A Longitudinal Disease Progression Model in Alzheimer's Disease (AD) to Assess the Clinical Dementia Rating Scale – Sum of Boxes (CDR-SB) in Tauriel

Samira Jamalian[1], Michael Dolton[2], Pascal Chanu[3], Vidya Ramakrishnan[1], Kristin Wildsmith[1], Bali Toth[1], Paul Manser[1], Edmond Teng[1], Jin Jin[1], Angelica Quartino[1], Joy C. Hsu[1]

[1]Genentech, Inc., South San Francisco, CA, USA; [2]Roche Products Australia Pty Ltd, Sydney, Australia; [3]Genentech/Roche, Lyon, France.

Introduction

Modeling longitudinal progression in AD

- Understanding the natural progression of Alzheimer's disease (AD) is essential to assessing the efficacy of treatment in AD[1]
- AD progression models have been developed with longitudinal data describing the trajectory of cognitive scores using mixed-effect modeling approaches[1,2]

Tauriel clinical trial

- Semorinemab is a humanized anti-tau antibody targeting all known isoforms of full-length tau
- The recent Tauriel (NCT03289143) Phase 2, randomized, double-blind, placebo-controlled trial investigated the efficacy and safety of semorinemab in people with prodromal-to-mild AD[3,4]
 - — The study was designed with the help of an AD model and clinical trial support[5,6]
- While semorinemab was well-tolerated, it did not slow clinical progression on the primary outcome, change from baseline at Week 73 in Clinical Dementia Rating Scale – Sum of Boxes (CDR-SB)[4]

Objective

- The objective of this modeling effort was to predict the natural progression (in absence of treatment) of CDR-SB score in the placebo and semorinemab treatment arms in Tauriel, to support analysis and decision making following the study readout

Methods

Model building dataset

- An equation describing the change in CDR-SB over time was fitted to data from the placebo arms of four interventional trials[7-10] and the Alzheimer's Disease Neuroimaging Initiative (ADNI) study[2] (N_{total} = 1044) (**Figure 1**)

Figure 1. Studies for model building and validation

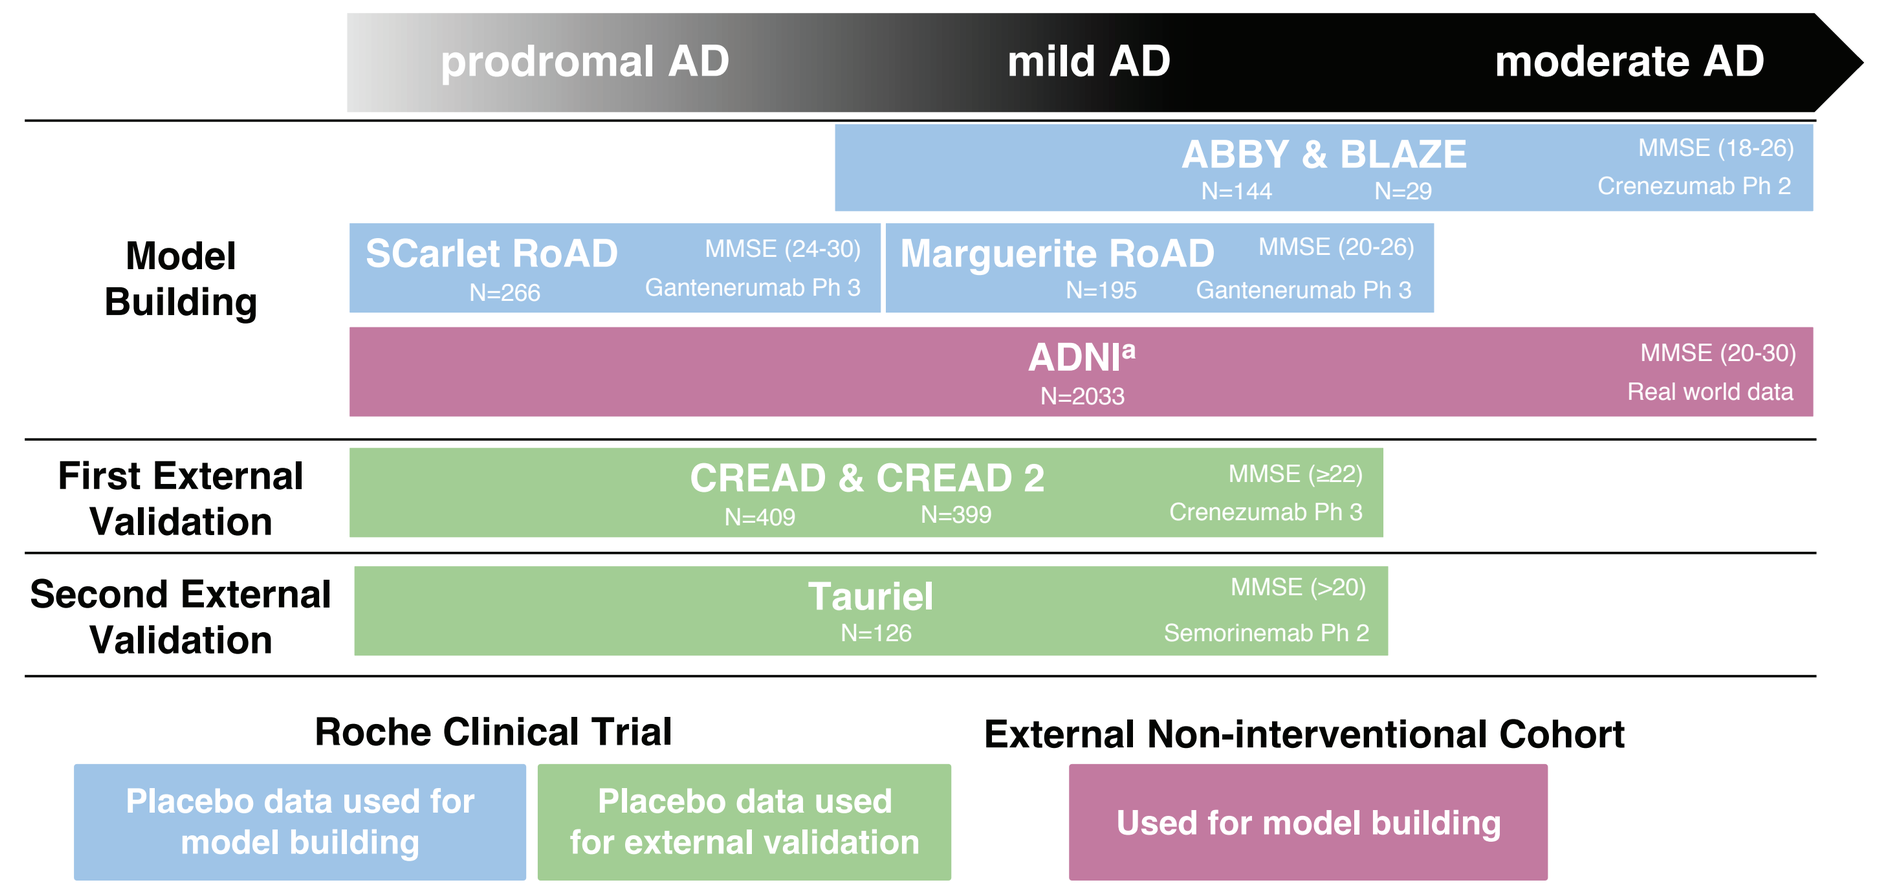
prodromal AD
mild AD
moderate AD
Model Building
ABBY & BLAZE
N=144 N=29
MMSE (18-26)
Crenezumab Ph 2
SCarlet RoAD
N=266
MMSE (24-30)
Gantenerumab Ph 3
Marguerite RoAD
N=195
MMSE (20-26)
Gantenerumab Ph 3
ADNIa
N=2033
MMSE (20-30)
Real world data
First External Validation
CREAD & CREAD 2
N=409 N=399
MMSE (≥22)
Crenezumab Ph 3
Second External Validation
Tauriel
N=126
MMSE (>20)
Semorinemab Ph 2
Roche Clinical Trial
Placebo data used for model building
Placebo data used for external validation
External Non-interventional Cohort
Used for model building

