2949

Health-Related Quality of Life (HRQoL) in Patients with Diffuse Large B-Cell Lymphoma (DLBCL) Treated with Polatuzumab Vedotin, Rituximab, Cyclophosphamide, Doxorubicin and Prednisone (Pola-R-CHP) versus Rituximab, Cyclophosphamide, Doxorubicin, Vincristine and Prednisone (R-CHOP) in the Phase III POLARIX Study

Jonathan W. Friedberg,[1] Carrie Thompson,[2] Marek Trněný,[3] Franck Morschhauser,[4] Gilles Salles,[5] Patrick M. Reagan,[6] Mark Hertzberg,[7] Piotr Smolewski,[8] Huilai Zhang,[9] Catherine Thieblemont,[10] Bei Hu,[11] Gustavo Fonseca,[12] Won Seog Kim,[13] Maurizio Martelli,[14] Amitkumar Mehta,[15] Avrita Campinha-Bacote,[16] Mark Yan,[17] Jamie Hirata,[16] Matthew Sugidono,[16] Calvin Lee,[16] Jeff Sharman[18]

Summary

Patient-reported outcomes were used to characterize the patient experience in POLARIX.

Clinically meaningful improvements in disease-related symptoms were seen in both treatment arms.

These data showed that the **patient experience was similar** between those without progression receiving Pola-R-CHP or R-CHOP.

Improvements in HRQoL measures were rapid and sustained.

[1]Wilmot Cancer Institute, University of Rochester, Rochester, NY, USA; [2]Mayo Clinic College of Medicine, Rochester, MN, USA; [3]First Faculty of Medicine, Charles University, General Hospital, Prague, Czech Republic; [4]University of Lille, CHU Lille, ULR 7365 – GRITA – Group de Recherche sur les formes Injectables et les Technologies Associées, Lille, France; [5]Memorial Sloan Kettering Cancer Center, New York City, NY, USA; [6]University of Rochester School of Medicine, Rochester, NY, USA; [7]Prince of Wales Hospital and University of NSW, Sydney, Australia; [8]Medical University of Lodz, Lodz, Poland; [9]Tianjin Medical University Cancer Hospital, Tianjin, China; [10]University of Paris, Assistance Publique – Hôpitaux de Paris, Saint-Louis Hospital, Hemato-oncology department, Paris, France; [11]Levine Cancer Institute, Atrium Health, Charlotte, NC, USA; [12]Florida Cancer Specialists North/Sarah Cannon Research Institute, St. Petersburg, FL, USA; [13]Sungkyunkwan University School of Medicine, Seoul, Republic of Korea; [14]Hematology Department of Translational and Precision Medicine, Sapienza University, Rome, Italy; [15]University of Alabama at Birmingham, AL, USA; [16]Genentech, Inc., South San Francisco, CA, USA; [17]Hoffmann-La Roche Ltd, Mississauga, ON, Canada; [18]Willamette Valley Cancer Institute/US Oncology, Eugene, OR, USA.

Background

- Patients with lymphoma report impaired HRQoL as a result of disease- and treatment-related symptoms.[1]
- However, there are limited dynamic HRQoL data using patient-reported outcome (PRO) measures from large prospective clinical trials in patients with previously untreated DLBCL.
- The Phase III POLARIX study (NCT03274492) demonstrated superior progression-free survival and a similar safety profile with Pola-R-CHP versus standard R-CHOP in patients with previously untreated DLBCL.[2]
- Analysis of patient-reported peripheral neuropathy symptoms in POLARIX using the Functional Assessment of Cancer Therapy/Gynecologic Oncology Group Neurotoxicity subscale have been reported previously.[3]
- In this analysis, PROs were used to measure QoL, physical functioning, fatigue, and lymphoma-related symptom scores to fully characterize the patient experience in POLARIX.

Methods

- In total, 879 patients with previously untreated DLBCL were randomized 1:1 to receive Pola-R-CHP (n=440) or R-CHOP (n=439).
- Lymphoma symptom scores were assessed using the Functional Assessment of Cancer Therapy-Lymphoma Subscale (FACT-LymS).[1]
- QoL, physical functioning, and fatigue were assessed using the European Organization for Research and Treatment of Cancer Quality of Life-Core 30 questionnaire (EORTC QLQ-C30).[4]
- Questionnaires were administered to patients without progression according to a defined schedule (**Figure 1**) until disease progression or study discontinuation.

Figure 1. Schedule of PRO questionnaire administration.

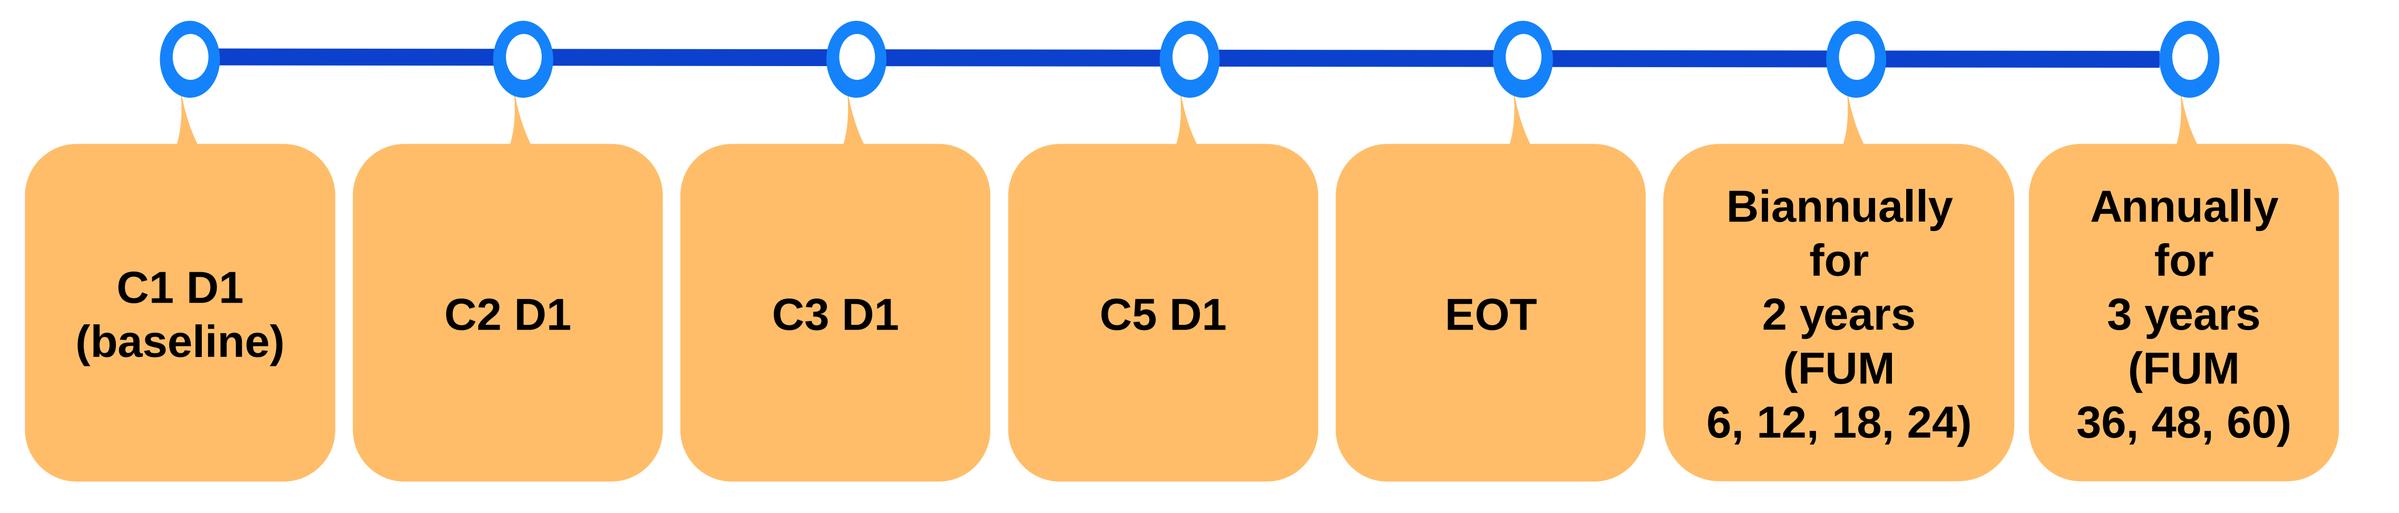
C1 D1
(baseline)
C2 D1
C3 D1
C5 D1
EOT
Biannually for 2 years (FUM 6, 12, 18, 24)
Annually for 3 years (FUM 36, 48, 60)

