1170

Characteristics and Healthcare Utilization of Patients with Mild or Moderate Hemophilia A in the US - An Analysis from the PicnicHealth Cohort

Anjali Sharathkumar MD[1]*, Yanrong Jiang MD, MRCP[2,3], Hwai Jing Hiew MD[3,4], Athanasios Siadimas MSc[5], Francis Nissen MD, PhD[5], Gillian Hanson ND[6], Emily Cibelli PhD[6], Katya Moreno MD[5], Johnny Wharton PhD[3], Angela C. Weyand MD[7]

*Presenting Author

Summary

Data from the PicnicHealth platform were used to evaluate the characteristics and utilization of healthcare facilities by people with mild or moderate hemophilia A (HA)

113 people with HA enrolled in the PicnicHealth platform met the inclusion criteria for this analysis

The data generated from this study may aid planning for, and delivery of, the most appropriate and effective care for this group of patients

Notable variations were seen in the geographical distribution of healthcare facility visits

Affiliations: [1]University of Iowa, Carver College of Medicine, Iowa City, USA; [2]University College London Hospitals NHS Foundation Trust, London, UK; [3]Roche Products Ltd, Welwyn Garden City, UK; [4]University Hospital Southampton NHS Trust, Southampton, UK; [5]F. Hoffmann-La Roche Ltd, Basel, Switzerland; [6]PicnicHealth, San Francisco, USA; [7]University of Michigan, Ann Arbor, USA

Background

- People with mild or moderate hemophilia A (HA) constitute 40–52% of all people with HA (PwHA); nonetheless, their bleeding phenotype is not well characterized in real-world data and published literature.[1]
- To better understand the real-world characteristics of this population, we used a novel research platform, PicnicHealth. This approach integrates medical data from patients' health records and patient-reported real-time outcomes, including bleeding occurrence, treatment for bleeds, and pain.[1,2]
- The primary aim of the study was to evaluate the epidemiology of the mild and moderate HA population; the secondary aim was to characterize the association between demography and healthcare utilization, assessed by visits to healthcare facilities, during the COVID-19 pandemic.

Methods

- PwHA have been enrolled into the PicnicHealth online record management platform from June 2020 through June 2022; patients provided consent prior to participating in the study.
- Inclusion criteria for this cohort with mild or moderate HA were based on baseline factor (F)VIII activity level (mild, >5–50%; moderate, 1–5%).
- Healthcare records were collected from all known healthcare providers and facilities. These records included all outpatient visits with providers (hematology, primary care, and other specialty care), laboratory reports, imaging reports, pathology, and inpatient encounters.
- Healthcare utilization was assessed based on provider visits, emergency room visits and hospitalizations before and after the COVID-19 pandemic; cost utilization was not studied.
- Descriptive statistical analyses were performed to summarize cohort characteristics.

Demographic and disease characteristics

- The study period was 2 years.
- 113 PwHA were included; 68 (60.2%) had mild HA and 45 (39.8%) had moderate HA (Table 1).

Table 1. Demographics of people with mild or moderate HA enrolled in the PicnicHealth cohort

	All (N=113)	Mild (n=68)	Moderate (n=45)
Gender, n (%)			
Female	23 (20.4)	23 (33.8)	0
Male	90 (79.6)	45 (66.2)	45 (100.0)
Race, n (%)			
Black or African American	8 (7.1)	3 (4.4)	5 (11.1)
White	77 (68.1)	48 (70.6)	29 (64.4)
More than one race / unreported	25 (22.1)	14 (20.6)	11 (24.4)
Other race	3 (2.7)	3 (4.4)	0
FVIII inhibitor status, n (%)			
Present at any time point	8 (7.1)	6 (8.8)	2 (4.4)
Not present	105 (92.9)	62 (91.2)	43 (95.6)
Anemia, n (%)	12 (10.6)	8 (11.8)	4 (8.9)
HJHS in the 2 years prior to enrolment			
Median (Q1, Q3)	4.0 (3.0, 9.0)	3.5 (2.3, 5.3)	17.0 (17.0, 17.0)
Min / Max	0 / 17.0	0 / 9.0	17.0 / 17.0
Missing, n (%)	108 (95.6)	64 (94.1)	44 (97.8)
Annualized bleed rate			
Median (Q1, Q3)	0.2 (0, 3.6)	0 (0, 4.4)	0.3 (0, 2.9)
Min / Max	0 / 122.0	0 / 23.0	0 / 122.0
Missing, n (%)	81 (71.7)	49 (72.1)	32 (71.1)
Treatment in the 2 years prior to enrolment, n (%)*			
FVIII prophylaxis	36 (31.9)	16 (23.5)	20 (44.4)
FVIII on demand	73 (64.6)	41 (60.3)	32 (71.1)
Emicizumab	10 (8.8)	4 (5.9)	6 (13.3)

*Participants could have received more than one type of treatment, or none of these treatments, during the 2-year period.
HJHS, Hemophilia Joint Health Score; Q, quartile.