Trial lengths ranged from 18 months–2 years.
[a]Late MCI and AD patients who were amyloid positive were selected from ADNI

AD, Alzheimer's disease; ADNI, Alzheimer's Disease Neuroimaging Initiative; MCI, mild cognitive impairment; MMSE, mini mental state examination; Ph, phase.

Model development

- Change in CDR-SB score was described via a differential equation[11,12] to estimate disease onset time (DOT) for each patient (**Figure 2A**)
- Individual change in CDR-SB was described by a global disease progression rate (RATE) and an exponential growth rate (ALPHA, α) (**Figure 2B**)
- Covariate analysis was conducted to identify factors that help explain between subject variability
- An additive residual error model was implemented

Figure 2. Example curve of score progression (A) and equation to estimate DOT, rate, and α (B)

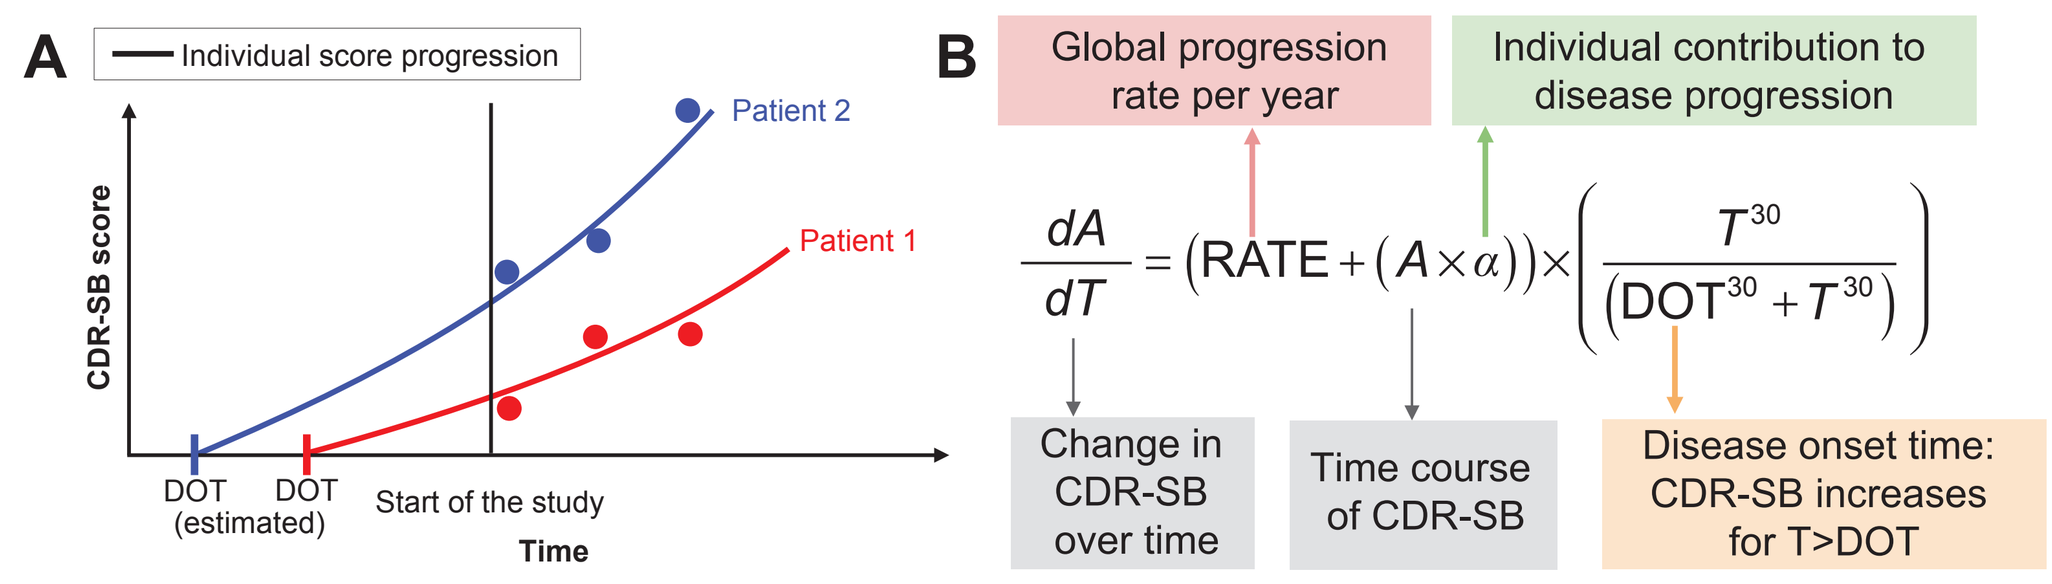
A
Individual score progression
Patient 2
Patient 1
CDR-SB score
DOT (estimated)
DOT
Start of the study
Time
B
Global progression rate per year
Individual contribution to disease progression
$\frac{dA}{dT} = (\mathrm{RATE} + (A \times \alpha)) \times \left(\frac{T^{30}}{(\mathrm{DOT}^{30} + T^{30})} \right)$
Change in CDR-SB over time
Time course of CDR-SB
Disease onset time: CDR-SB increases for T>DOT

Model developed in NONMEM (non-linear mixed-effect modeling).