Note: Patients who experienced disease progression or discontinued treatment stopped reporting PROs.
C, Cycle; D, Day; EOT, end of treatment; FUM, Follow-up Month.

- Percentage of patients with clinically meaningful improvements, worsening, and no changes from baseline for HRQoL measures (lymphoma symptom scores, QoL, physical functioning, and fatigue) were reported based on validated thresholds[1,4] (**Table 1**).
- Changes over time in HRQoL measures were assessed.

Table 1. Thresholds* for clinically meaningful differences in PROs.

PRO Measures	Clinically Meaningful Improvement	No Change	Clinically Meaningful Worsening
Lymphoma Symptom Score[1]	≥3-point increase	-2-points to +2-points	≥3-point decrease
QoL[4]	≥8-point increase	-9-points to +7-points	≥10-point decrease
Physical Functioning[4]	≥7-point increase	-6-points to +9-points	≥10-point decrease
Fatigue[4]	≥9-point increase	-9-points to +8-points	≥10-point decrease

*Thresholds validated based on the FACT-LymS index and EORTC QLQ-C30 tool.[1,4]

Results

- Questionnaire response rates for all PROs were high (>80%) throughout the study.

Lymphoma symptom scores

- Lymphoma symptom scores were similar in both treatment arms at baseline.
- Clinically meaningful improvements at any time point were seen in 82.3% of patients receiving Pola-R-CHP and 81.3% of those receiving R-CHOP
 - Both regimens caused rapid, sustained improvements in lymphoma symptom score in most patients after Cycle 1, which plateaued by the first follow-up assessment after EOT (**Figure 2**).

Figure 2. FACT-LymS lymphoma symptom score by (A) clinically meaningful difference* and (B) mean changes over time.†, ‡