Healthcare facilities utilization in the 2 years prior to enrolment

- Overall, there were 1,079 visits to a healthcare facility reported.
- PwHA visited a healthcare facility a median (Q1, Q3) of 7 (3, 14) times and a hematology practitioner a median (Q1, Q3) of 2 (1, 3) times.
- The majority of the visits were to hematology physicians (93%; 239/256), with a smaller number to nurse practitioners or allied health professionals (7%; 17/256).
- There was no notable difference in the median number of visits to healthcare facilities or to hematology providers according to disease severity, race, or inhibitor status (Tables 2, 3 and 4).

Table 2. Healthcare utilization – number of visits to healthcare facilities in the 2 years prior to enrolment according to disease severity

	Disease severity		
	All (N=113)	Mild (n=68)	Moderate (n=45)
Visits to healthcare facilities per person, median	7	7	6
Visits to hematologist per person, median	2	2	1
Overall number of hematology provider visits			
Physician	239	123	116
Nurse	12	4	8
Allied healthcare professional	5	4	1

Table 3. Healthcare utilization – number of visits to healthcare facilities in the 2 years prior to enrolment according to race

	Race			
	Black / African American (n=8)	White (n=77)	More than one race / unreported (n=25)	Other race (n=3)
Visits to healthcare facilities per person, median	4	8	3	7
Visits to hematologist per person, median	1	2	1	0
Overall number of hematology provider visits				
Physician	7	187	44	1
Nurse	0	11	1	0
Allied healthcare professional	0	5	0	0

Table 4. Healthcare utilization – number of visits to healthcare facilities in the 2 years prior to enrolment according to FVIII inhibitor status

	FVIII inhibitor status	
	Patients with inhibitors (n=8)	Patients without inhibitors (n=105)
Visits to healthcare facilities per person, median	12	7
Visits to hematologist per person, median	1.5	2
Overall number of hematology provider visits		
Physician	24	215
Nurse	0	12
Allied healthcare professional	1	4

Geographical distribution of facility utilization

- Visits to a total of 379 healthcare facilities were reported, with 1,079 visits in total.
- The five states with the highest proportions of healthcare facilities visited by PwHA were California (22.7%), Indiana (7.7%), Texas (6.3%), Michigan (5.3%), and Louisiana (4.5%).
- The five states with the highest proportion of visits to healthcare facilities were California (25.3%), Indiana (6.9%), Virginia (5.2), Louisiana (4.4%), and Missouri (4.3%; Figure 1).

