1171

Comorbidities and Mental Health Concerns Reported by Participants in the FARE (Food Allergy Research & Education) Patient Registry

Thomas B. Casale,[1] Christopher Warren,[2] Yamina Rajput,[3] Robert Schuldt,[3] Rongrong Wang,[3] Ahmar Iqbal,[3] Sachin Gupta,[3] Ruchi Gupta[2]

[1]University of South Florida, Tampa, FL, USA; [2]Northwestern University, Chicago, IL, USA; [3]Genentech, Inc., South San Francisco, CA, USA

Background

- Immunoglobulin E (IgE)-mediated food allergies carry a risk of severe reactions, including anaphylaxis.[1]
 - >170 foods have been associated with allergic reactions. The most frequently reported allergens include peanuts, tree nuts, egg, milk, shellfish, soy, wheat, and finned fish.[2]
 - The vigilance and lifestyle changes necessary to prevent exposure to allergens can impart significant psychosocial burdens on patients and caregivers.[3]
- The clinical phenotypes and experiences of patients with food allergy are specific to each individual and may be influenced by concomitant factors, including comorbidities and mental health status.
- However, neither the relationship between specific food allergies and the frequency of comorbidities nor the mental health status of patients with food allergy have been well studied.

Objective

- To compare self-reported characteristics, including comorbidities and mental health status, of patients with single food allergy to peanuts, tree nuts, milk, or egg versus patients with multiple food allergies using the Food Allergy Research & Education (FARE) Patient Registry database.

Methods

- The FARE Patient Registry[4] (NCT04653324) conducts online surveys of patients with food allergies, and caregivers acting on behalf of patients, and collects cross-sectional data regarding their experiences.
 - FARE has >13,000 members, aged <1 to >80 years, and tracks >200 allergens.
- Using data from the FARE Patient Registry from May 2017 to January 2021, we selected patients with the following characteristics: completed survey (n=6,167), resided in the United States, and reported ≥1 food allergy.
 - Patients were categorized to single food allergy (allergy to only 1 of peanuts, tree nuts, milk, or egg) or complex multiple food allergies (allergy to peanuts plus ≥2 to tree nuts, milk, or egg, as defined by the OUtMATCH Trial [NCT03881696]).
- A total of 1,233 patients met inclusion criteria.
- A descriptive analysis of outcomes, including comorbidities and mental health status, was conducted (note that self-reported refers to self-reported or parent/caregiver reported).
 - To identify comorbidities, responses to the survey question "Has the participant ever been diagnosed with any of the following conditions?" were collated.
 - Comorbidity event rates were calculated using total person-years of disease duration (defined as age at survey completion minus age at diagnosis).
 - To assess mental health status, responses to the mental health survey module were collated (n=342/1,233; 28%).

Results

Patient Characteristics

- Demographic characteristics were generally similar between patients with single and multiple food allergies (**Table 1**).
 - Overall, most patients:
 - Were diagnosed when they were young (87% before age 6 years)
 - Experienced food allergy for many years (12 years mean duration of disease)
 - Were evenly split by sex (50% female).

Table 1. Patient Characteristics

	Patients With Single Food Allergy					Patients With MFA
Characteristic	Overall (N=1,233)	Peanuts (n=347)	Tree Nuts (n=217)	Milk (n=138)	Egg (n=77)	Peanuts Plus ≥2 to Tree Nuts, Milk, or Egg (n=454)
Age at survey completion, y, mean (SD)	15 (15)	14 (15)	18 (15)	25 (19)	16 (20)	12 (10)
Age at food allergy diagnosis, y, mean (SD)	4 (10)	2 (7)	7 (10)	10 (18)	7 (15)	1 (3)
Duration of disease, y, mean (SD)	12 (11)	11 (13)	12 (12)	15 (11)	9 (14)	11 (9)
Female, n (%)	618 (50)	171 (49)	133 (61)	86 (62)	47 (61)	181 (40)
White, n (%)	1,005 (82)	282 (81)	184 (85)	112 (81)	64 (83)	363 (80)

MFA, multiple food allergies.

