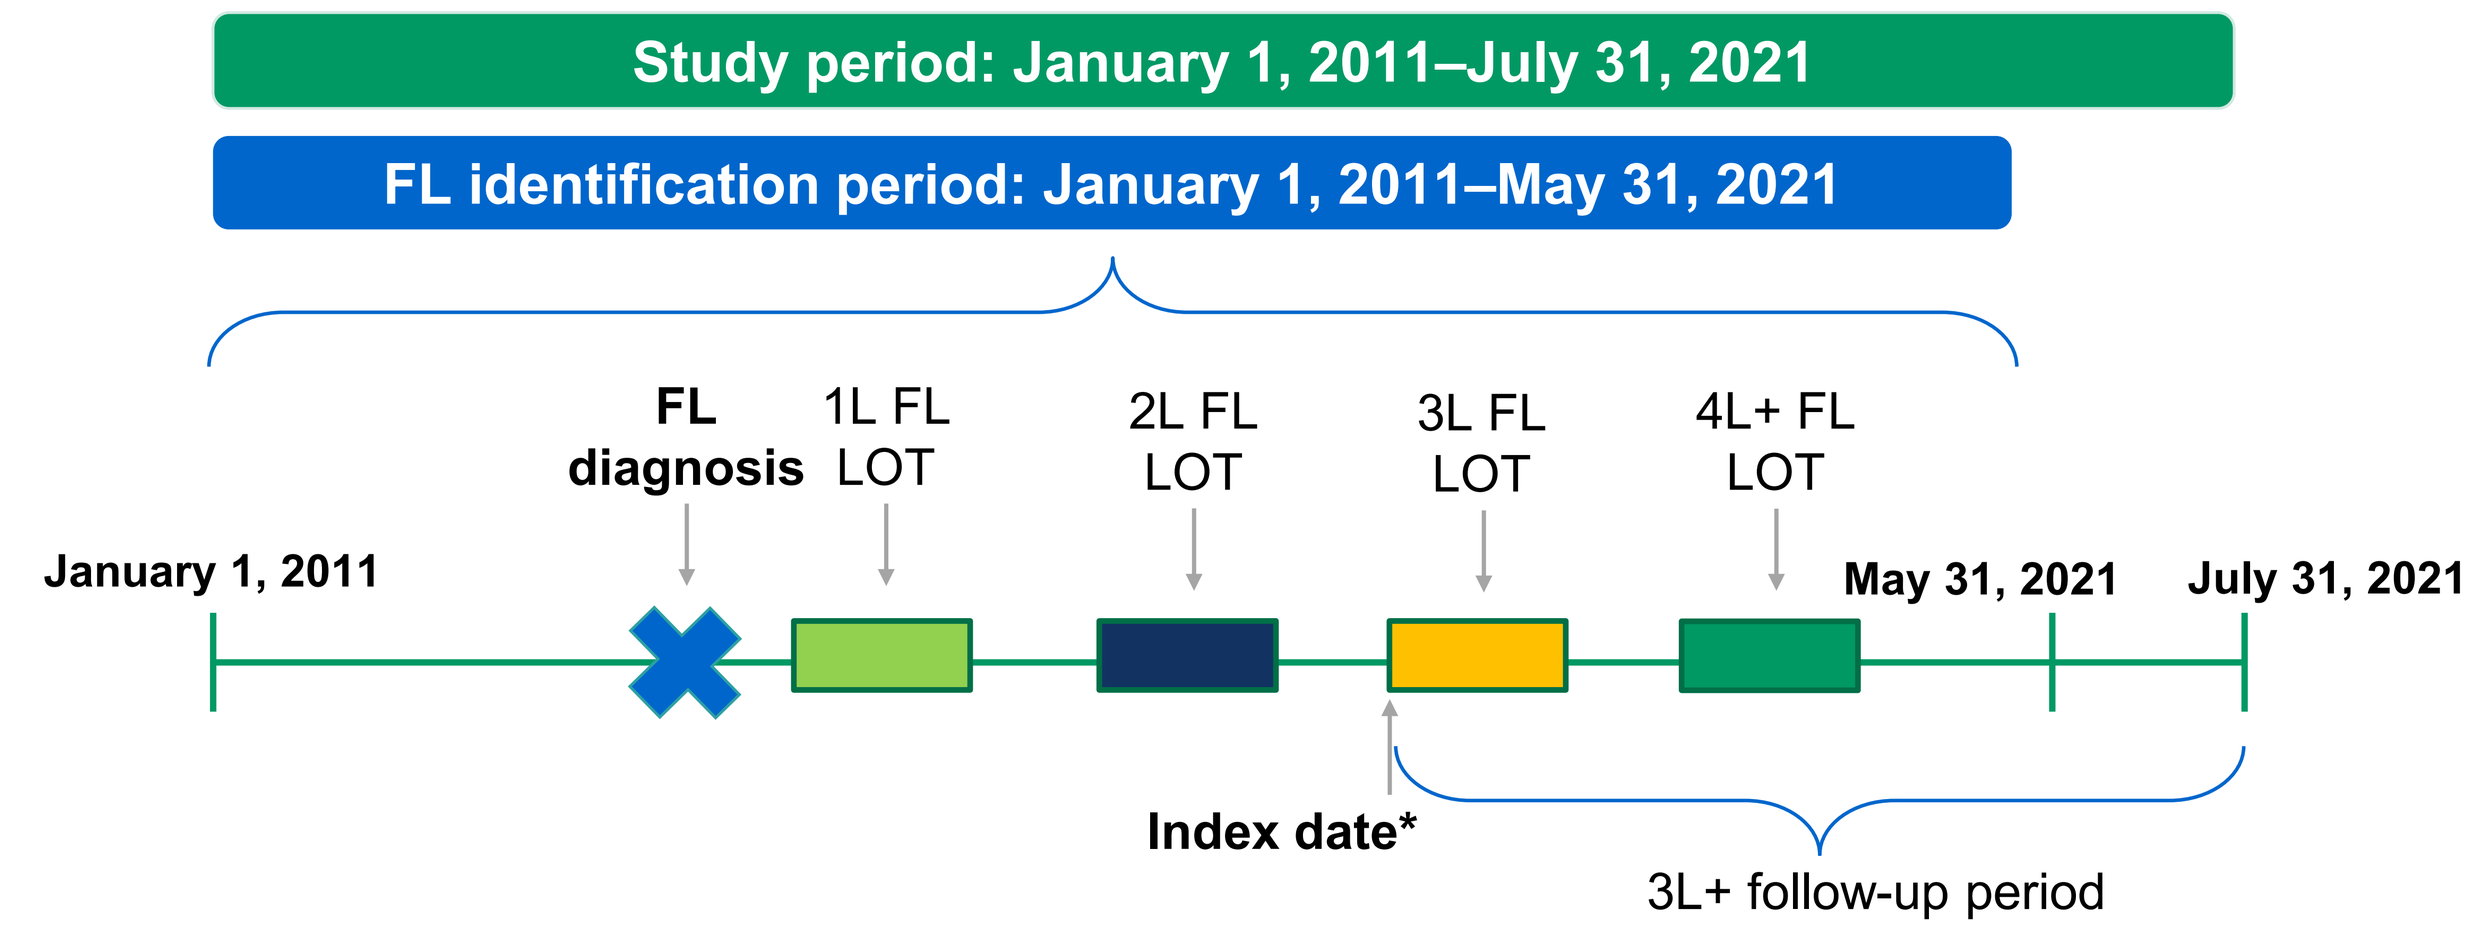

Study period: January 1, 2011–July 31, 2021
FL identification period: January 1, 2011–May 31, 2021
January 1, 2011
FL diagnosis
1L FL LOT
2L FL LOT
3L FL LOT
4L+ FL LOT
May 31, 2021
July 31, 2021
Index date*
3L+ follow-up period

*The index date was the 3L treatment initiation date.
1L, first line; 2L, second line; 4L+, fourth and later lines; SCT, stem cell transplantation.

Of 3,024 patients receiving treatment for FL during the study period, 164 patients who received ≥3 LOTs for FL were included (Figure 2)

Figure 2: Cohort attrition

Patients in Flatiron Health FL database with ≥2 clinical encounters between January 2011–July 2021 (N=5,638)

↓

Patients ≥18 years with an initial FL diagnosis between January 2011–May 2021 and recorded treatment in the Flatiron Health FL database (N=3,827)

↓

Patients receiving treatment for FL and no evidence of clinical trial participation at any time during the study (N=3,551)

↓

Patients without evidence of SCT or anticancer therapies 12 months prior to first FL LOT (N=3,515)

↓

Patients without evidence of transformation to aggressive lymphoma (DLBCL or Burkitt's lymphoma) on or prior to first FL LOT (N=3,127)

↓

FL-treated population: patients without Grade 3b FL at diagnosis who initiated:
≥1L treatment for FL (N=3,024)*
≥2L treatment for FL (N=655)*
≥3L+ treatment for FL (N=164)*

DLBCL, diffuse large B-cell lymphoma. *Patients with evidence of transformation on or prior to each FL LOT were excluded.

Median age at the time of 3L FL treatment was 70 years, and 78.0% of patients had stage III–IV disease at diagnosis (Table 1)

- The majority of patients were treated at community oncology practices (n=146 [89.0%]).
- Median time to 3L FL treatment from diagnosis was 36.7 months, and median follow-up after 3L FL treatment was 14.5 months.
- Overall, 64.6% of patients with evaluable Follicular Lymphoma International Prognostic Index (FLIPI) scores had high-risk FLIPI at diagnosis, and 65.9% of patients had evidence suggestive of progression within 24 months of 1L FL treatment.