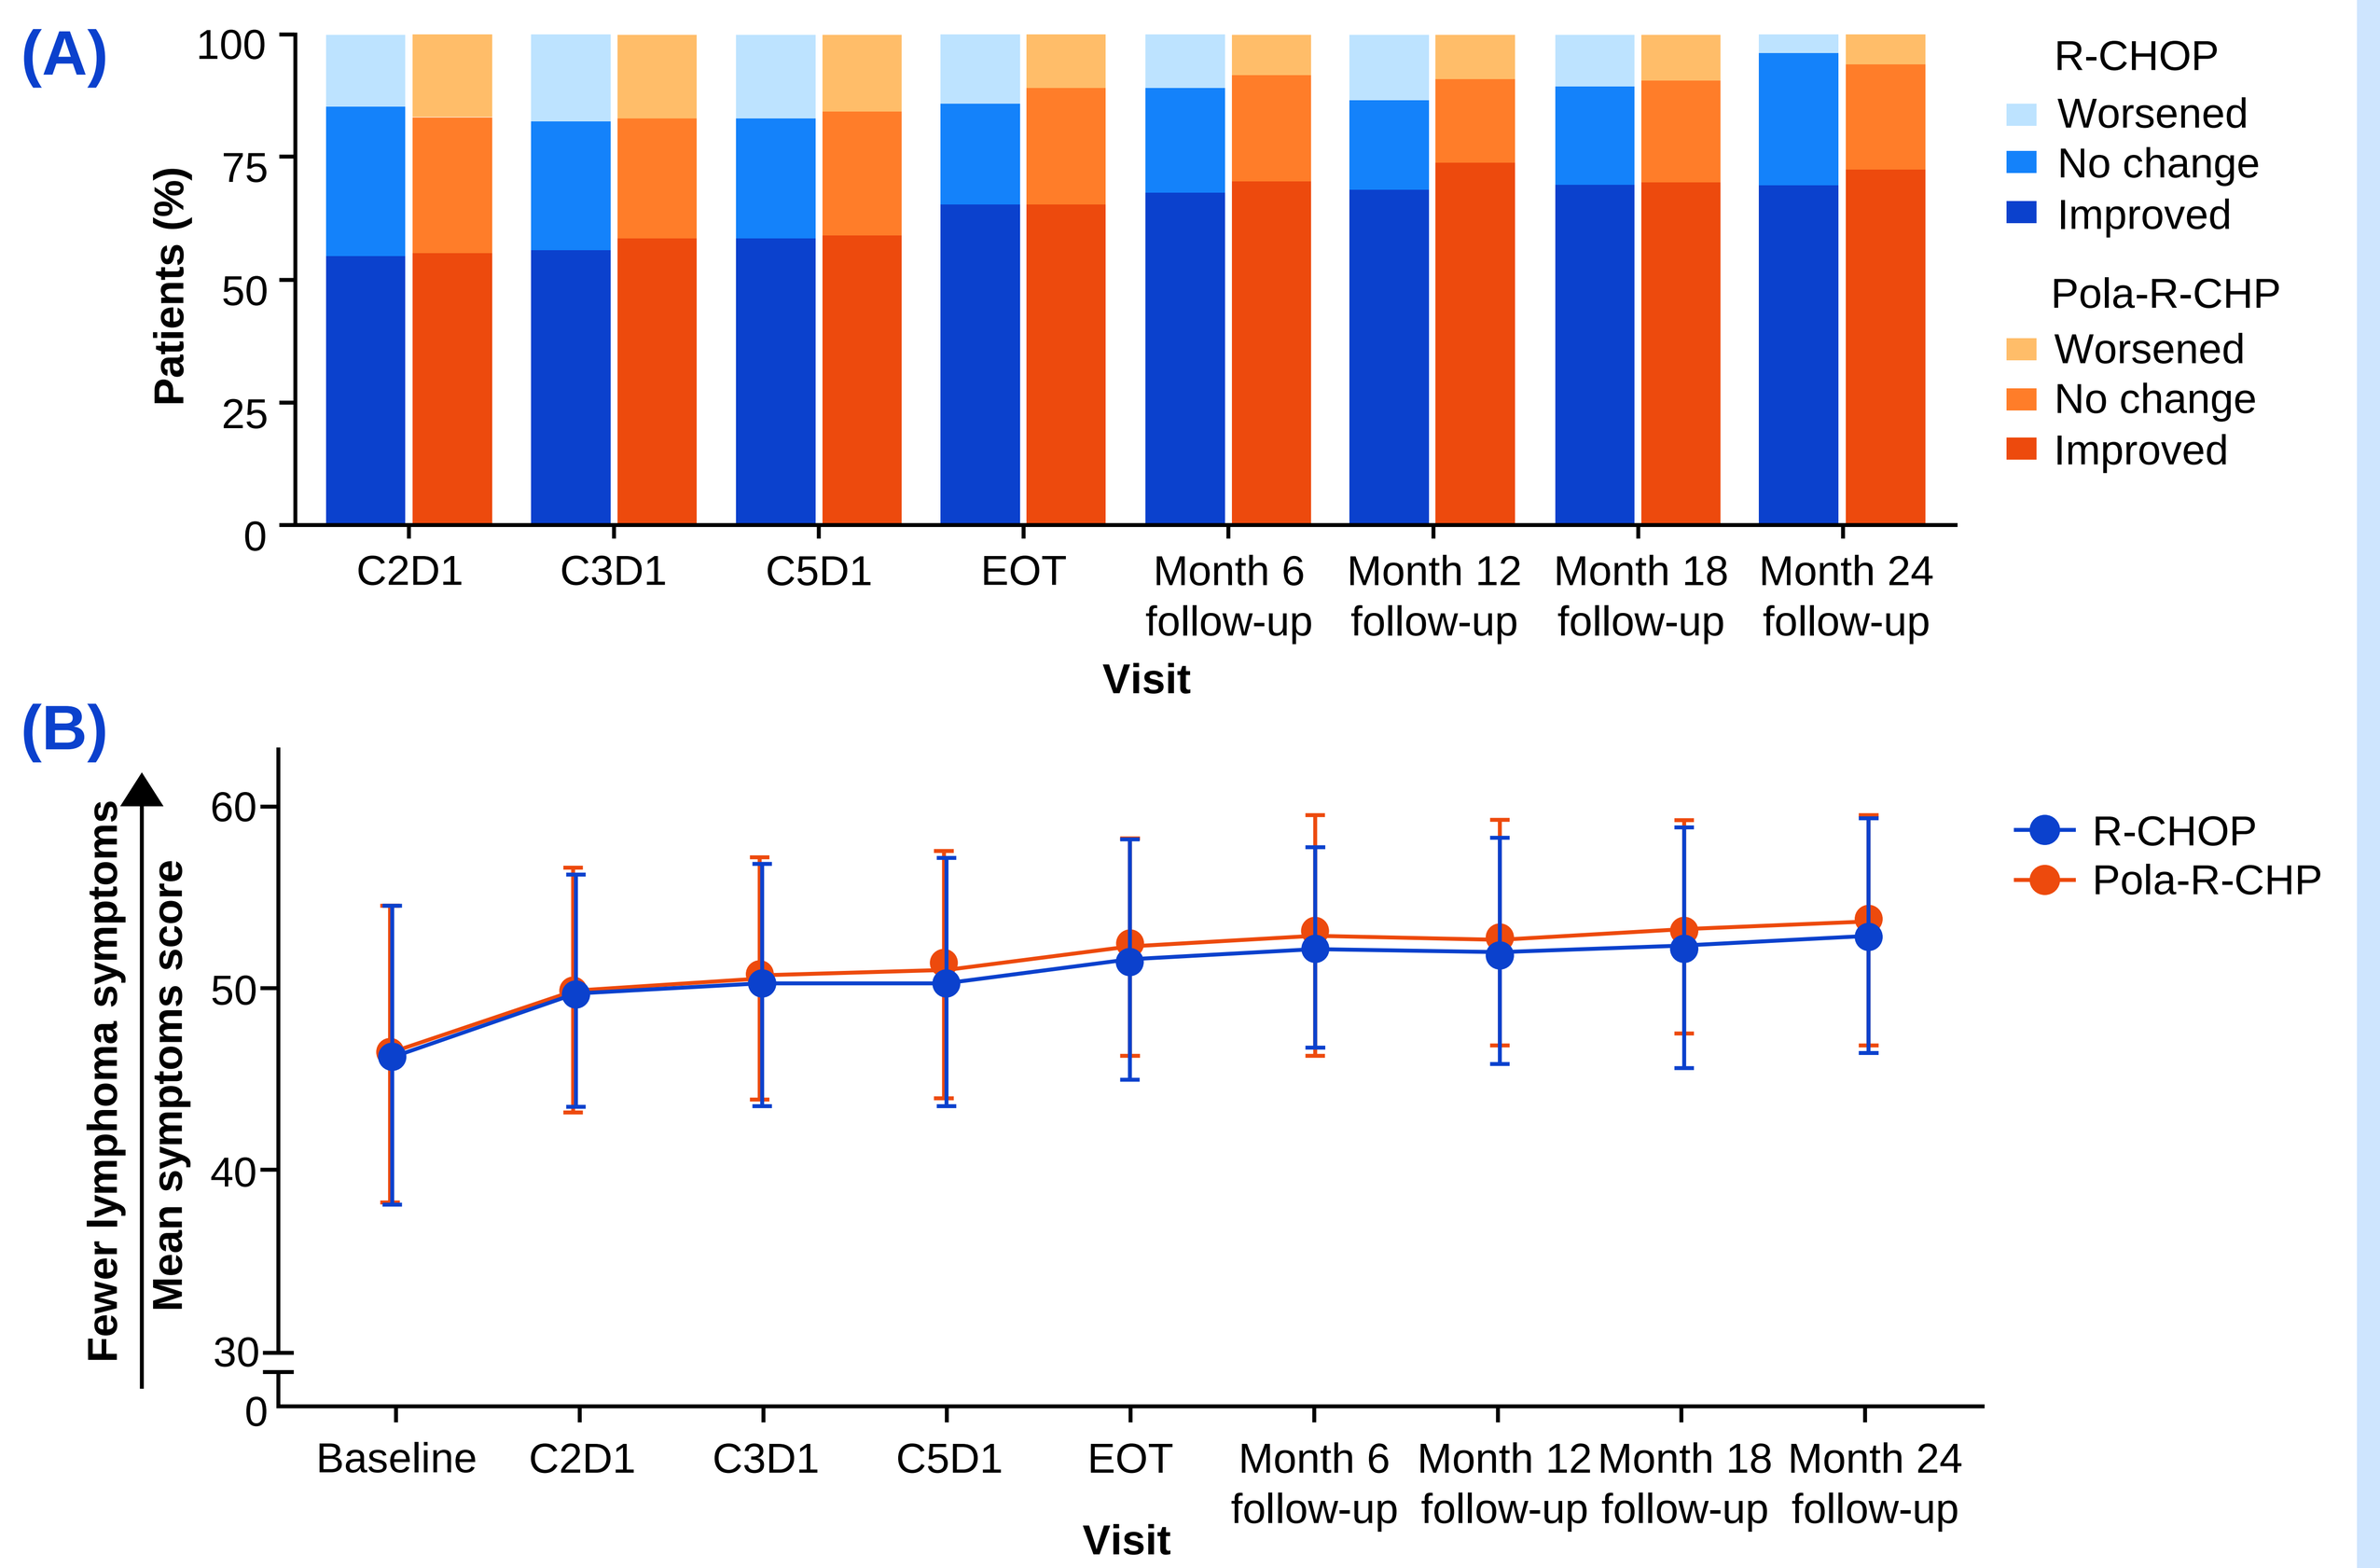
(A)
Patients (%)
0
25
50
75
100
C2D1
C3D1
C5D1
EOT
Month 6 follow-up
Month 12 follow-up
Month 18 follow-up
Month 24 follow-up
Visit
R-CHOP
Worsened
No change
Improved
Pola-R-CHP
Worsened
No change
Improved
(B)
Fewer lymphoma symptoms
Mean symptoms score
0
30
40
50
60
Baseline
C2D1
C3D1
C5D1
EOT
Month 6 follow-up
Month 12 follow-up
Month 18 follow-up
Month 24 follow-up
Visit
R-CHOP
Pola-R-CHP

*Clinically meaningful difference in symptom score categorized by improvement, worsening, or no change were defined as ≥3-point increase, decrease, and in between, respectively; †Scored on a subscale of 0–60, with higher scores representing fewer symptoms; ‡Error bars represent mean score ± one standard deviation.