Food Allergy Comorbidities

- The most common comorbidities were atopic dermatitis, asthma, and allergic rhinitis, and the frequency of each comorbidity varied by food allergen (**Figure 1**, **Table 2**).
 - Milk allergy alone was associated with more reports of asthma by percentage of patients (**Figure 1**).
 - Atopic dermatitis was reported by more patients with multiple food allergies by percentage of patients (**Figure 1**) and by person-year adjusted comorbidity rate (**Table 2**).
- Across all groups, 39% of patients experienced anaphylaxis, which varied by food allergen.
 - Anaphylaxis was most reported by patients with milk allergy by percentage of patients (peanuts, 31%; tree nuts, 42%; milk, 57%; egg, 25%; multiple food allergies, 41%) and had the highest rate of anaphylaxis when adjusted for total person-years (peanuts, 2.72%; tree nuts, 3.63%; milk, 3.82%; egg, 2.80%, multiple food allergies, 3.65%) (**Figure 2**).

Table 2. Adjusted Rates of Comorbidity

	Patients With Single Food Allergy				Patients With MFA
Characteristic, %	Peanuts (n=347)	Tree Nuts (n=217)	Milk (n=138)	Egg (n=77)	Peanuts Plus ≥2 to Tree Nuts, Milk, or Egg (n=454)
Atopic dermatitis	3.53	3.20	1.84	3.97	5.60
Asthma	2.56	2.68	3.24	2.80	3.67
Allergic rhinitis	1.83	2.80	1.79	2.50	2.62
Contact dermatitis	0.66	0.72	0.68	1.18	1.13
Drug allergy	0.43	0.96	1.26	1.77	0.97
Heartburn	0.63	1.08	1.16	1.32	0.70

MFA, multiple food allergies.