α, exponential growth rate; CDR-SB, Clinical Dementia Rating Scale – Sum of Boxes; DOT, disease onset time; RATE, population disease progression rate; T, time.

Model evaluation

- The final model with covariates was selected by careful examination of goodness of fit plots together with minimization of the objective function

Results:

Model development

- The progression of CDR-SB score for the entire population was captured (**Figure 3**) by including the following covariates:
 - — **Baseline CDR-SB** on DOT and RATE
 - — **Baseline MMSE** score on ALPHA
- Interindividual variability was implemented on DOT and ALPHA
- All parameters were well estimated
 - — DOT was estimated as 3.3 years before entering the trial (or start of study for ADNI) (relative standard error [RSE] 1.5%)
 - — Global progression rate (RATE) was estimated at 0.305 (/y) [5%]

Figure 3. Disease trajectory and individual predictions for 100 randomly selected patients from the model building dataset

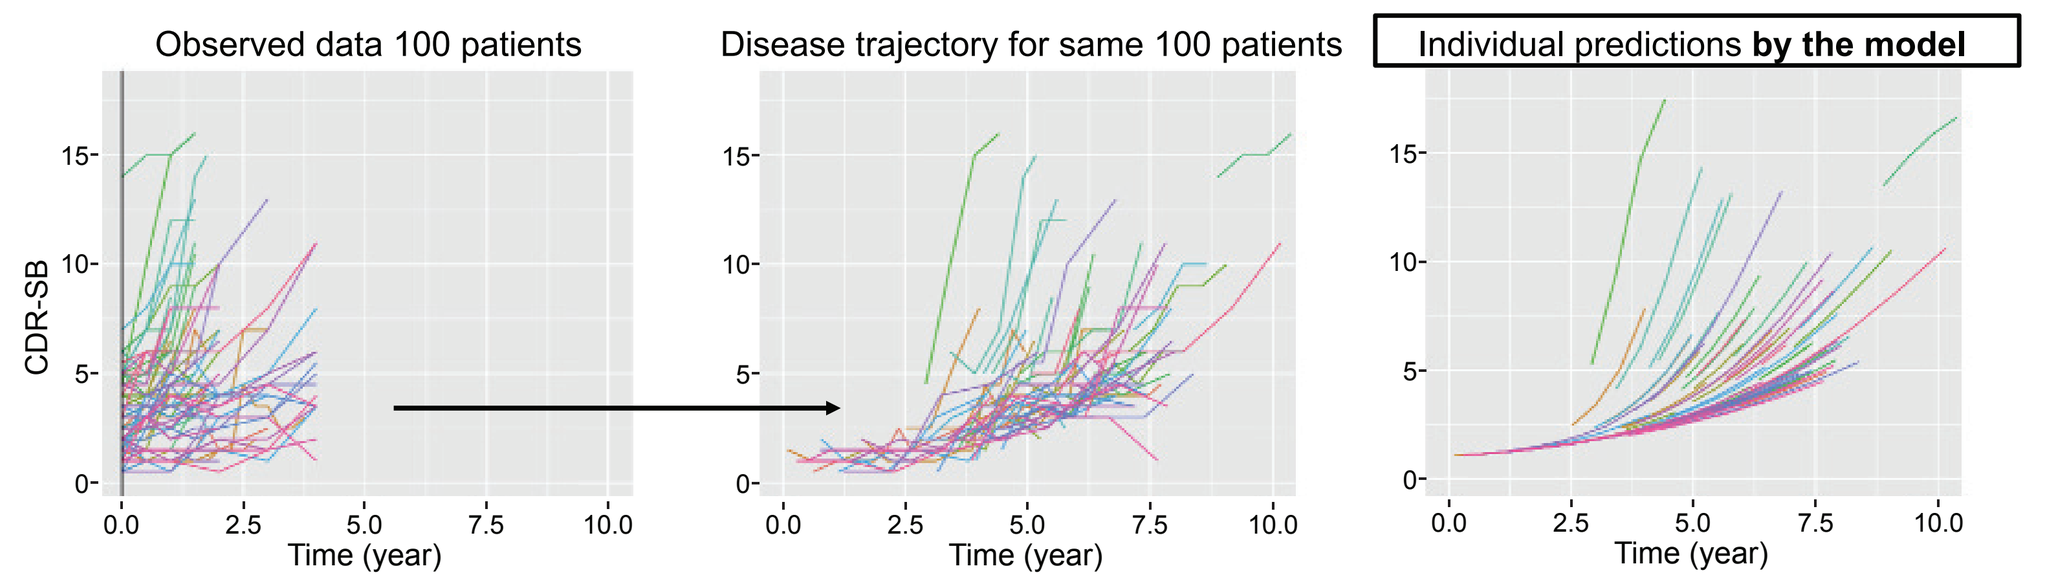
Observed data 100 patients
Disease trajectory for same 100 patients
Individual predictions by the model
CDR-SB
Time (year)

CDR-SB, Clinical Dementia Rating Scale – Sum of Boxes

Model evaluation

- Internal validation with a visual predictive check (VPC) confirmed that the model performs adequately in capturing the observed data from each trial used in model building (**Figure 4**)