QoL

- Baseline scores for QoL were similar in both treatment arms.
- Clinically meaningful improvements in QoL scores at any time point compared with baseline were comparable in patients from either treatment group and plateaued by the first assessment after EOT (**Figure 3**).

Figure 3. EORTC QLQ-C30 QoL by (A) clinically meaningful difference* and (B) mean changes over time.†, ‡

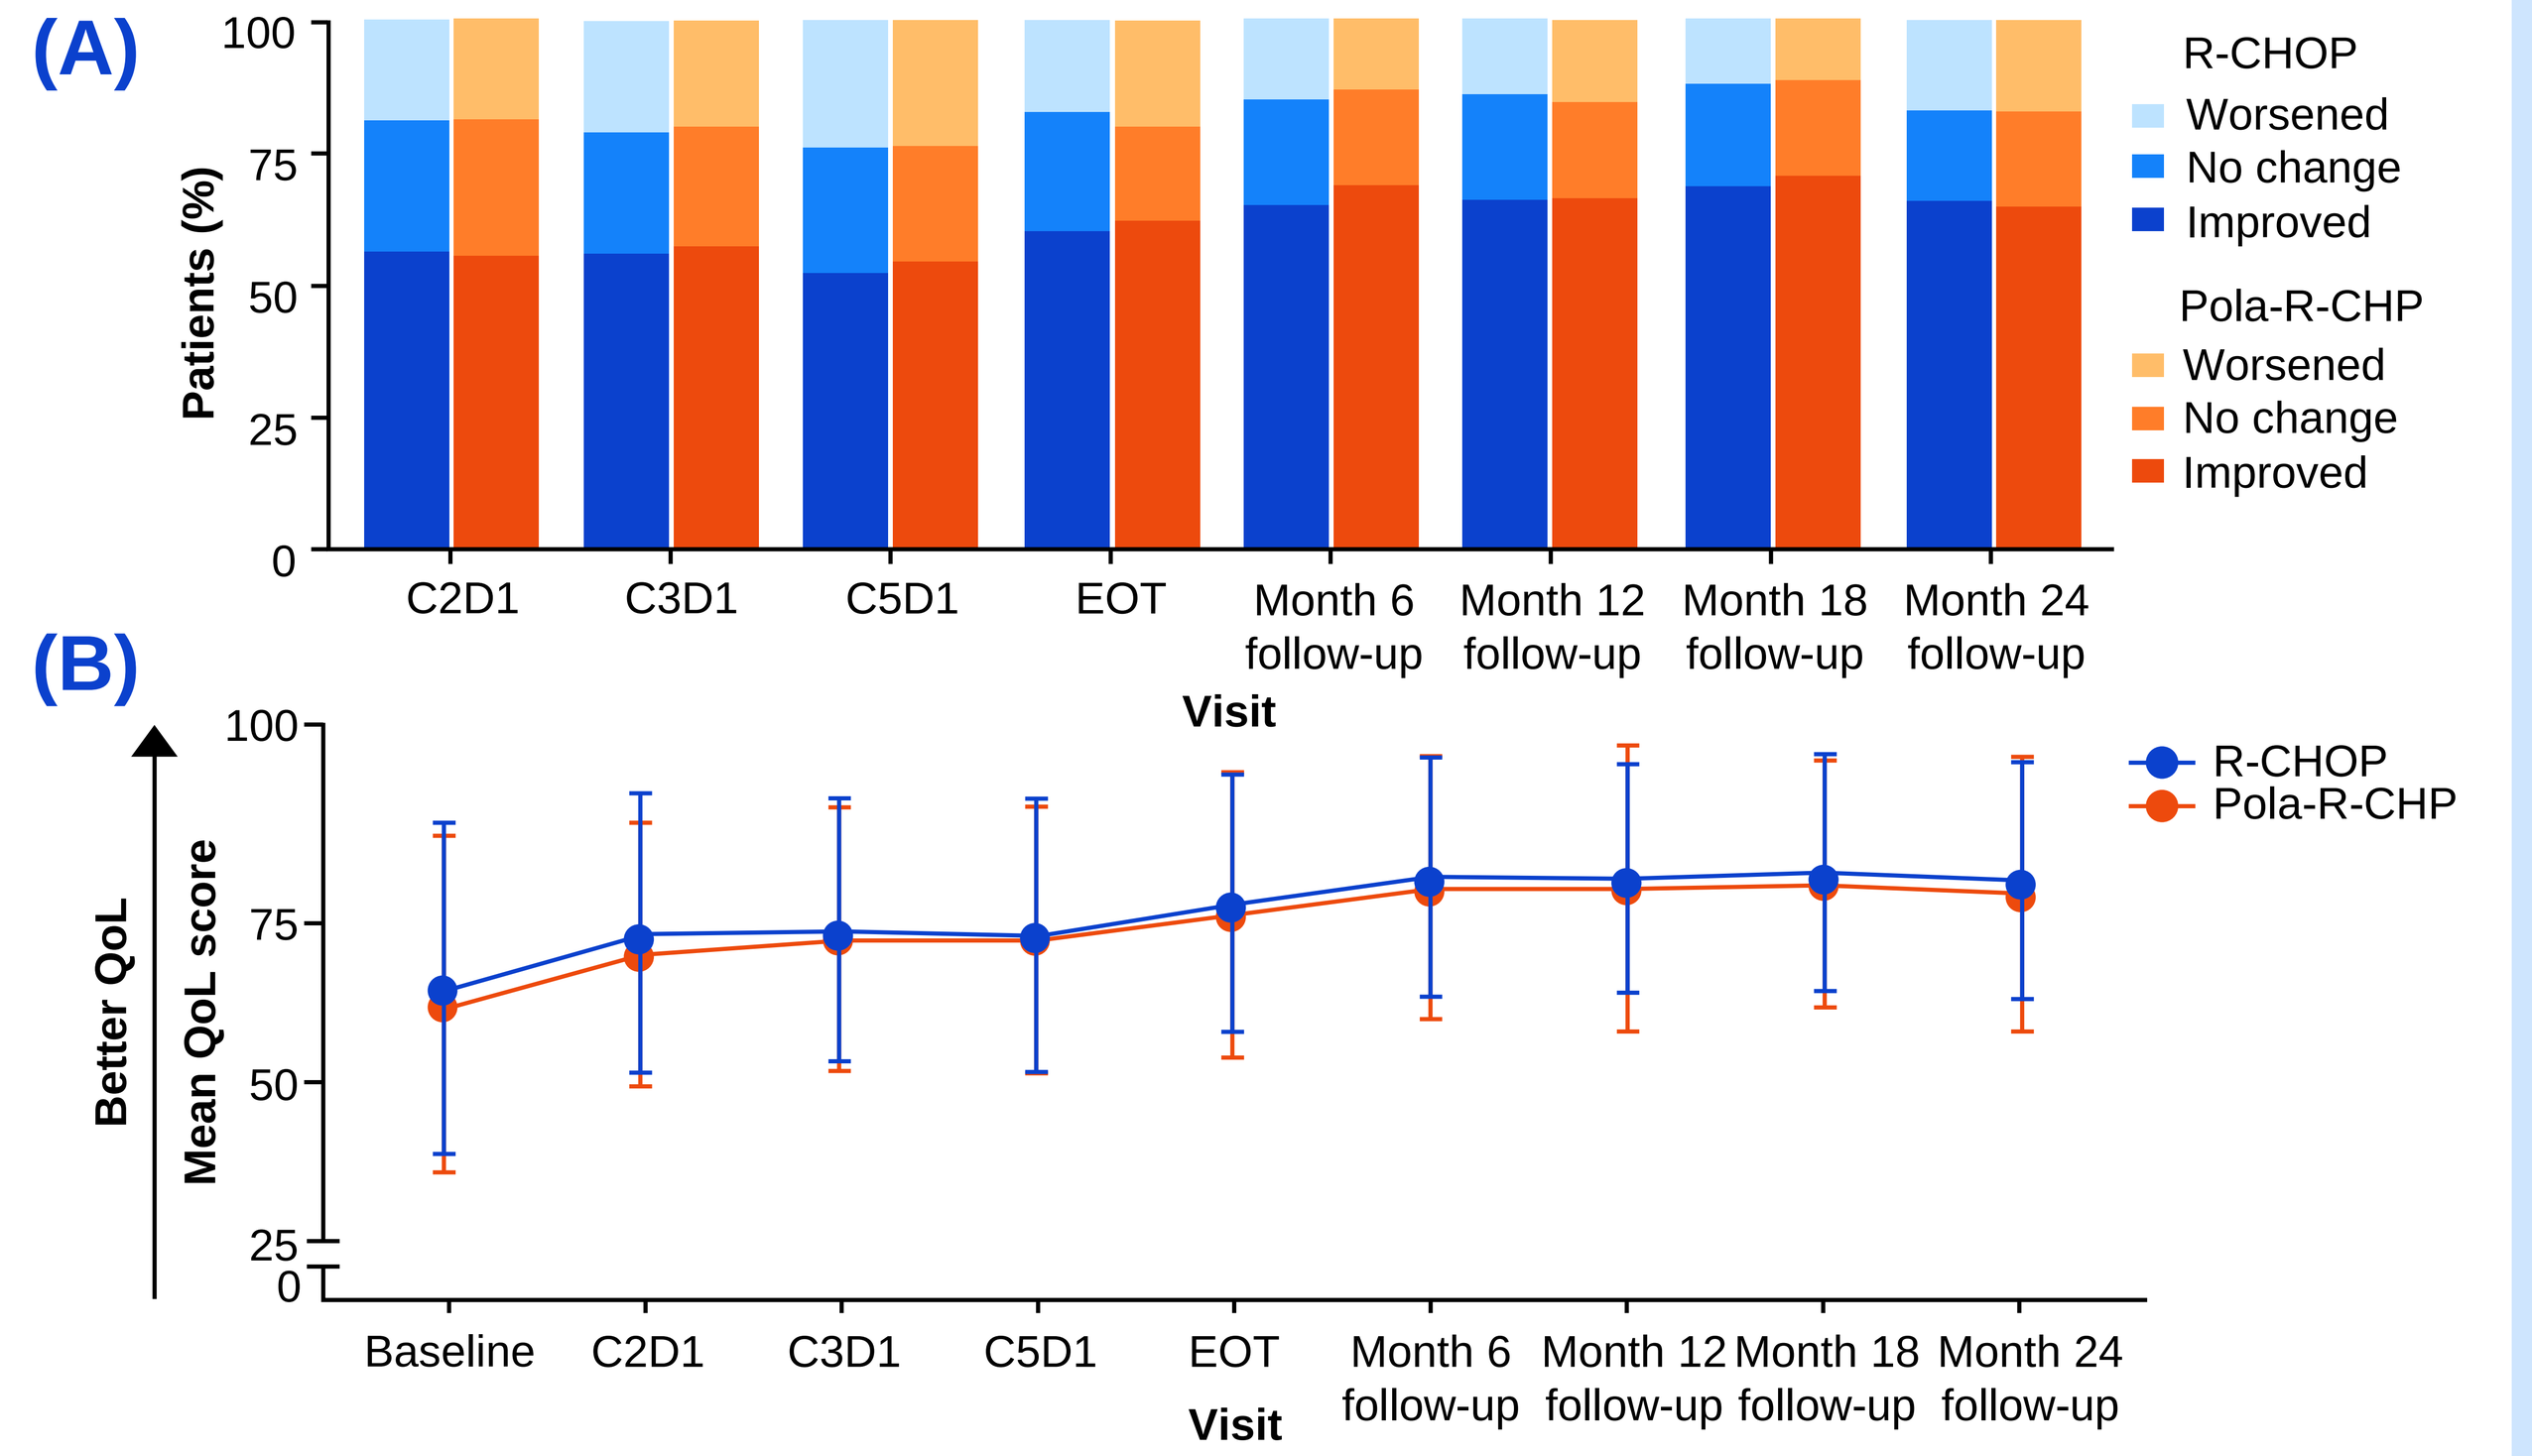
(A)
Patients (%)
0
25
50
75
100
C2D1
C3D1
C5D1
EOT
Month 6 follow-up
Month 12 follow-up
Month 18 follow-up
Month 24 follow-up
Visit
R-CHOP
Worsened
No change
Improved
Pola-R-CHP
Worsened
No change
Improved
(B)
Better QoL
Mean QoL score
0
25
50
75
100
Baseline
C2D1
C3D1
C5D1
EOT
Month 6 follow-up
Month 12 follow-up
Month 18 follow-up
Month 24 follow-up
Visit
R-CHOP
Pola-R-CHP