Figure 1. Percent of Patients Experiencing Comorbidities

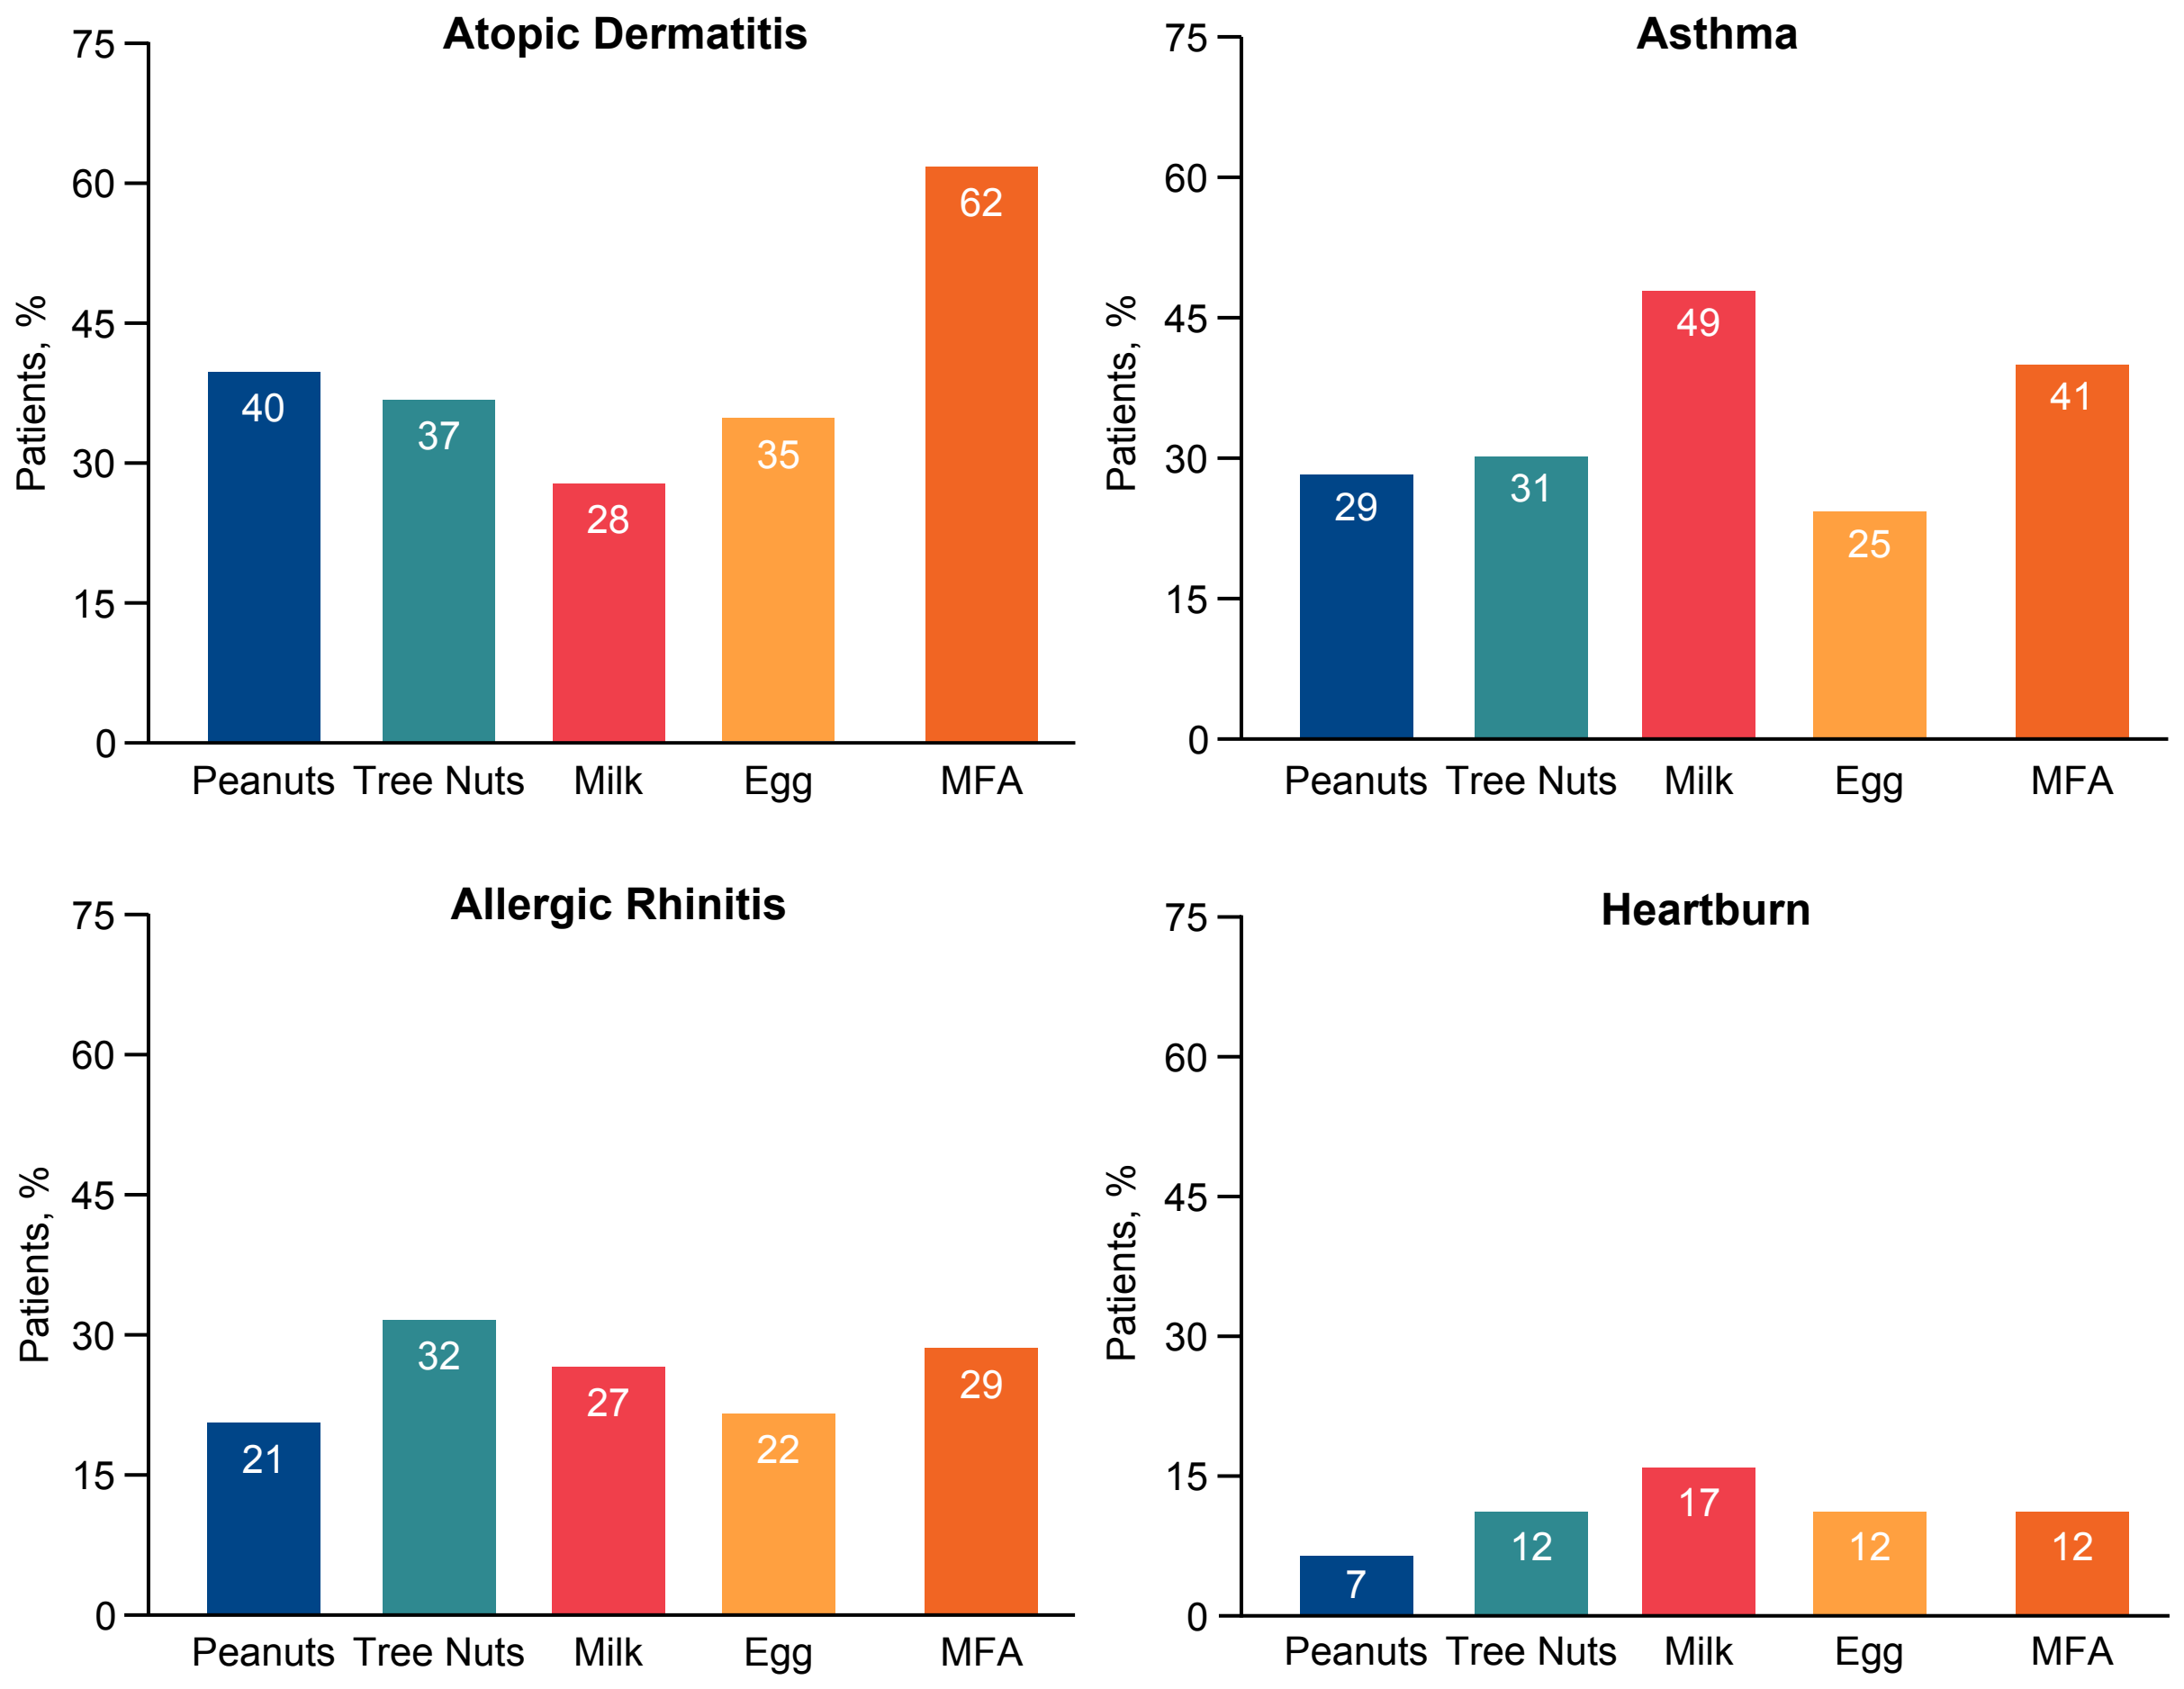
Atopic Dermatitis
Asthma
Allergic Rhinitis
Heartburn
Patients, %
0
15
30
45
60
75
Peanuts
Tree Nuts
Milk
Egg
MFA
40
37
28
35
62
29
31
49
25
41
21
32
27
22
29
7
12
17
12
12