Figure 4. Example of VPC internal validation of the model

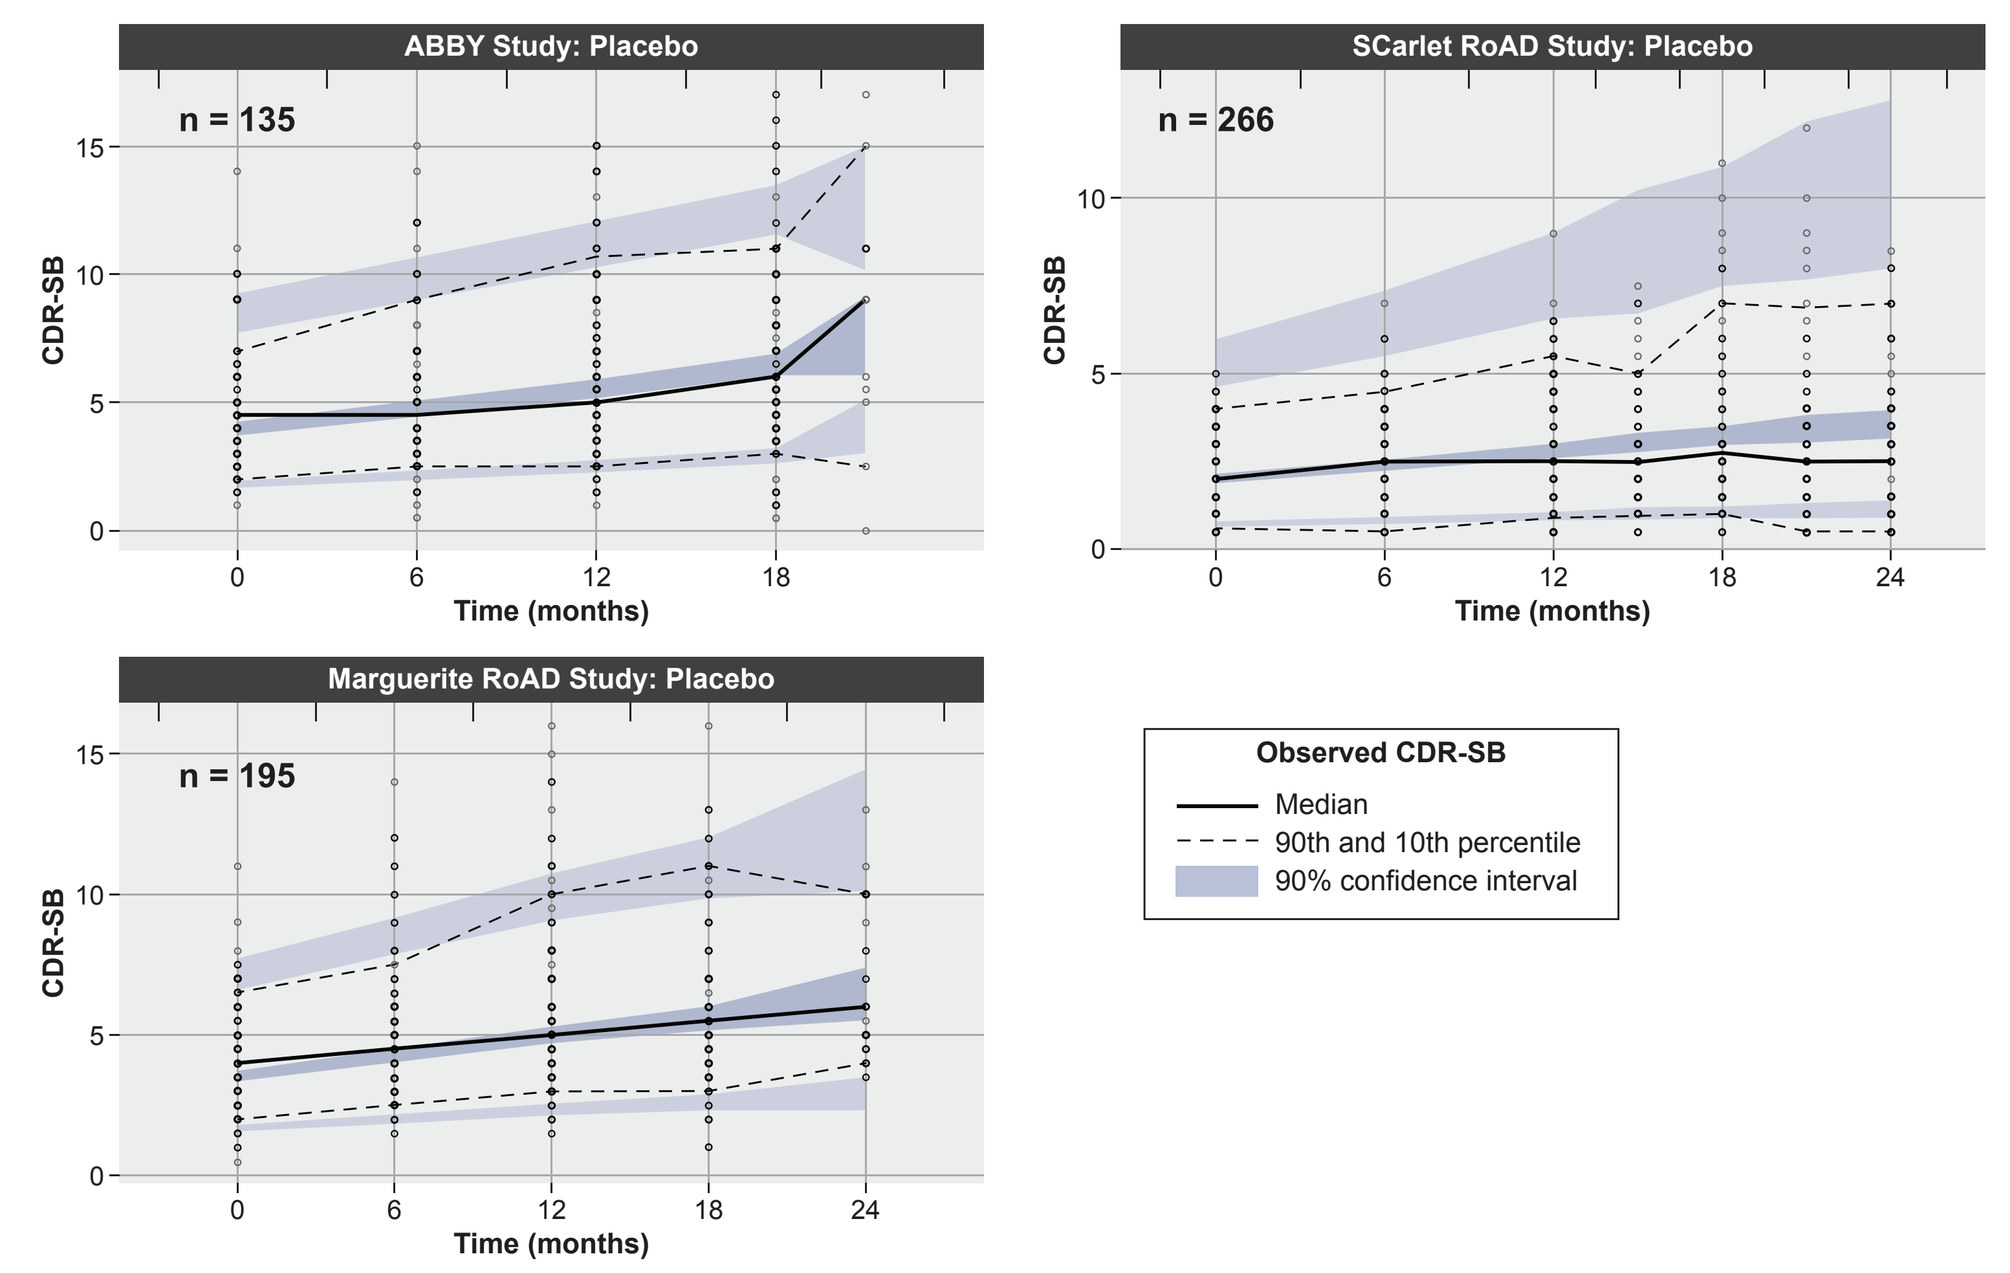
ABBY Study: Placebo
n = 135
SCarlet RoAD Study: Placebo
n = 266
Marguerite RoAD Study: Placebo
n = 195
CDR-SB
Time (months)
Observed CDR-SB
Median
90th and 10th percentile
90% confidence interval

1000 simulations each.