Table 1: Demographic and clinical characteristics

Characteristic	3L+ FL population (N=164)
Male, n (%)	83 (50.6)
Median age at 3L FL treatment, years (Q1, Q3)	70 (60, 78)
Age at 3L FL treatment, n (%)	
≤60 years	43 (26.2)
>60 years	121 (73.8)
Year of initial diagnosis, n (%)	
2011–2015	109 (66.5)
2016–2021	55 (33.5)
Race, n (%)	
White	116 (70.7)
Black or African American	8 (<5)
Asian	5 (<5)
Other race	23 (14.0)
Unknown/not documented	12 (7.3)
Practice type, n (%)	
Academic	18 (11.0)
Community	146 (89.0)
Geographic location, n (%)	
South	67 (40.9)
West	27 (16.5)
Northeast	27 (16.5)
Midwest	16 (9.8)
Puerto Rico or unknown/not documented	27 (16.5)
ECOG PS at diagnosis, n (%)	
0-2	46 (28.0)
Unknown/not documented	118 (72.0)
Grade at diagnosis, n (%)	
1–2	130 (79.3)
3a	24 (14.6)
Unknown/not documented	10 (6.1)
Ann Arbor stage at diagnosis, n (%)	
Stage I/II	36 (22.0)
Stage III/IV	128 (78.0)
FLIPI score at diagnosis, n (%)	
FLIPI score available	79 (48.2)
High risk (≥3)	51 (64.6)
Intermediate risk (2)	21 (26.6)
Low risk (≤1)	7 (8.9)
Missing	85 (51.8)
Presence of B symptoms at diagnosis, n (%)	
No	106 (64.6)
Yes	39 (23.8)
Unknown/not documented	19 (11.6)
POD24*, n (%)	108 (65.9)
Transformation, n (%)	17 (10.4)
Median time from diagnosis to 3L FL treatment initiation, months (Q1, Q3)	36.7 (24.0, 59.0)
Median follow-up time after 3L FL treatment, months (Q1, Q3)	14.5 (5.0, 31.7)
Total lines of FL therapy received, n (%)	
3	110 (67.1)
4	35 (21.3)
5+	19 (11.6)

*POD24 was defined as initiation of 2L anti-lymphoma treatment or evidence of transformation to an aggressive lymphoma, within 24 months after the first FL LOT. ECOG PS, Eastern Cooperative Oncology Group performance status; POD24, progression of disease within 24 months; Q, quartile.

The most common 3L FL treatment regimens were R monotherapy, R plus bendamustine, R plus lenalidomide, G monotherapy, idelalisib, and G plus bendamustine (Table 2)

- Overall, 54 patients (32.9%) received a subsequent 4L FL therapy.

Table 2: Most common FL treatment regimens by LOT

	3L FL treatment regimen	3L (N=164), n (%)
1st	R monotherapy	33 (20.1)
2nd	R bendamustine	22 (13.4)
2nd	R lenalidomide	22 (13.4)
3rd	G monotherapy	15 (9.1)
4th	Idelalisib	13 (7.9)
5th	G bendamustine	12 (7.3)
	4L FL treatment regimen	**4L (N=54), n (%)**
1st	R lenalidomide	11 (20.4)
2nd	R monotherapy	9 (16.7)
2nd	R bendamustine	9 (16.7)
3rd	G monotherapy	6 (11.1)
4th	R-CVP	*
5th	R-CHOP	*
5th	Idelalisib	*
5th	Copanlisib	*
5th	G bendamustine	*

*Masked due to small patient numbers (n <5). CHOP, cyclophosphamide, doxorubicin, vincristine, and prednisone; CVP, cyclophosphamide, vincristine, and prednisone.

Significant heterogeneity in FL treatment sequences among patients receiving 3L+ FL therapy was observed in real-world clinical practice (Figure 3)

Figure 3: FL treatment sequencing pre- and post-3L therapy up to 5L (3L+ FL; N=164)