*Clinically meaningful difference in QoL categorized by improvement, worsening, or no change were defined as ≥8-point increase, ≥10-point decrease, and in between, respectively; †Scored on a subscale of 0–100, with higher scores representing fewer symptoms; ‡Error bars represent mean score ± one standard deviation.

Physical functioning scores

- At baseline, scores for physical functioning were similar in both treatment arms.
- Clinically meaningful improvements in physical functioning scores at any time point compared with baseline were similar between patients who received Pola-R-CHP (42.4%) or R-CHOP (39.6%).
- Improvements plateaued by the first assessment after EOT (**Figure 4**).

Figure 4. Mean changes over time* in EORTC QLQ-C30 physical functioning score.†

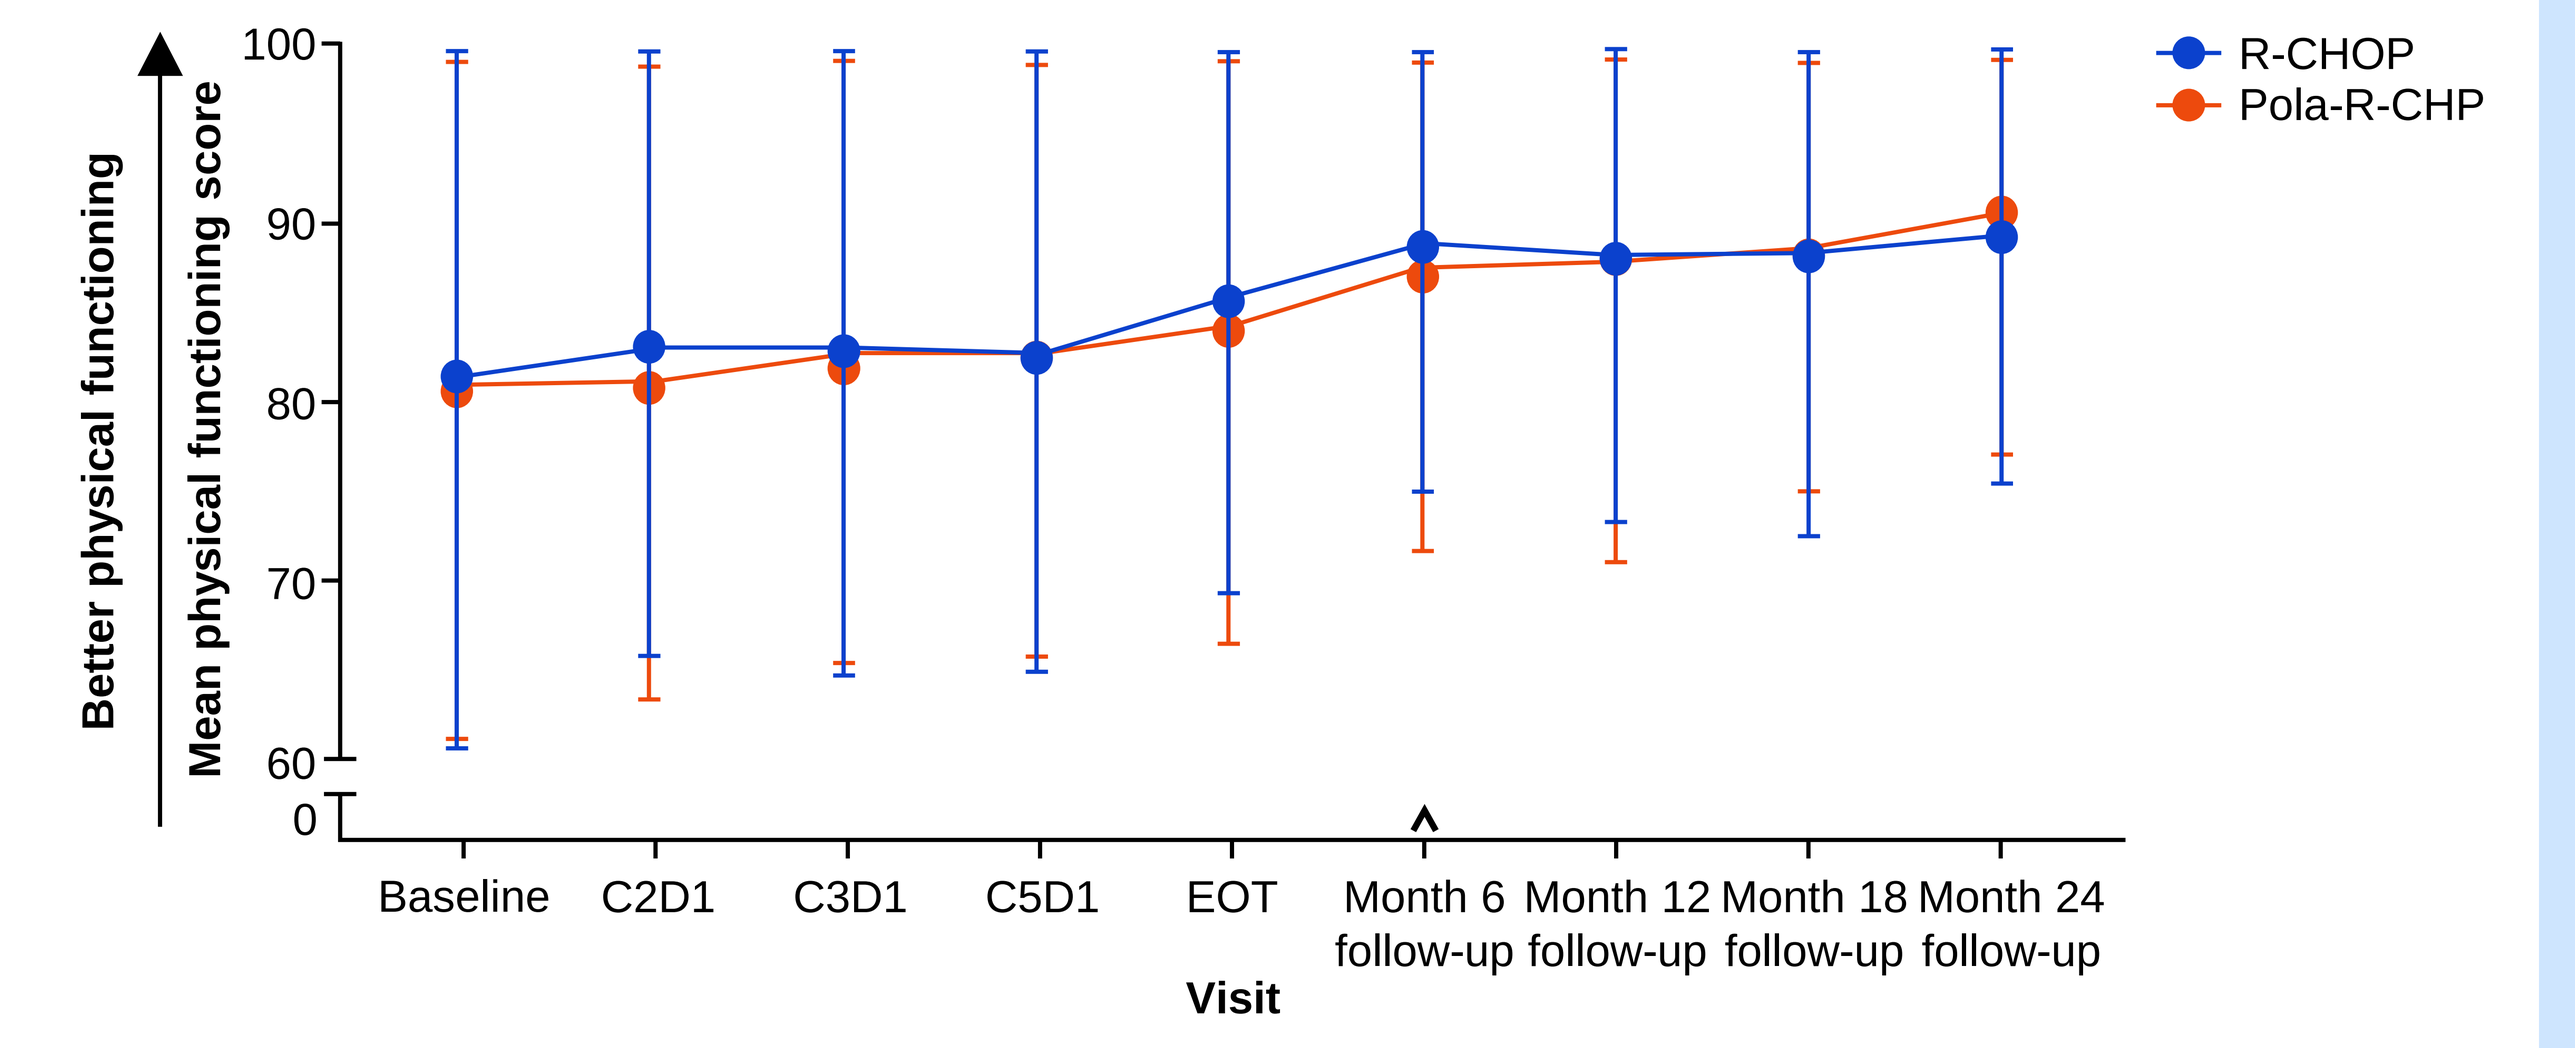
Better physical functioning
Mean physical functioning score
0
60
70
80
90
100
Baseline
C2D1
C3D1
C5D1
EOT
Month 6 follow-up
Month 12 follow-up
Month 18 follow-up
Month 24 follow-up
Visit
^
R-CHOP
Pola-R-CHP

*Scored on a subscale of 0–100, with higher scores representing fewer symptoms; †Error bars represent mean score ± one standard deviation; ^31.5% of patients in the Pola-R-CHP arm and 30.9% of patients in the R-CHOP arm showed clinically meaningful improvements in physical functioning at Month 6 follow-up based on **Table 1**.

Fatigue score

- At baseline, scores for fatigue were similar in both treatment arms.
- Clinically meaningful improvements in fatigue symptom scores at any time point compared with baseline were also similar between patients who received Pola-R-CHP (74.8%) or R-CHOP (68.2%).
- Improvements plateaued by the first assessment after EOT (**Figure 5**).