CDR-SB, Clinical Dementia Rating Scale – Sum of Boxes; VPC, visual predictive check.

- External validation via VPC of the CREAD and CREAD2 studies[13] confirmed that the model performs adequately in predicting the observed data from trials not used in model building (**Figure 5**)

Figure 5. First VPC external validation of the model in CREAD and CREAD2

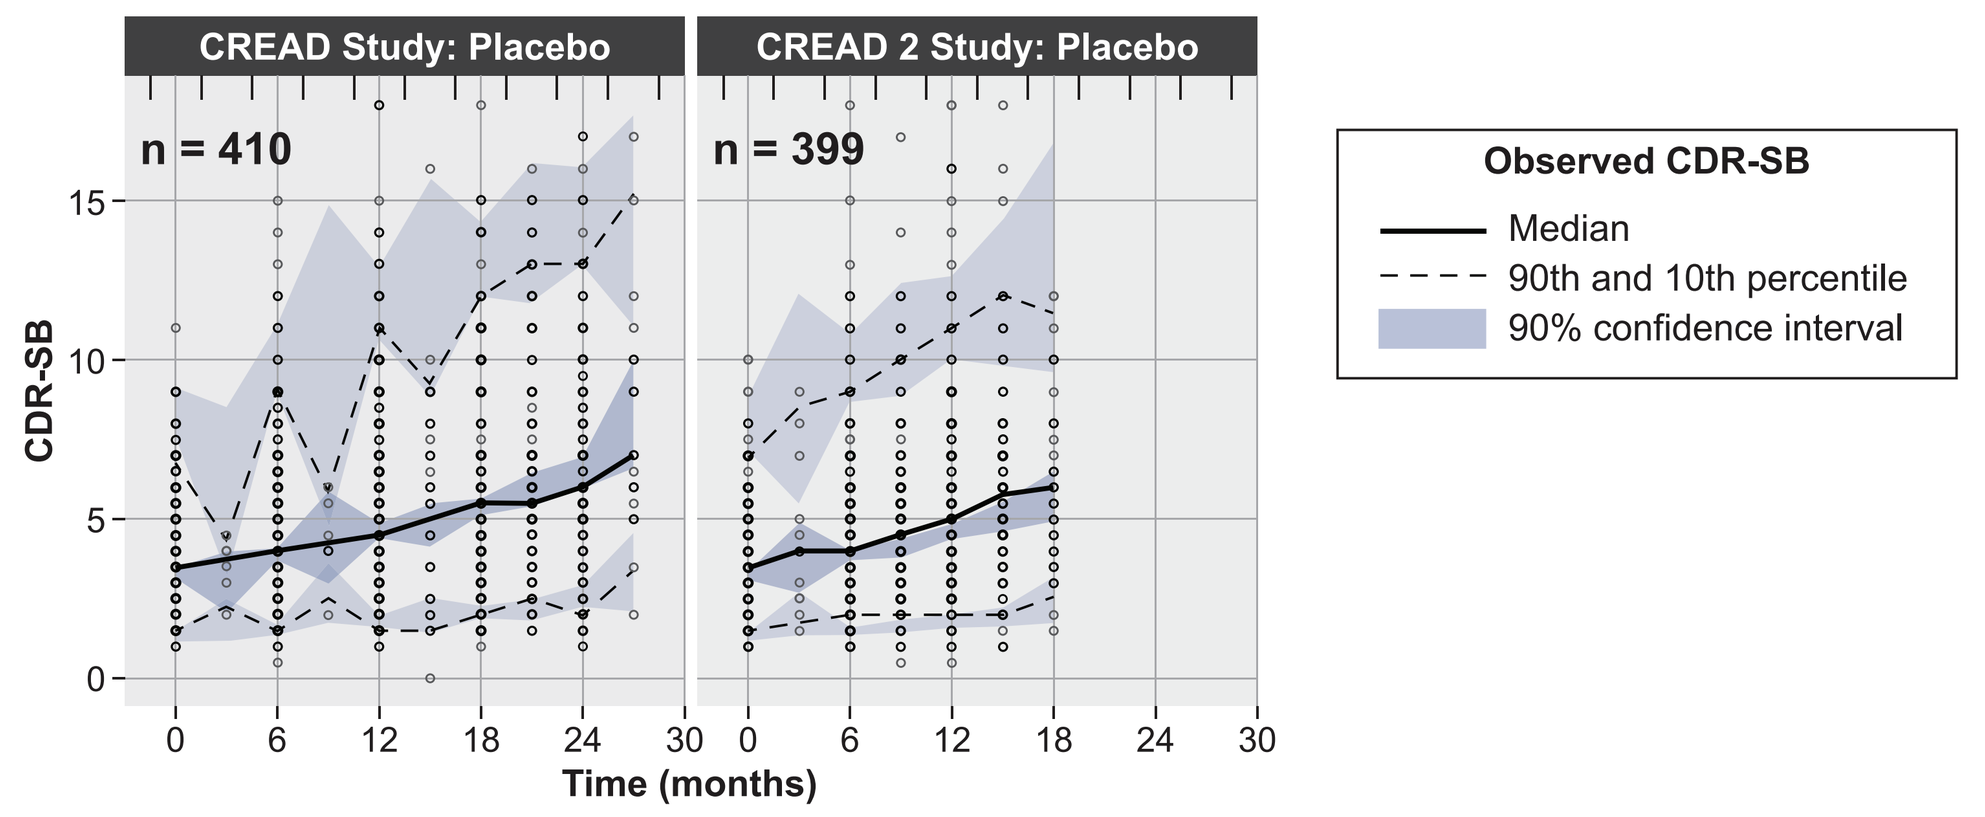
CREAD Study: Placebo
n = 410
CREAD 2 Study: Placebo
n = 399
CDR-SB
Time (months)
Observed CDR-SB
Median
90th and 10th percentile
90% confidence interval

Base, baseline; CDR-SB, Clinical Dementia Rating Scale – Sum of Boxes; VPC, visual predictive check; W, week.