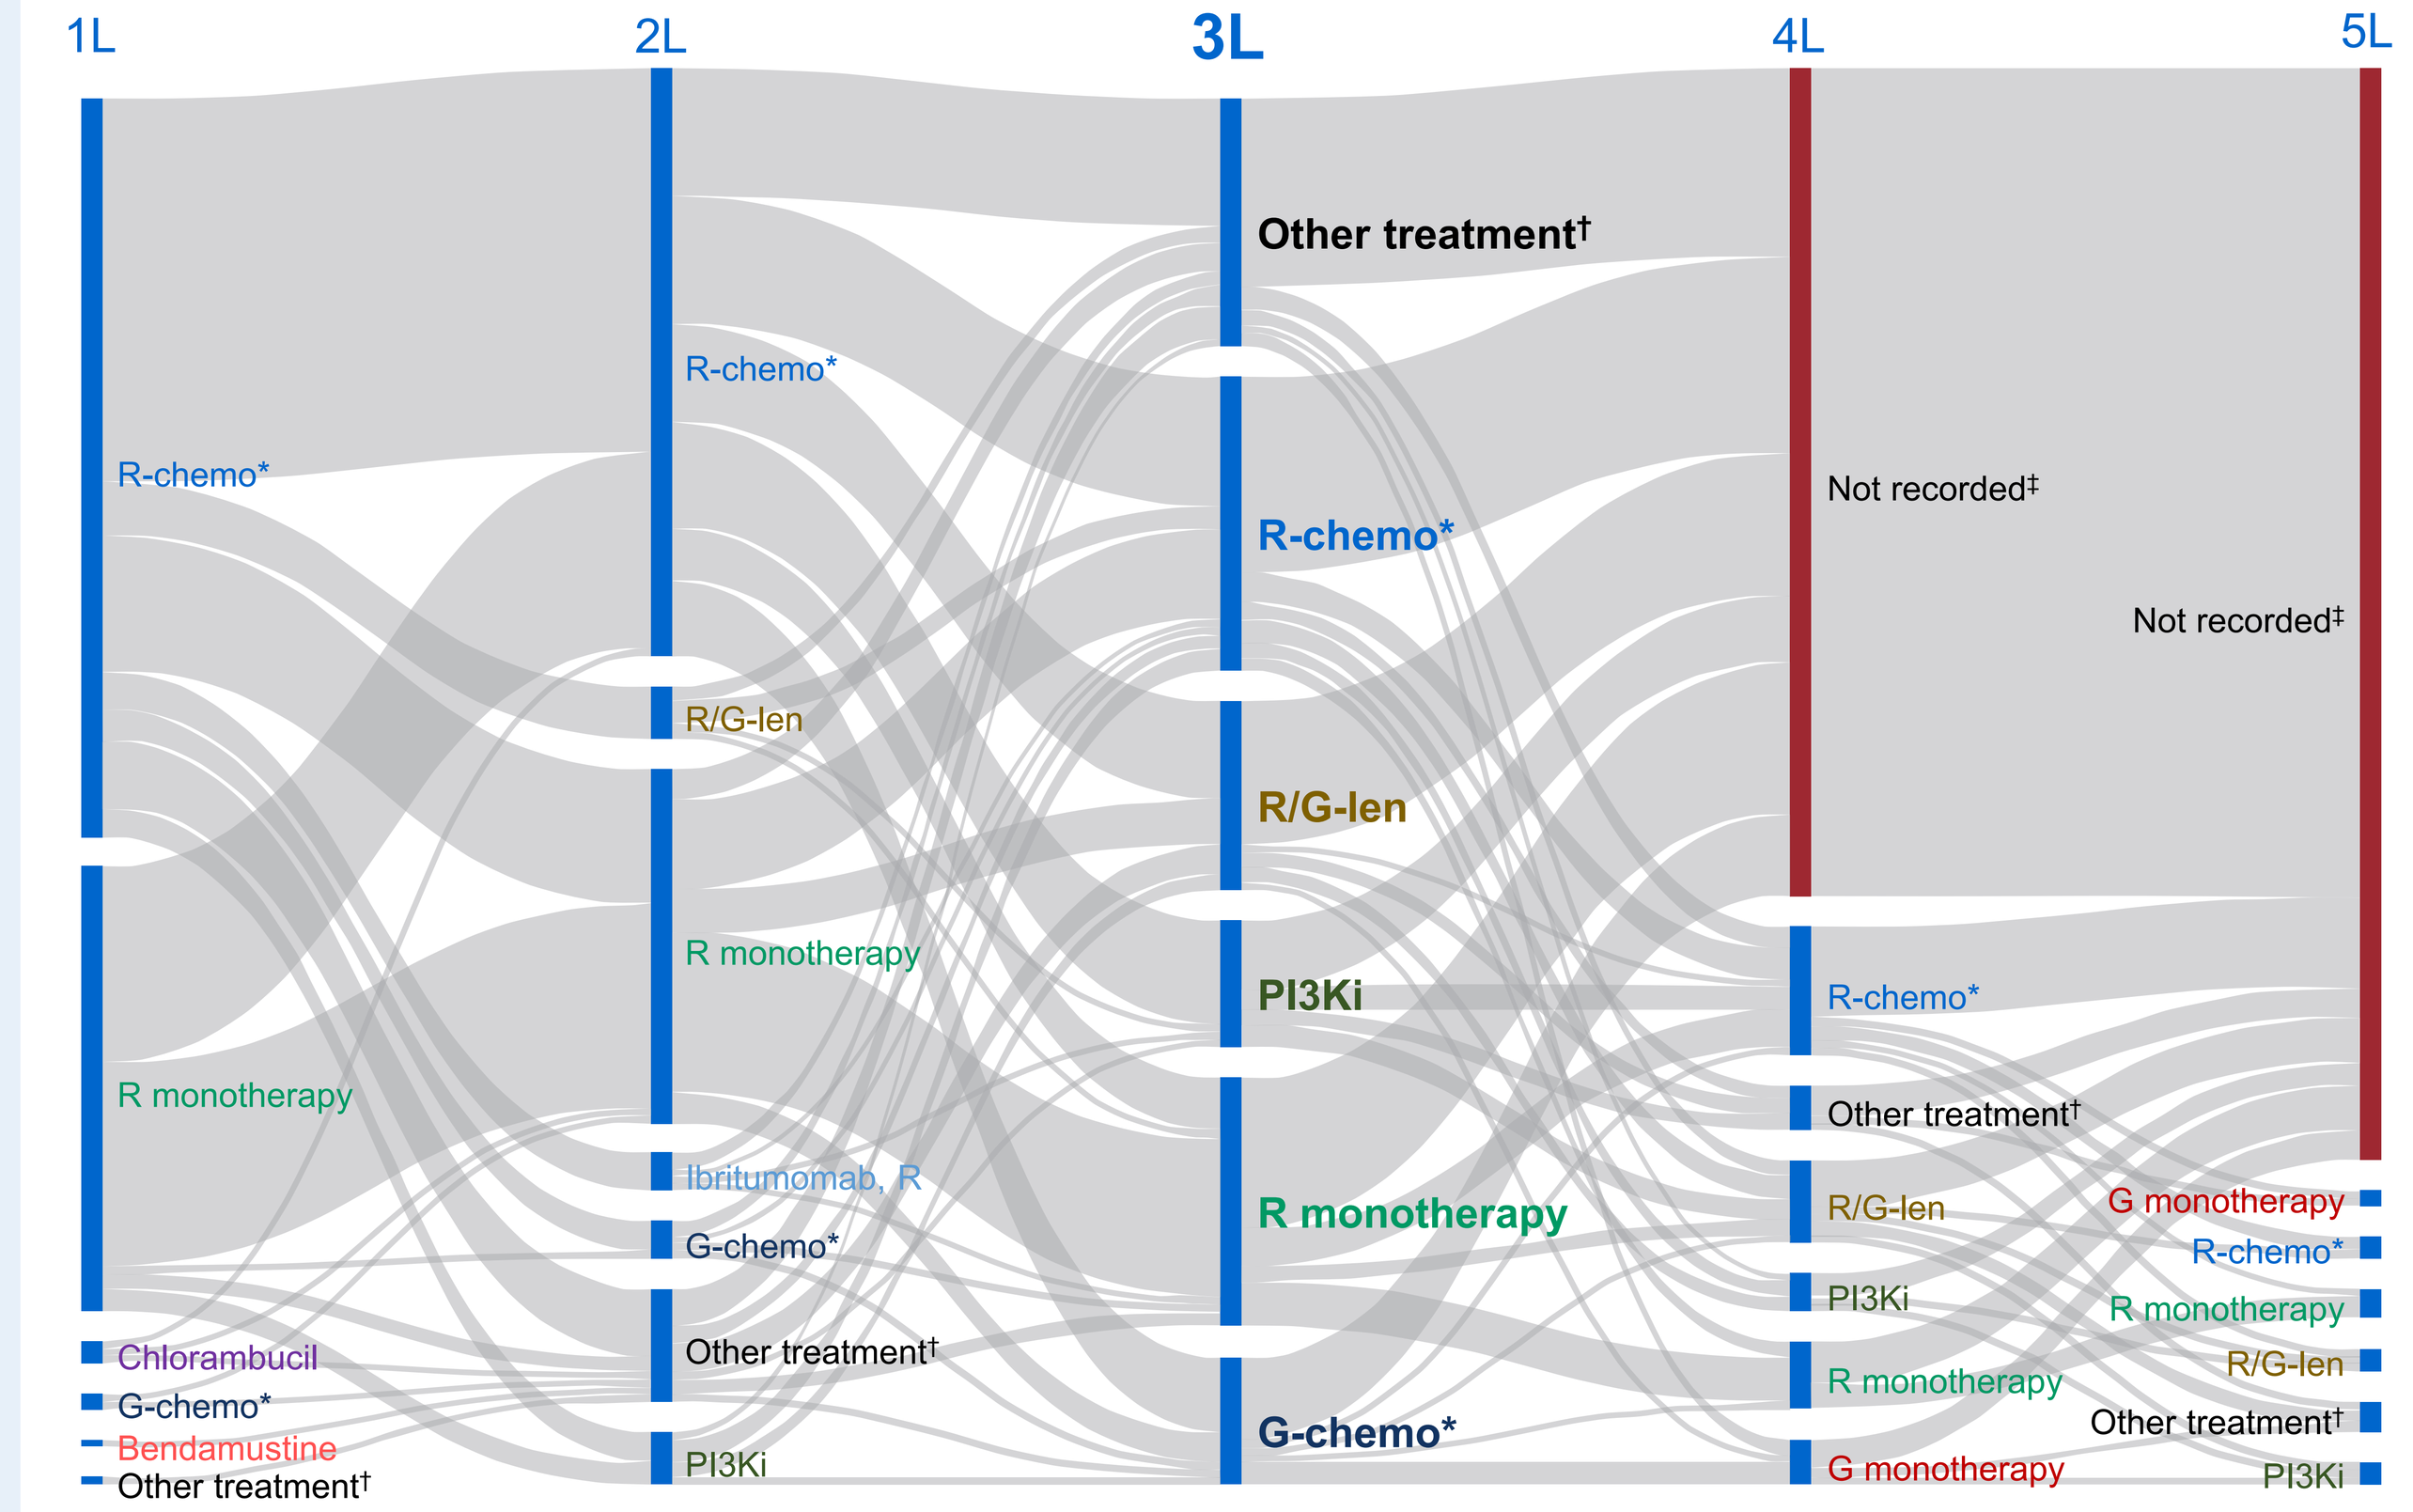

1L
2L
3L
4L
5L
R-chemo*
R monotherapy
Chlorambucil
G-chemo*
Bendamustine
Other treatment†
R-chemo*
R/G-len
R monotherapy
Ibritumomab, R
G-chemo*
Other treatment†
PI3Ki
Other treatment†
R-chemo*
R/G-len
PI3Ki
R monotherapy
G-chemo*
Not recorded‡
R-chemo*
Other treatment†
R/G-len
PI3Ki
R monotherapy
G monotherapy
Not recorded‡
G monotherapy
R-chemo*
R monotherapy
R/G-len
Other treatment†
PI3Ki

*R-chemo/G-chemo includes R/G in combination with a chemo regimen (e.g. bendamustine, CHOP, CVP). †Other regimens included FL treatments that fell outside of the top five most common FL therapy classes in each line. ‡No recorded treatment indicates patients did not have documented subsequent FL treatments prior to last activity date in EHR or death. 5L, fifth line; chemo, chemotherapy; len, lenalidomide; PI3Ki, phosphoinositide 3-kinase inhibitor.