Figure 1. Geographical distribution of visits in the 2 years prior to enrolment

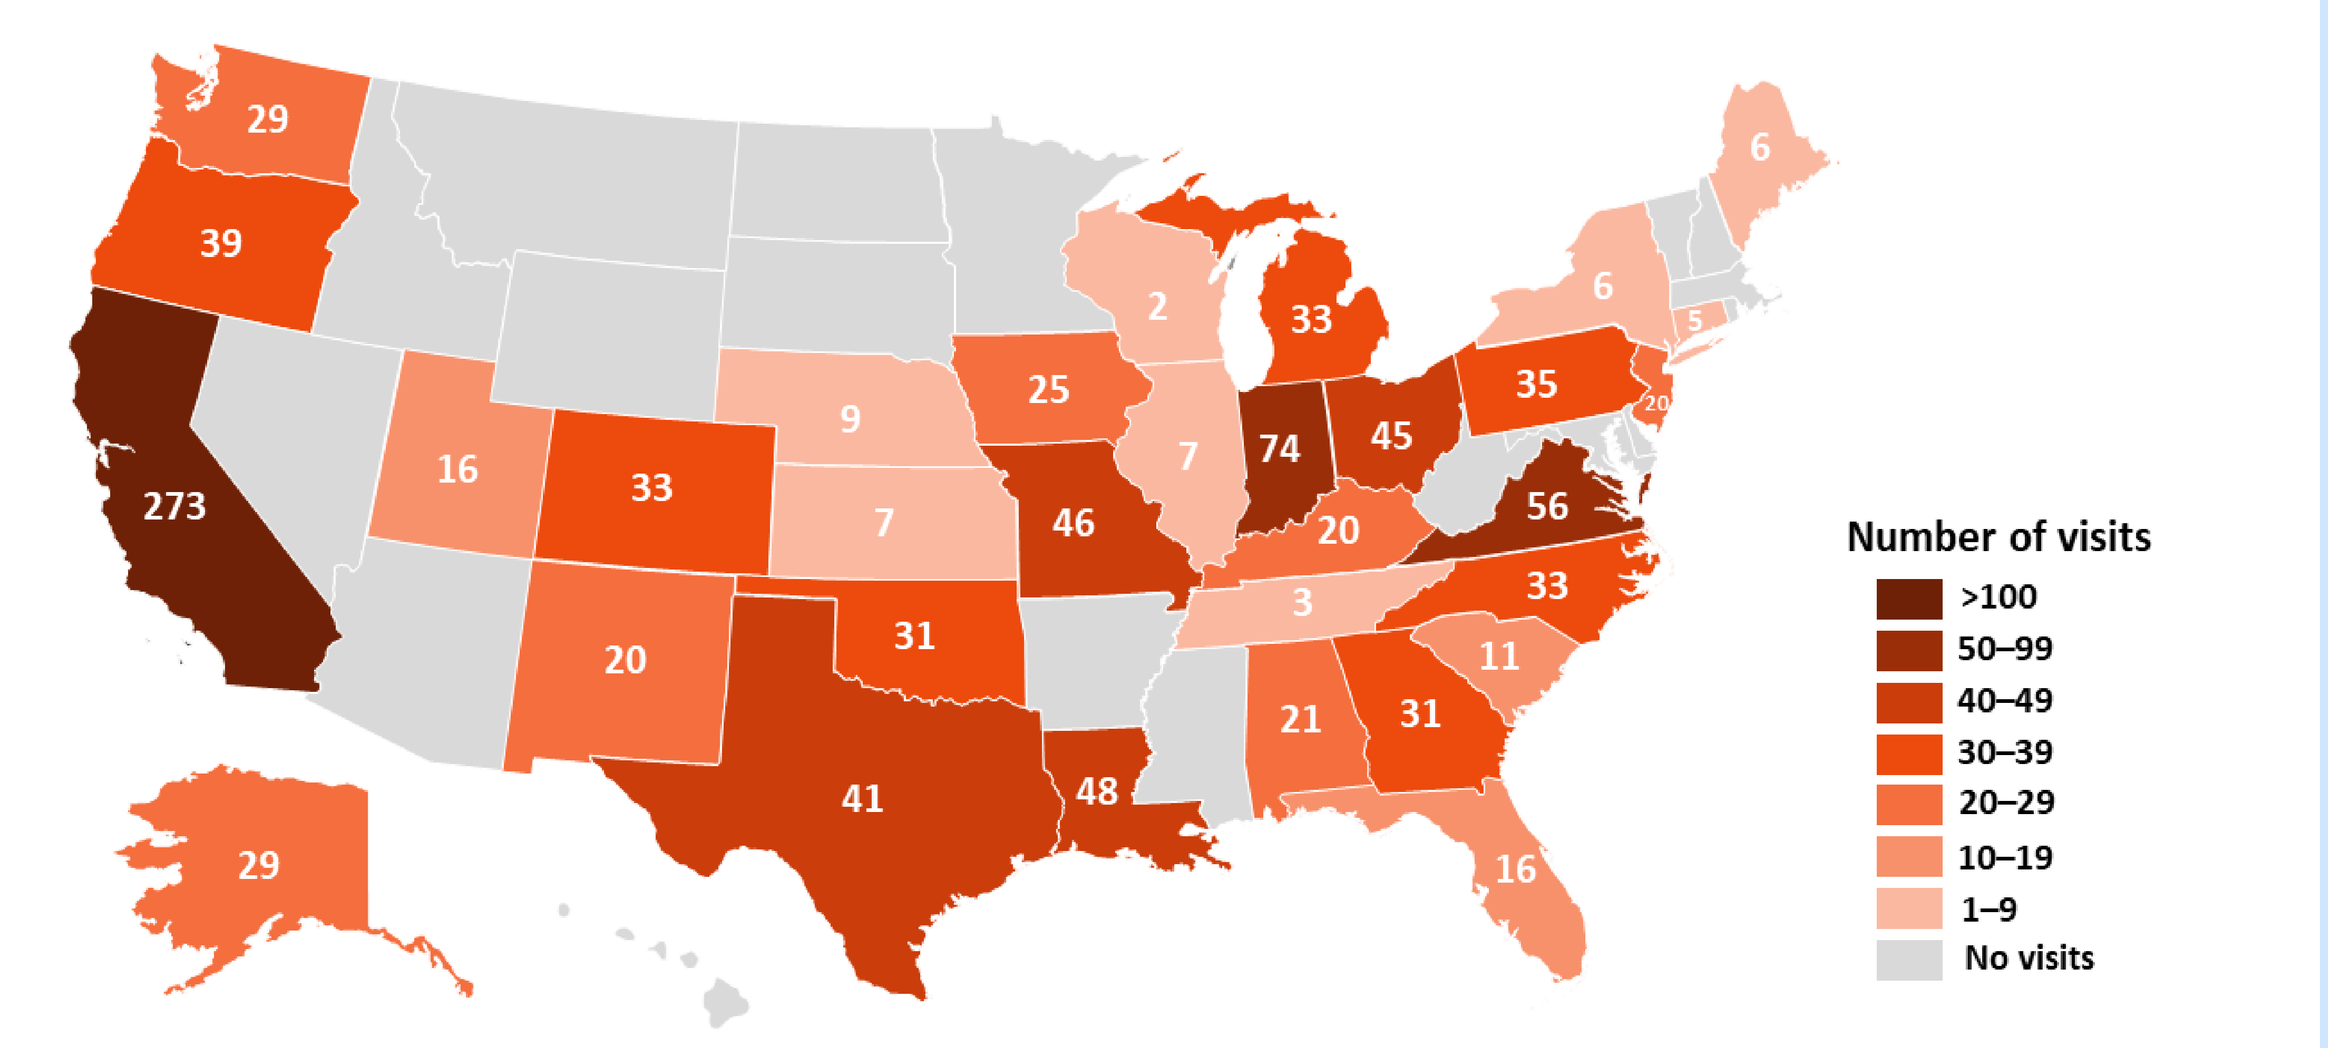

Number of visits
>100
50–99
40–49
30–39
20–29
10–19
1–9
No visits

Impact of COVID-19 pandemic on healthcare facility utilization

- There were no changes to emergency care visits from the pre-pandemic period (Jan 2019 to Jan 2020) to during the pandemic (Feb 2020 to Jun 2021).
- For routine care visits, there were variations since the pandemic began, which may be attributable to lockdown restrictions, although the number of these does not strictly correlate with COVID-19 case numbers (Figure 2).

Figure 2. Line charts of the number of PwHA with emergency and routine care visits from January 2019 to June 2021 overlaid on a bar-plot showing COVID-19 cases in the US reported to the CDC[3]