Model application in Tauriel

- A second external validation of the model was conducted using data from the placebo arm of the Tauriel study (**Figure 6**)

Figure 6. Second VPC external validation of the model in Tauriel

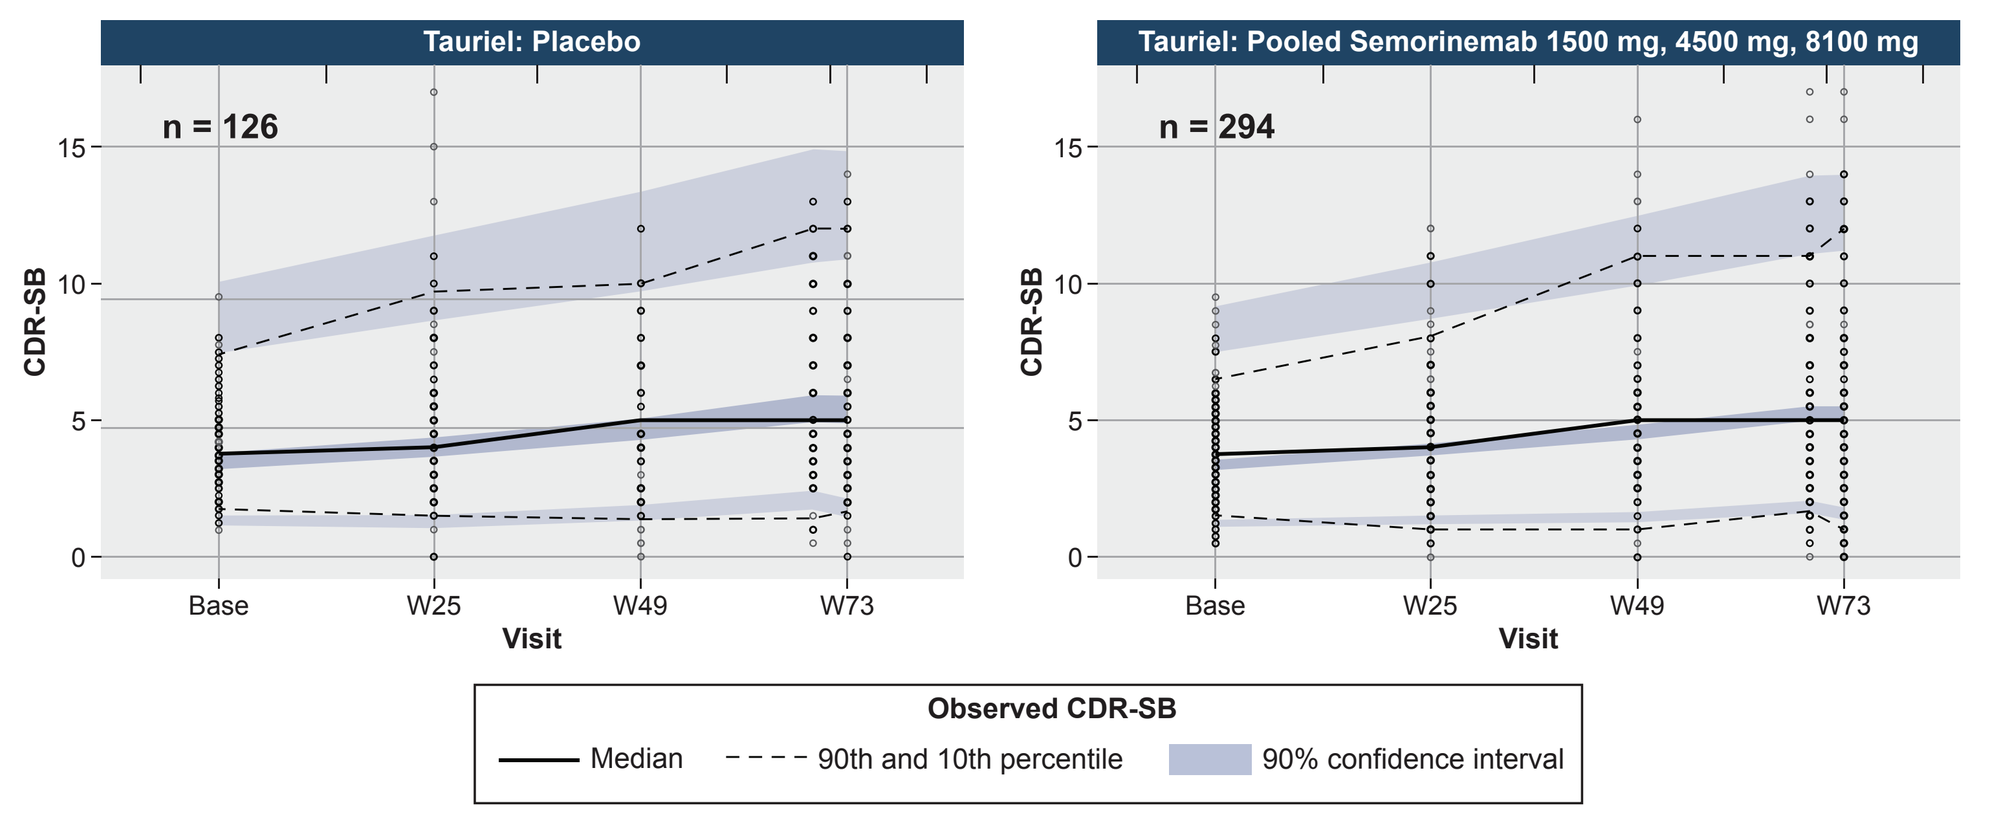
Tauriel: Placebo
n = 126
Tauriel: Pooled Semorinemab 1500 mg, 4500 mg, 8100 mg
n = 294
CDR-SB
Visit
Base
W25
W49
W73
Observed CDR-SB
Median
90th and 10th percentile
90% confidence interval

Base, baseline; CDR-SB, Clinical Dementia Rating Scale – Sum of Boxes; VPC, visual predictive check; W, week.

Model prediction for Tauriel

- The validated model with covariates predicted natural progression of CDR-SB score (without treatment effect) in placebo and semorinemab treatment arms in Tauriel (**Table 1, Figure 7**)

Table 1. Mean change from baseline at Week 73 in CDR-SB in Tauriel

Group	Model Prediction, mean CFB (95% PI)	Observed, mean CFB (95% CI)
Placebo	2.21 (1.8–2.7)	1.97 (1.55–2.38)
Pooled semorinemab	2.17 (1.9–2.5)	2.10 (1.79–2.43)

Mean computed from 1000 simulations

CDR-SB, Clinical Dementia Rating Scale – Sum of Boxes; CFB, change from baseline.