MFA, multiple food allergies (peanuts plus ≥2 to tree nuts, milk, or egg).

Figure 2. Percent of Patients Experiencing Anaphylaxis

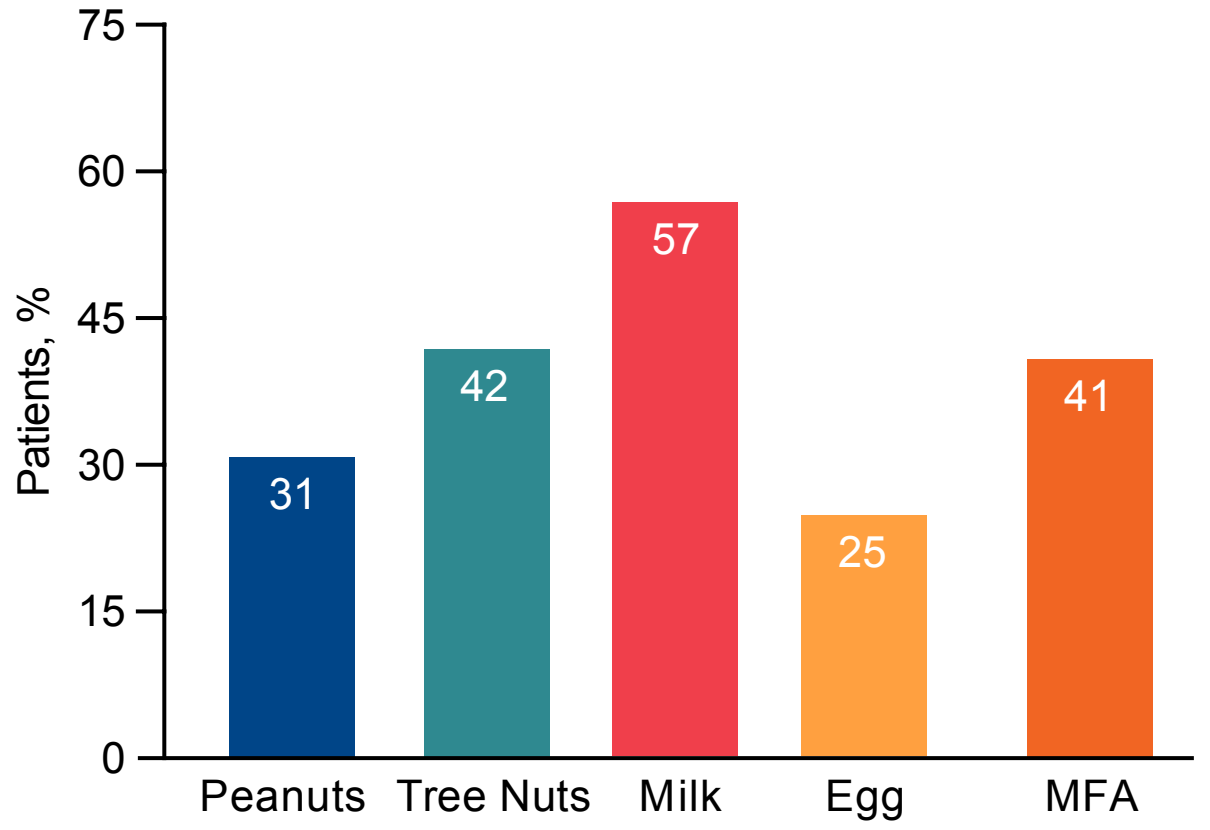
Patients, %
0
15
30
45
60
75
Peanuts
Tree Nuts
Milk
Egg
MFA
31
42
57
25
41