Median TTNTD after 3L FL treatment was 14.1 months (95% CI: 10.6–23.3; Figure 4)

Figure 4: Kaplan-Meier curve for TTNTD* (N=164) following 3L FL treatment†

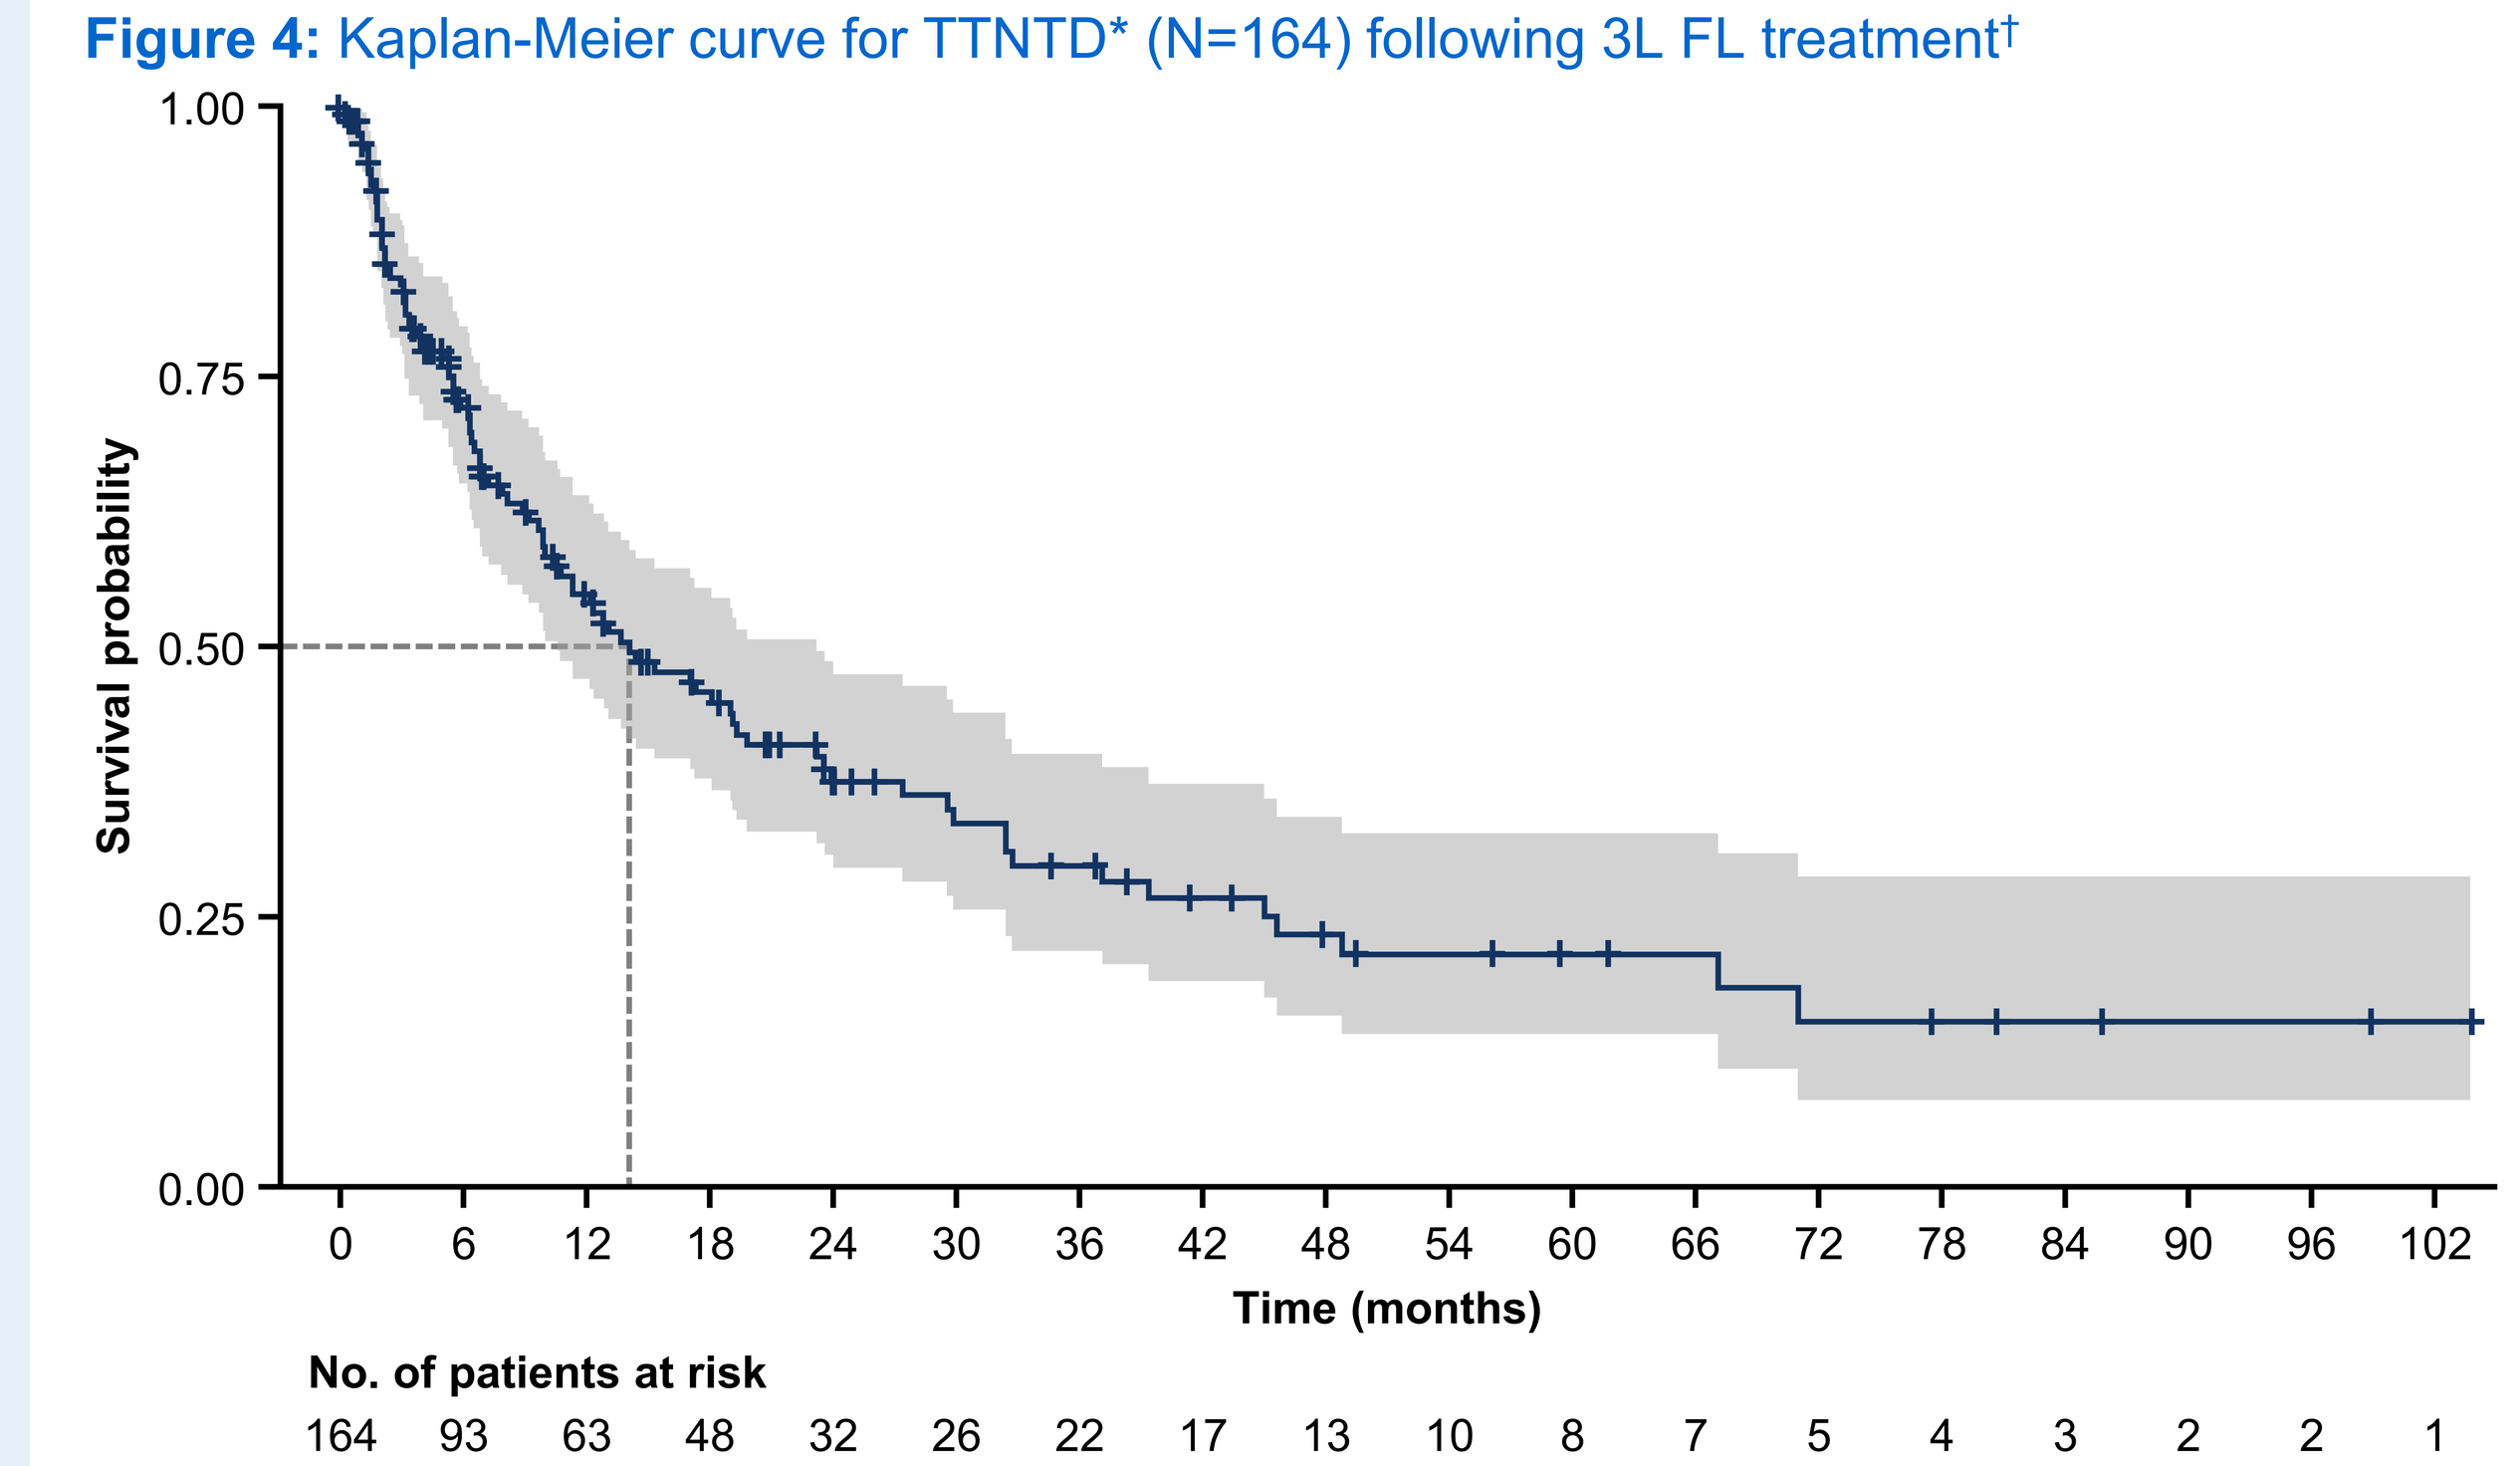

Survival probability
1.00
0.75
0.50
0.25
0.00
0 6 12 18 24 30 36 42 48 54 60 66 72 78 84 90 96 102
Time (months)
No. of patients at risk
164 93 63 48 32 26 22 17 13 10 8 7 5 4 3 2 2 1

*Time from initiation of 3L FL treatment to next anti-lymphoma treatment or death; patients censored at last activity in EHR.
†Median follow-up after 3L FL treatment was 14.5 months

Conclusions

- Our analysis provides an update on the heterogeneous treatment landscape for R/R FL in the US for patients receiving 3L+ FL therapy in real-world clinical practice, the majority of whom were treated in community oncology practices.
- Many patients requiring 3L+ FL treatment had clinical characteristics predictive of poor prognosis, based on evidence suggestive of early progression and a high-risk FLIPI score at diagnosis.