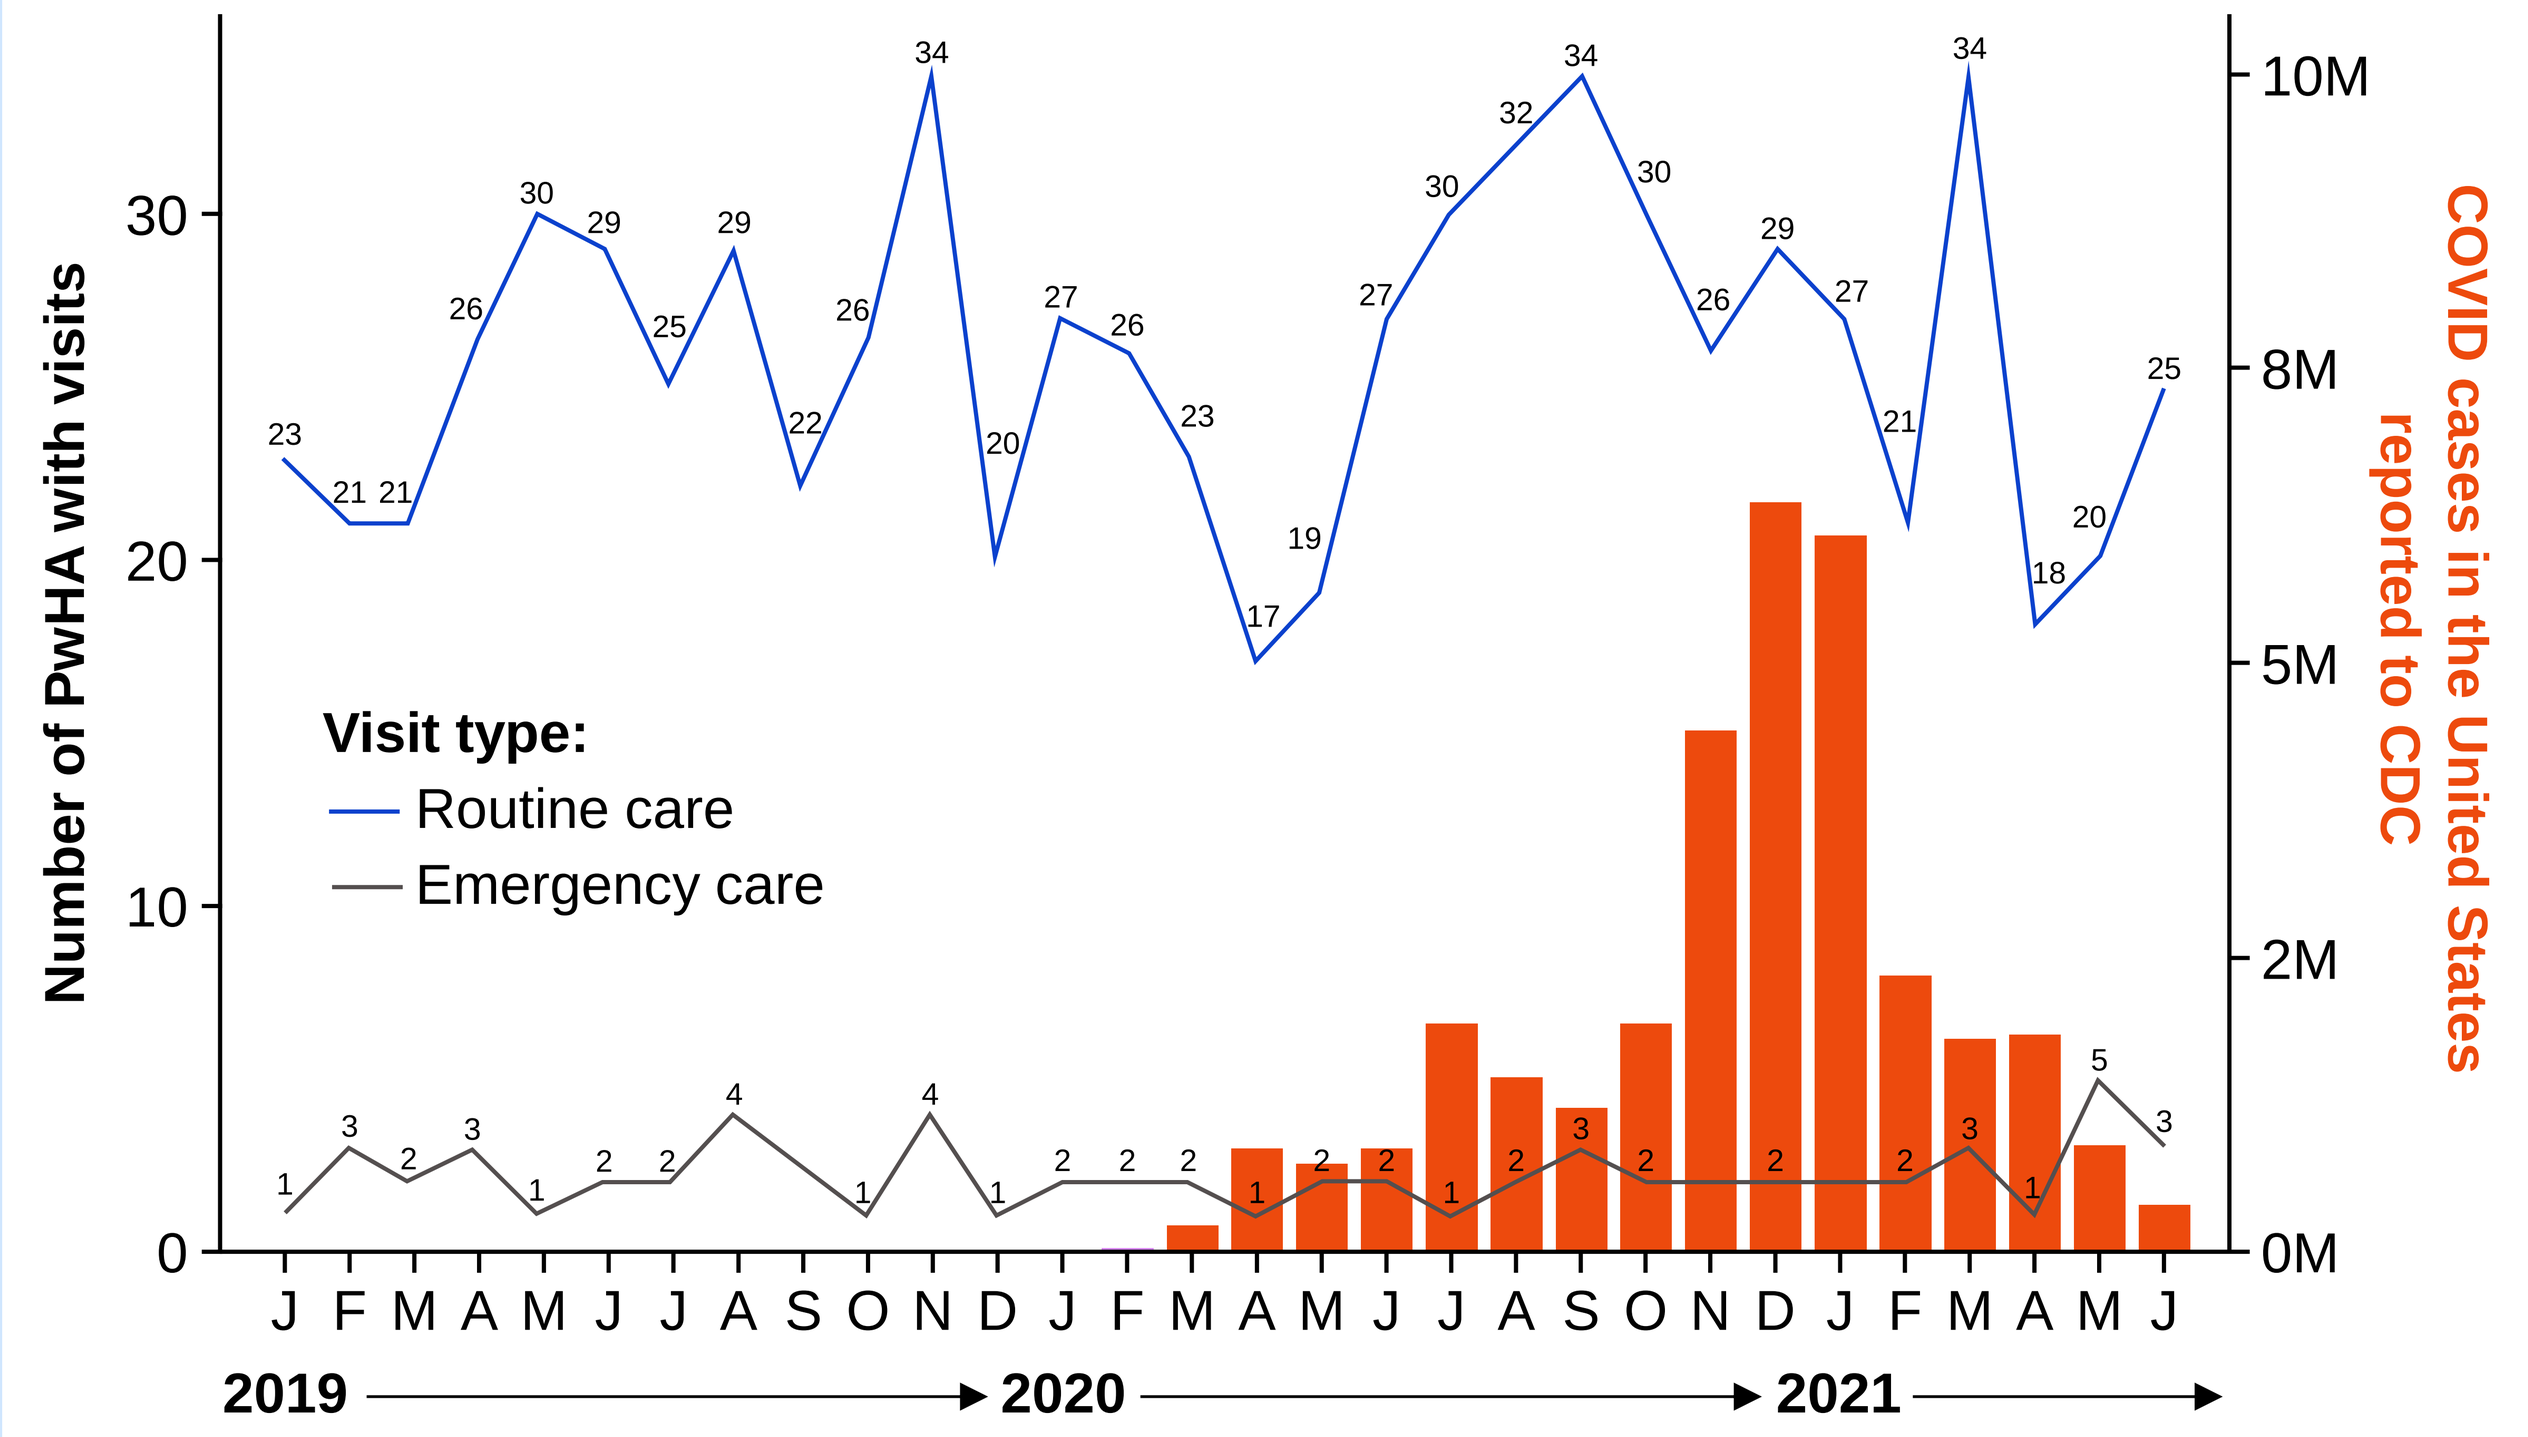

Number of PwHA with visits
COVID cases in the United States reported to CDC
Visit type:
Routine care
Emergency care
0M
2M
5M
8M
10M
J F M A M J J A S O N D J F M A M J J A S O N D J F M A M J
2019
2020
2021

Details of SARS-CoV-2 infections were not collected in the PicnicHealth database.
CDC, Centers for Disease Control and Prevention; M, million.

Conclusions

- Our study provides valuable insights into the characteristics and utilization of healthcare of the under-represented cohort of people with mild or moderate HA.
- We found notable variation in the geographical distribution of healthcare facility visits by PwHA across different states.
- Though routine visits by PwHA to healthcare facilities fluctuated, they remained relatively stable during the COVID-19 pandemic.
- The study is limited by potential bias resulting from participant reporting and the retrospective nature of the data collection.