MFA, multiple food allergies (peanuts plus ≥2 to tree nuts, milk, or egg).

Figure 3. Percent of Patients Reporting Specific Mental Health Concerns

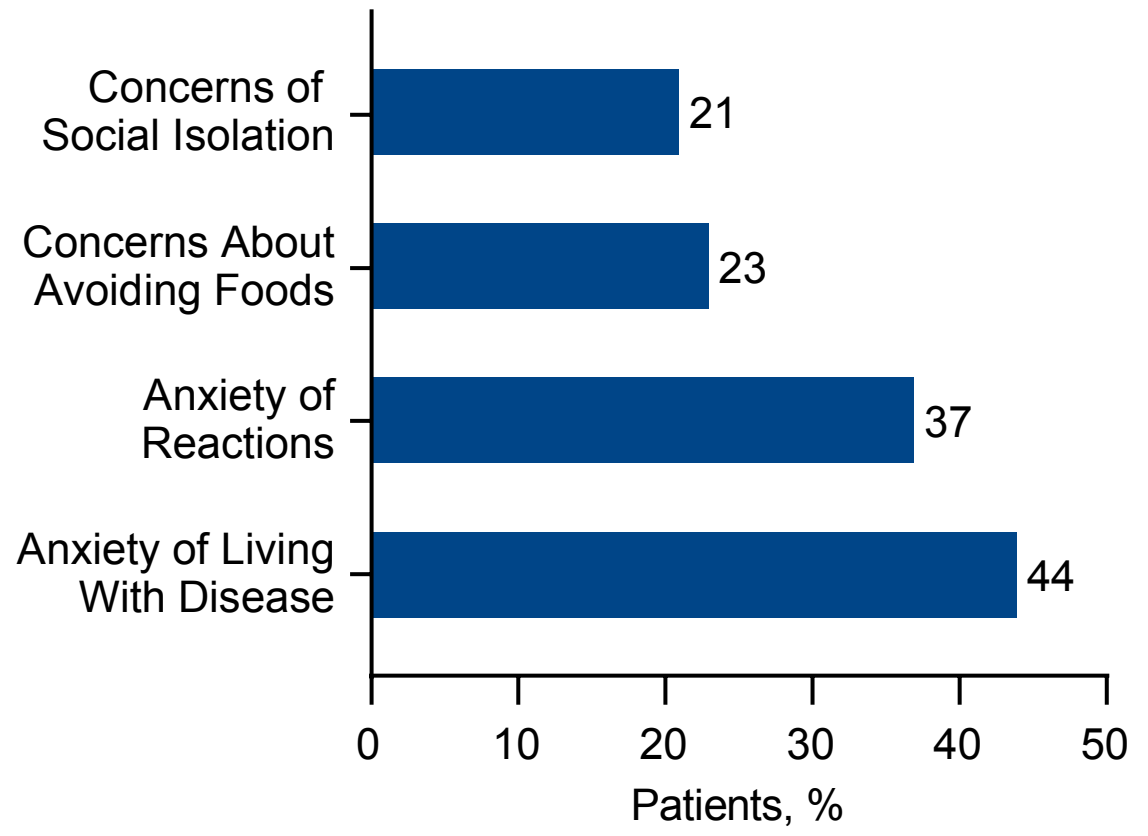
Concerns of Social Isolation
Concerns About Avoiding Foods
Anxiety of Reactions
Anxiety of Living With Disease
21
23
37
44
0
10
20
30
40
50
Patients, %