- Treatment outcomes following 3L FL therapy remain poor, and approximately one-third of patients required additional treatment beyond 3L.
- Limitations of this analysis include those inherent to real-world observational databases and the relatively short follow-up of patients with indolent FL in the Flatiron Health database, particularly following 3L FL treatment.
- Future studies with additional follow-up are warranted. Nevertheless, these findings highlight the ongoing unmet need for novel, effective treatments in this setting in order to improve patient outcomes.

Presented at the 63rd ASH Annual Meeting and Exposition | 11–14 December 2021

References
1. Batlevi, et al. Blood Cancer J 2020;10:74
2. Ma, et al. medRxiv 2020.03.16.20037143. Available from: https://www.medrxiv.org/content/10.1101/2020.03.16.20037143v2
3. Birnbaum, et al. arXiv 2021.09765. Available from: https://arxiv.org/abs/2001.09765

Acknowledgements
This study was sponsored by Genentech, Inc. Third party editorial assistance, under the direction of all authors, was provided by Zoe Toland, BSc, of Ashfield MedComms, an Ashfield Health company, and was funded by Genentech, Inc.

Disclosures
Jamie T. Ta is employed by Genentech, Inc. and has equity interests in F. Hoffmann-La Roche; Stella Arndorfer is employed by and has a consultancy role with Genesis Research; Cristina Julian is employed by Genentech, Inc. and has equity interests in F. Hoffmann-La Roche; Mei Wu is employed by Genentech, Inc. and has equity interests in F. Hoffmann-La Roche; Dominic Lai is employed by Genentech, Inc., was previously employed by AbbVie, and has equity interests in BMS, AbbVie, and Genentech, Inc.; Sheila Shapouri is employed by Genentech, Inc. and has equity interests in F. Hoffmann-La Roche.