Figure 5. Mean changes over time* in EORTC QLQ-C30 fatigue score.†

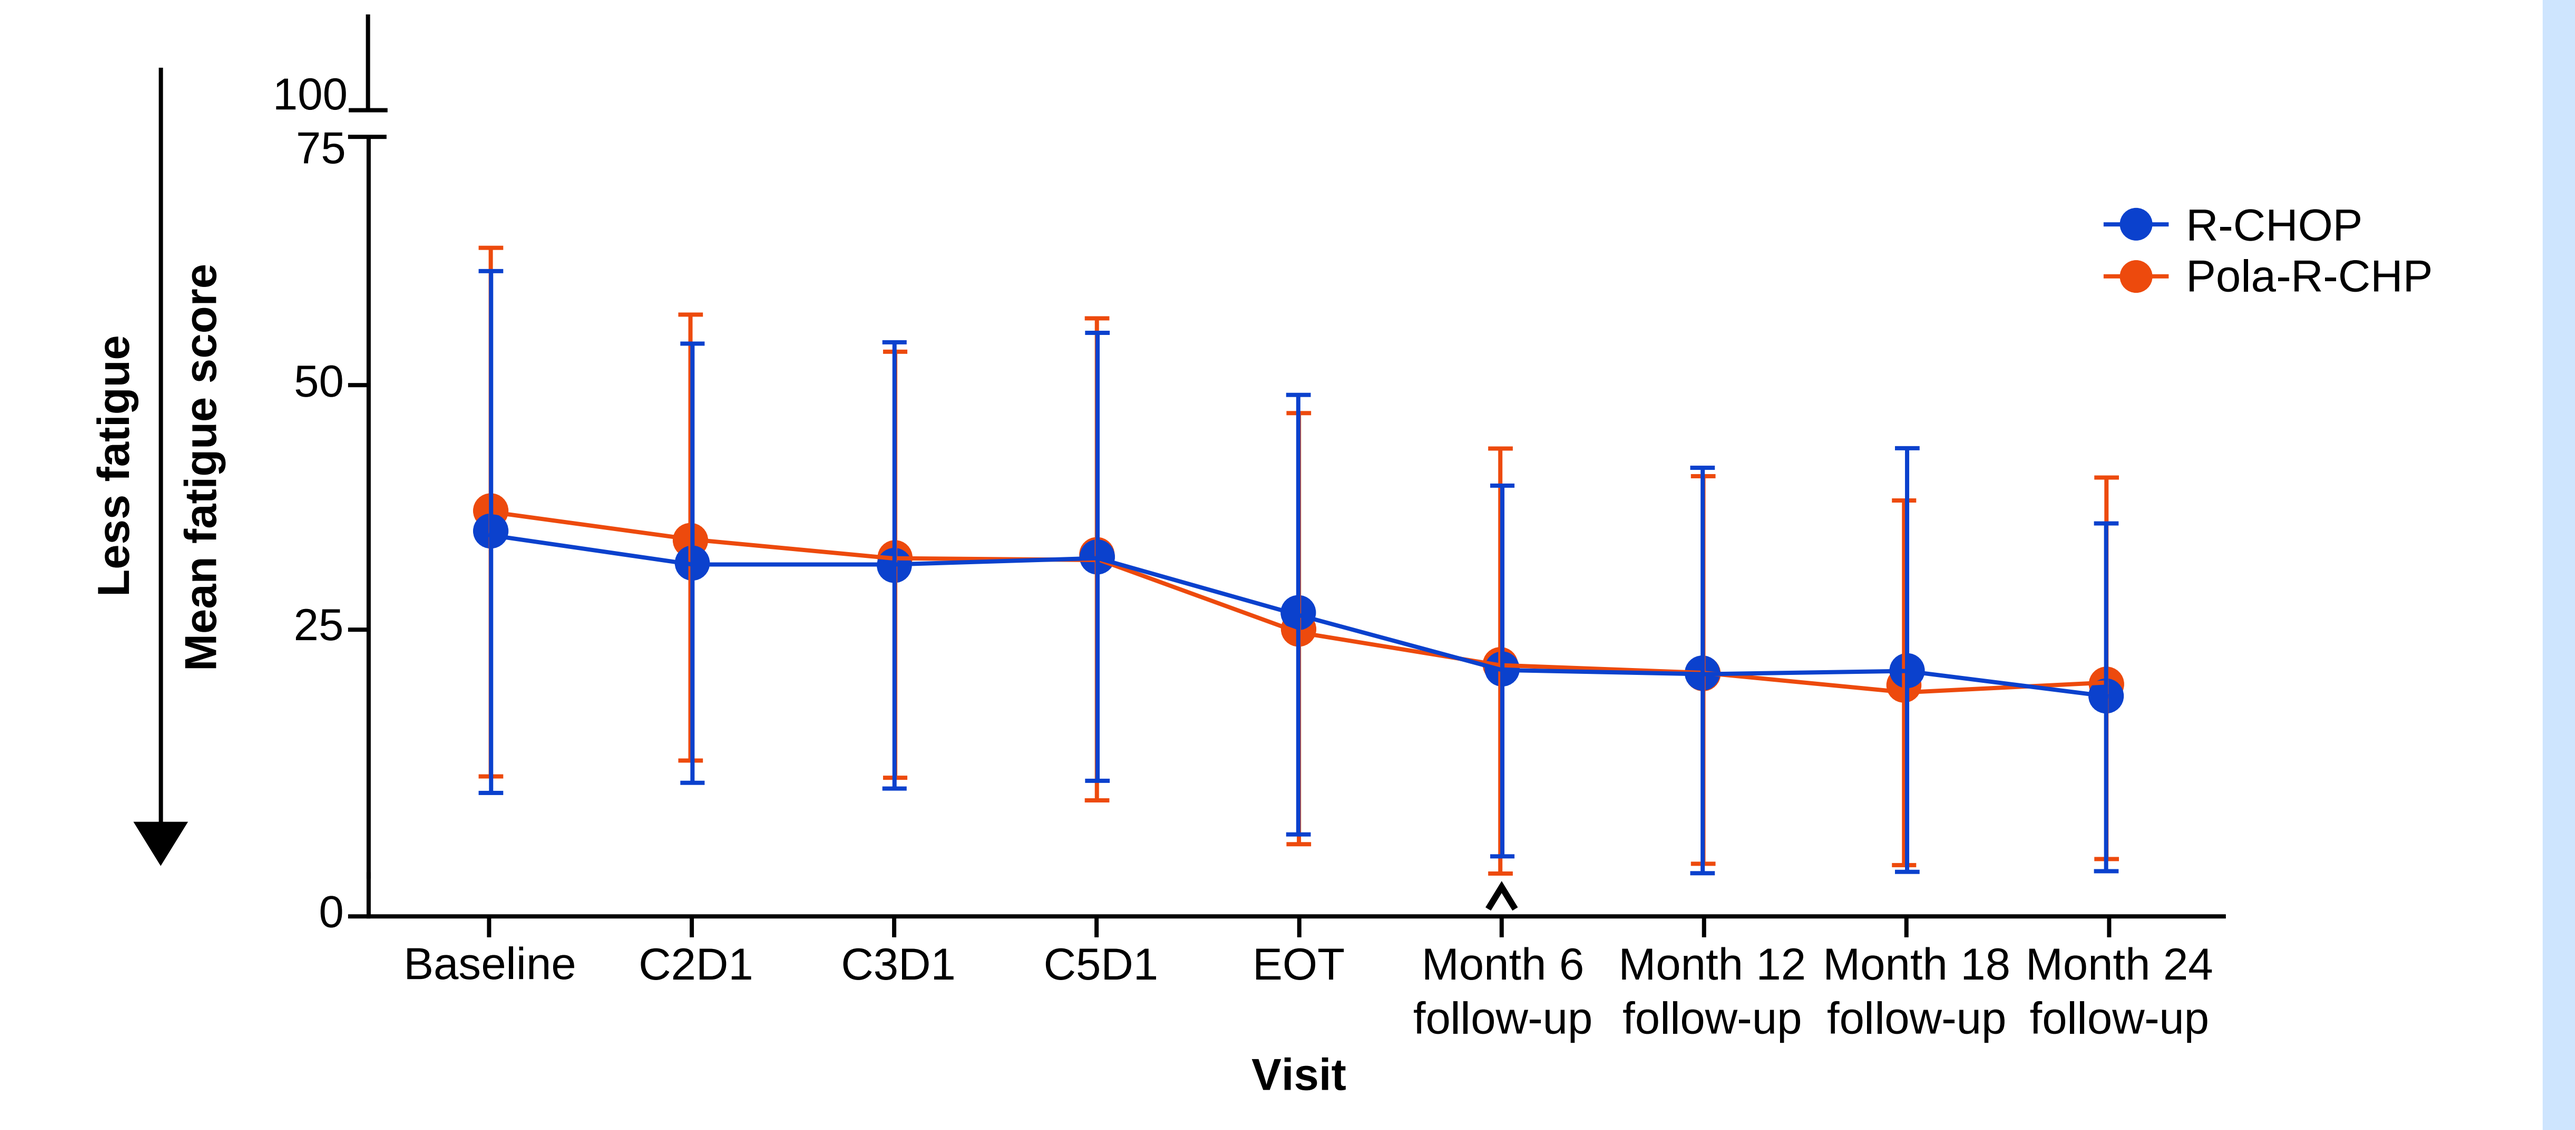
Less fatigue
Mean fatigue score
0
25
50
75
100
Baseline
C2D1
C3D1
C5D1
EOT
Month 6 follow-up
Month 12 follow-up
Month 18 follow-up
Month 24 follow-up
Visit
^
R-CHOP
Pola-R-CHP