- The data generated from this study may aid planning for, and delivery of, healthcare for this patient population.

Presented at the 2022 American Society of Hematology Annual Meeting | December 10–13, 2022

References

1. Witkop M, et al. Blood 2021;138(Suppl.1):2107.
2. Skinner MW, et al. Blood 2021;138(Suppl.1):59.
3. Centers for Disease Control and Prevention COVID Data Tracker; available from https://covid.cdc.gov/covid-data-tracker/#trends_dailycases; Accessed 19 July 2022.

Acknowledgments

This study was sponsored by F. Hoffmann-La Roche Ltd. Third party editorial assistance, under the direction of the authors, was provided by Katie Smith, PhD, of Ashfield MedComms, an Inizio company, and was funded by F. Hoffmann-La Roche Ltd.

Disclosures

AS: employment: University of Iowa; research funding: Amgen, CDC/ATHN/HRSA; honoraria: Genentech, Takeda, CSL Behring; membership on an entity's Board of Directors or advisory committees: University of Iowa Clinical Committee; YJ: employment: Roche; previous employment: Whittington Health NHS Trust; HJH: employment: Roche; previous employment: University Hospital Dorset; AS: employment, stockholder, research funding: Roche; FN, JW: employment, stock options: Roche; GH, EC: employment, stockholder, stock options: Picnic Health; KM: employment and equity: Roche; ACW: consultancy, honoraria, membership on an entity's Board of Directors or advisory committees: Genentech; consultancy, honoraria, membership on an entity's Board of Directors or advisory committees, research funding: Sanofi; consultancy, membership on an entity's Board of Directors or advisory committees, research funding: Takeda, research funding: Pfizer, Novo Nordisk; membership on an entity's Board of Directors or advisory committees: Bayer; consultancy: Spark Therapeutics.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the lead author of this poster.

Download this presentation: https://bit.ly/3tjKhjo