Figure 7. Model and observed CDR-SB progression of patients in Tauriel

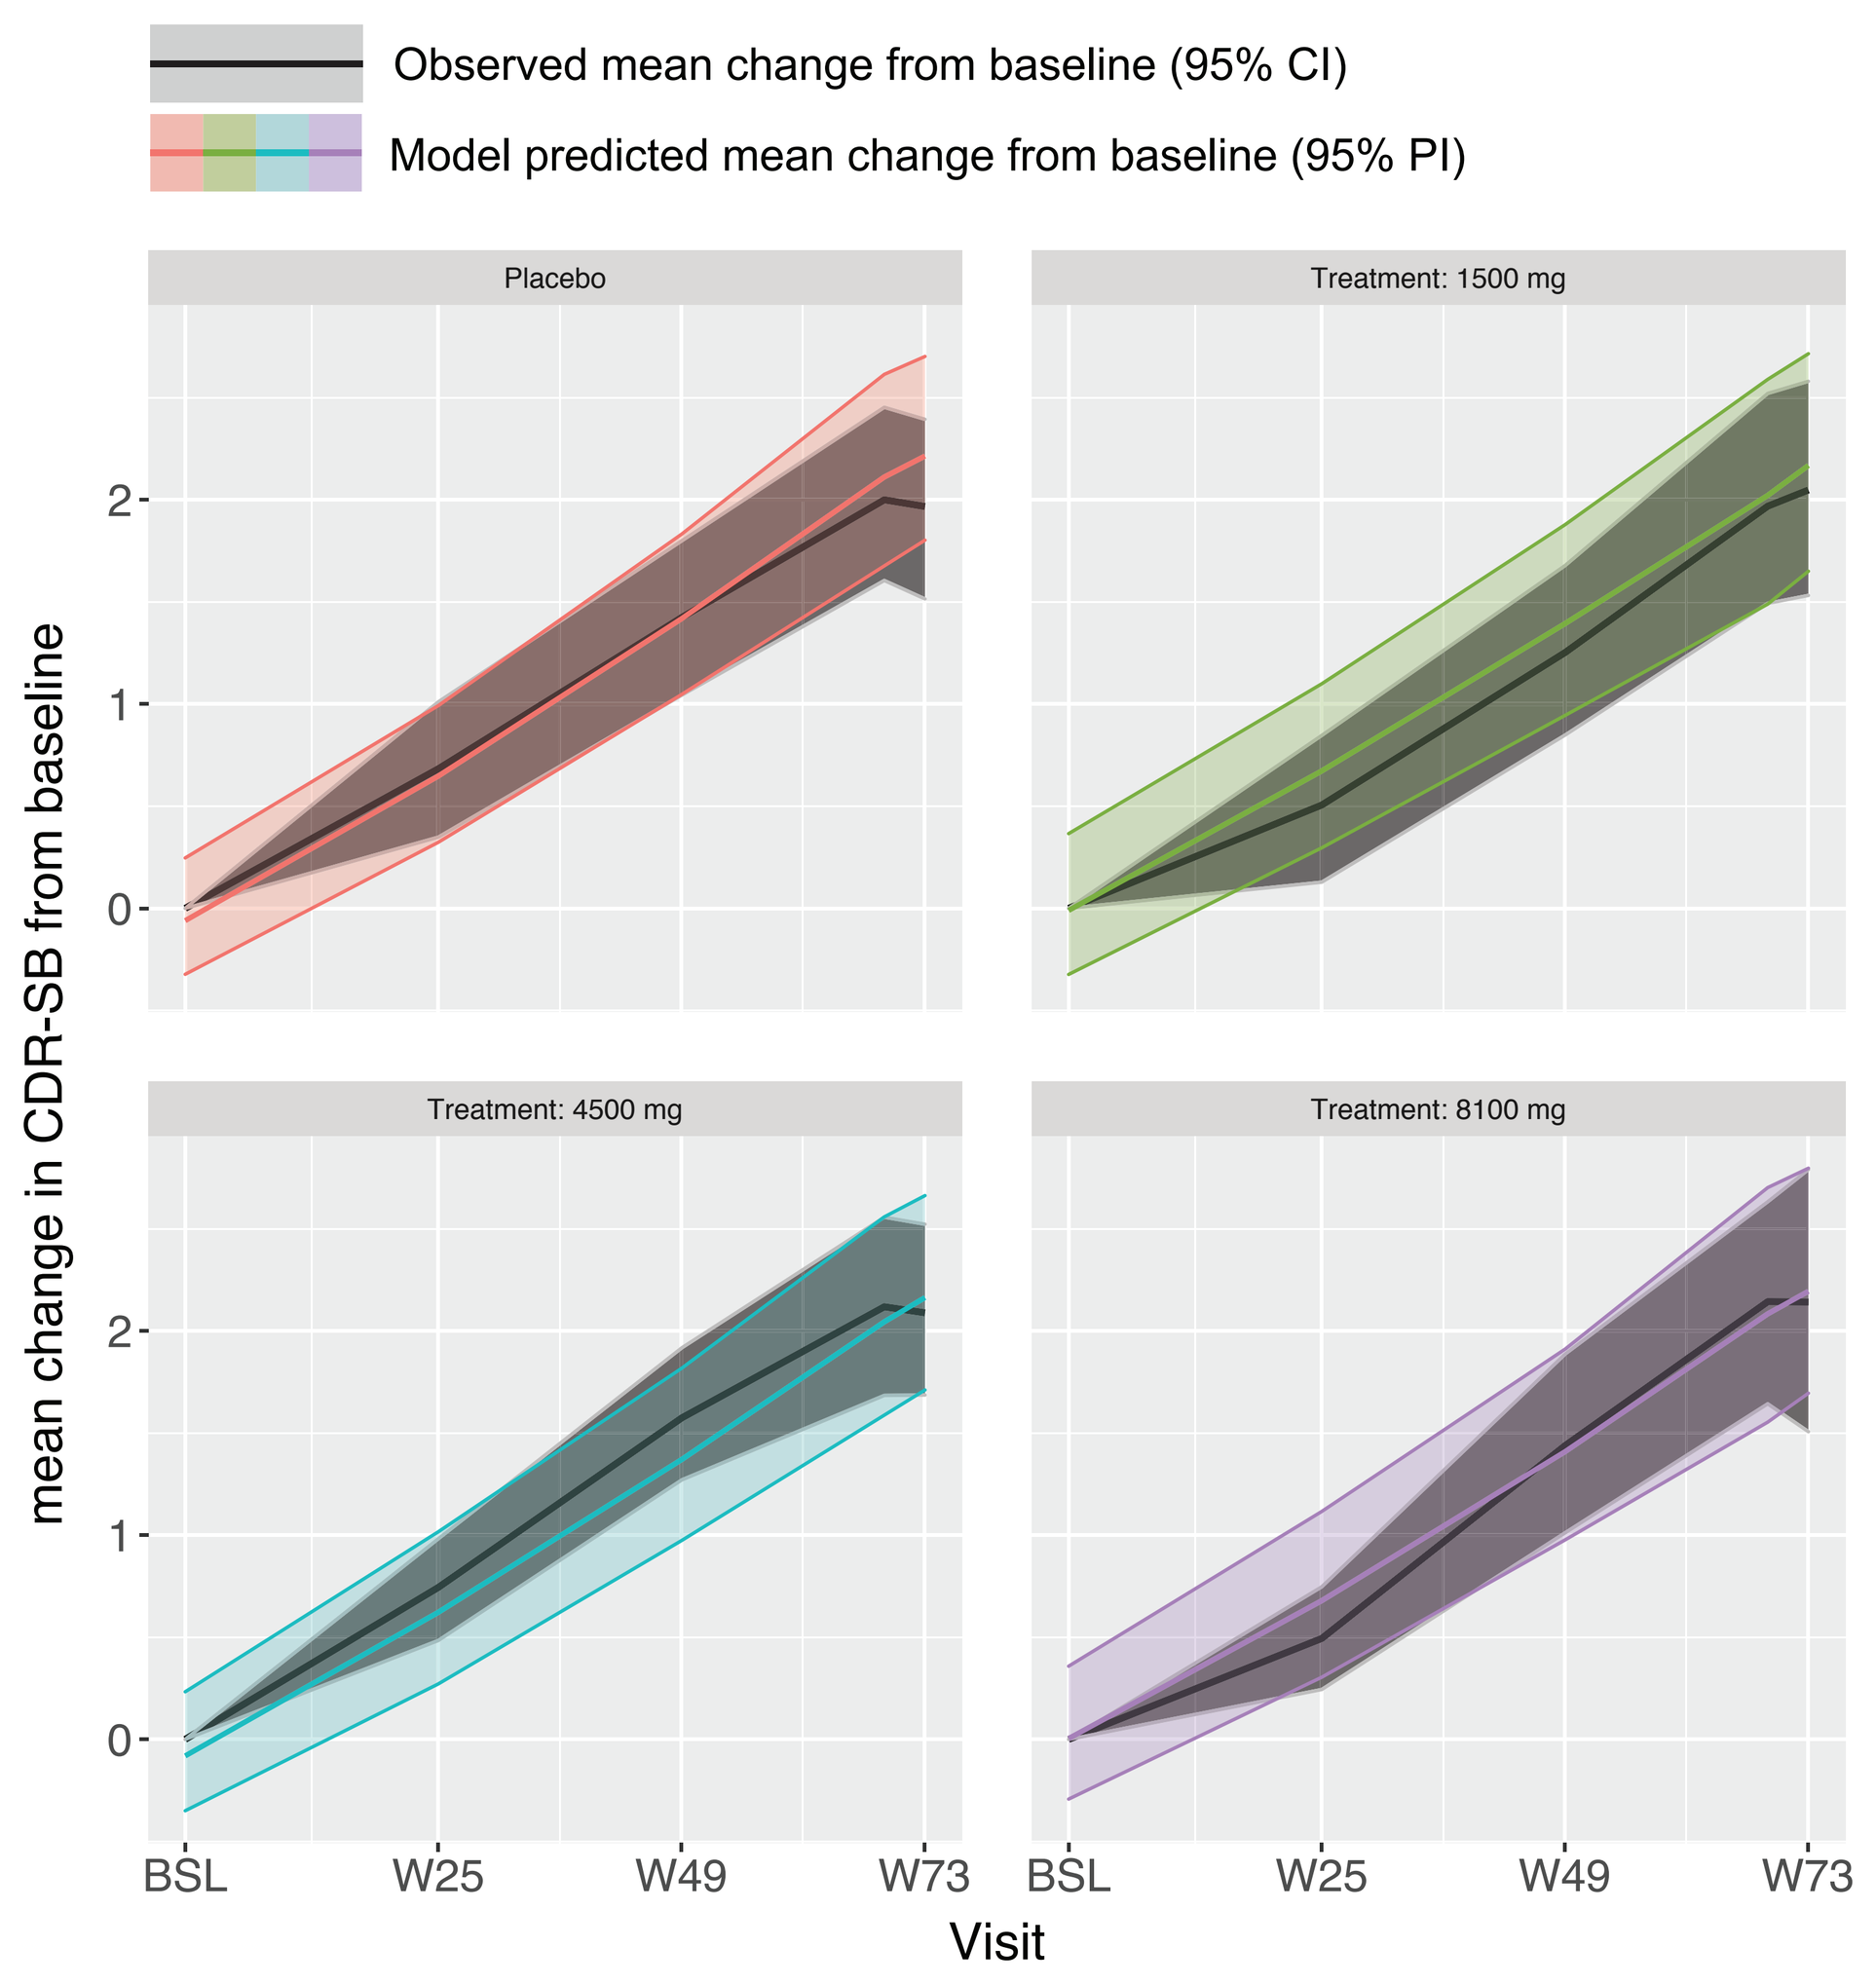
Observed mean change from baseline (95% CI)
Model predicted mean change from baseline (95% PI)
Placebo
Treatment: 1500 mg
Treatment: 4500 mg
Treatment: 8100 mg
mean change in CDR-SB from baseline
BSL
W25
W49
W73
Visit