*Scored on a subscale of 0–100, with lower scores representing fewer symptoms; †Error bars represent mean score ± one standard deviation; ^57.4% of patients in the Pola-R-CHP arm and 56.1% of patients in the R-CHOP arm showed clinically meaningful improvements in fatigue at Month 6 follow-up based on **Table 1**.

Conclusions

- Both Pola-R-CHP and R-CHOP led to rapid improvements in aspects of HRQoL that were sustained through EOT and follow-up.
- Improvements in lymphoma symptom scores were seen in the majority of patients after Cycle 1 and were maintained in most patients.
- QoL, physical functioning and fatigue scores were similar at baseline between arms and were improved during and after treatment.
- These data may represent a benchmark for patient-reported HRQoL in frontline DLBCL in the modern era.

Presented at the 2022 American Society of Hematology Annual Meeting | December 10–13, 2022

References

1. Hlubocky FJ, et al. Leuk Lymphoma 2013;54:1942–1946.
2. Tilly H, et al. N Engl J Med 2022;386:351–363.
3. Trněný M, et al. J Clin Oncol 2022;40:7561.
4. Aaronson NK, et al. J Natl Cancer Inst 1993;85:365–376.

Acknowledgments

The POLARIX study (NCT03274492) was sponsored by F. Hoffmann-La Roche Ltd and Genentech, Inc. Third-party editorial assistance, under the direction of the authors, was provided by Leen Al-Mohammad, BSc, of Ashfield MedComms, an Inizio company, and was funded by F. Hoffmann-La Roche Ltd.

Disclosures

JWF, CTho: report no COI; **MT:** consultancy: Takeda, Bristol-Myers Squibb, Incyte, AbbVie, Amgen, Roche, Gilead Sciences, Janssen, MorphoSys, Novartis, Zentiva; research funding: Roche, Takeda, Novartis; Honoraria: Takeda, Bristol-Myers Squibb, Incyte, AbbVie, Amgen, Roche, Gilead Sciences, Janssen, MorphoSys, Novartis, Zentiva; **FM:** Consultancy: Roche, Gilead, AbbVie; membership on an entity's Board of Directors or advisory committees: Roche, Gilead, Novartis, Bristol-Myers Squibb, AbbVie, Genmab, Miltenyi, Allogene therapeutics, AstraZeneca, Janssen; **GS:** consultancy: AbbVie, BeiGene, Bristol-Myers Squibb/Celgene, Epizyme, Genentech/Roche, Genmab, Incyte, Janssen, Kite/Gilead, Loxo, Miltenyi, Molecular Partners, MorphoSys, Nordic Nanovector, Novartis, Rapt, Takeda; Debiopharm/VelosBio/Ipsen; stockholder: Owkin; honoraria: AbbVie, Bayer, Incyte, Kite/Gilead. MorphoSys, Novartis, Regeneron; **PMR:** consultancy: Kite, Caribou Bio; research funding: Genentech, Seagen; **MH:** advisory and/or honoraria: Roche, Takeda, Otsuka, BeiGene, Gilead, Janssen, Novartis, Mundipharma; **PS:** employment: Medical University of Lodz, Poland, consultancy: Hematology, Internal diseases; research funding and honoraria: Roche, Takeda; **HZ:** no COI; **CThi:** employment: Hôpital Saint-Louis; honoraria: Roche, Incyte, Novartis, Kite/Gilead, Amgen, Takeda: research funding: Roche; membership on an entity's Board of Directors or advisory committees: Roche, Incyte, Novartis, Kite/Gilead, Amgen, Takeda; **GF:** employment: Florida Cancer Specialists; honoraria: Morphosys, Pfizer, Amgen; **BH:** membership on an entity's Board of Directors or advisory committees: BeiGene, Incyte and ADC Therapeutics; **WSK:** research funding: Sanofi, BeiGene, Boryong, Roche, Kyowa Kirin, Dong-A; **MM:** speaker's bureau: Gilead, Novartis, Takeda, Roche, Membership on an entity's Board of Directors or advisory committees: Gilead, Novartis, Takeda, Roche, Eusapharma, Sandoz, Celgene; **AM:** employment: University of Alabama at Birmingham; consultancy: Gilead, AstraZeneca, Pharmacyclics, Seattle Genetics, Incyte, MorphoSys/Incyte, TG Therapeutics, Kyowa Kirin, Novartis, Bristol-Myers Squibb, BeiGene; research funding: Incyte, Takeda, Forty seven Inc/Gilead, Juno pharmaceuticals/Bristol-Myers Squibb, Celgene/Bristol-Myers Squibb, Innate pharmaceuticals, Seattle Genetics, TG Therapeutics, Affimed, Merck, Kite/Gilead, Roche/Genentech, I-MAB; speaker's bureau: Gilead, AstraZeneca, Pharmacyclics, Seattle Genetics, Incyte, MorphoSys/Incyte, Kyowa Kirin, Bristol-Myers Squibb, BeiGene; **ACB:** employment and equity: Roche/Genentech; **MY:** employment and equity: Roche; **JH:** employment: Genentech; stockholder: Roche; **MS:** employment, equity and stockholder: Genentech; **CL:** employment: Genentech and equity: Roche; **JS:** AbbVie, AstraZeneca, ADC Therapeutics, BeiGene, Bristol-Myers Squibb, Lilly, TG Therapeutics; research funding: AbbVie, AstraZeneca, ADC Therapeutics, BeiGene, Bristol-Myers Squibb, Lilly, TG Therapeutics; membership on an entity's Board of Directors or advisory committees: Araris.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the lead author of this poster.

Download this presentation: https://bit.ly/3UAHrmk