Food Allergy Mental Health Survey

- Across all food allergens (n=342), only 24% of patients who completed the mental health survey reported a formal mental health diagnosis, although 53% reported specific mental health concerns.
- The most common mental health concerns were anxiety of living with the disease (44%), anxiety of reactions (37%), avoiding foods (23%), and social isolation (21%) (**Figure 3**).

Limitations

- Given that the FARE Patient Registry is voluntary, the data may be reflective of the type of patient responding to a registry request and may not be representative of all patients with food allergies.
- Data in FARE are self-reported or reported by a parent or caretaker on behalf of a child and, therefore, could be subject to recall bias.
- Defining multiple food allergies as peanuts plus ≥2 to tree nuts, milk, or egg may not capture all patients with multiple food allergies.

Conclusions

- Allergic comorbidities are frequently experienced by patients with food allergy, and their relative frequency varies based on food allergen.
- Many people living with food allergy who have mental health concerns lack a formal mental health diagnosis.
- Therefore, health care providers who treat patients with food allergy should provide attention to comorbidities and mental health concerns, which may have a positive impact on the burden of food allergy.

References
1. Gupta RS, et al. *JAMA Netw Open*. 2019;2:e185630.
2. Burks AW, et al. *J Allergy Clin Immunol*. 2012;129:906–20.
3. Warren CM, et al. *Curr Allergy Asthma Rep*. 2020;20:6.
4. FARE Patient Registry. FARE website. Accessed May 5, 2022. https://www.foodallergypatientregistry.org/.

Disclosures **TBC:** consultant and speaker bureau member for Genentech, Inc.; consultant for Novartis Pharmaceuticals Corporation; chief medical advisor for FARE. **CW:** institutional research funding from FARE, the National Institutes of Health, and the Sunshine Charitable Foundation. **YR, RS, RW, AI, SG:** employees of Genentech, Inc.; stockholders in Roche. **RG:** research grant support from Allergy and Asthma Network, FARE, Genentech, Inc., Melchiorre Family Foundation, National Confectioners Association, National Institutes of Health, Stanford Sean N. Parker Center for Allergy Research, Sunshine Charitable Foundation, Thermo Fisher Scientific, UnitedHealth Group, Walder Foundation; medical consultant/advisor for FARE, Genentech, Inc., and Novartis; ownership interest in YoBee Care, Inc.

Acknowledgments
- The authors would like to thank Arpamas Seetasith, Genentech, Inc., for her contributions to the preparation of the poster.
- This study was funded by Genentech, Inc., a member of the Roche Group, and Novartis Pharma AG. Medical writing assistance was provided by Nikolas C. Vann, PhD, of Envision Pharma Group, and funded by Genentech, Inc., a member of the Roche Group.

Scan code for ePoster/PDF or visit https://bit.ly/3zDAxoy

Presented at the European Academy of Allergy & Clinical Immunology (EAACI) Hybrid Congress 2022, July 1–3, 2022, Prague, Czech Republic

Copyright © 2022. All rights reserved. Contact Sachin Gupta for permission.