BSL, baseline; CDR-SB, Clinical Dementia Rating Scale – Sum of Boxes; CI, confidence interval; PI, prediction interval; W, week.

- Individual parameter predictions for Tauriel were similar between treatment arms (**Table 2**)

Table 2. Distribution of individual parameter values from Tauriel

Group	DOT (range), years	RATE (range), per year	α (range)
Placebo	4.34 (6.52, 2.03)	0.33 (0.24, 0.43)	0.06 (0.03, 0.12)
Semorinemab 1500 mg	4.37 (6.12, 0.72)	0.33 (0.20, 0.40)	0.06 (0.03, 0.12)
Semorinemab 4500 mg	4.26 (5.89, 0.42)	0.33 (0.20, 0.41)	0.06 (0.02, 0.14)
Semorinemab 8100 mg	4.34 (6.13, 1.36)	0.33 (0.22, 0.43)	0.05 (0.02, 0.12)

α, exponential growth rate; CDR-SB, Clinical Dementia Rating Scale – Sum of Boxes; DOT, disease onset time; RATE, population disease progression rate.

Conclusions

- The developed model reliably predicts longitudinal natural progression of CDR-SB based on baseline CDR-SB and baseline MMSE scores
- In Tauriel, observed progression in mean change from baseline in the placebo group was in agreement with the AD model projected progression
- Model predictions confirmed that patients in the Tauriel semorinemab treatment arms progressed as expected based on their natural progression, without a treatment effect in the model
- Alignment between the model predicted and observed progression in the semorinemab treatment groups confirmed that there was no significant semorinemab treatment effect in prodromal-to-mild AD

References

1. Ito K, et al. Alzheimers Dementia. 2011;7(2):151-60.
2. Alzheimer's Disease Neuroimaging Initiative (ADNI) database (adni.loni.usc.edu).
3. ClinicalTrials.gov. https://clinicaltrials.gov/show/NCT03289143. Accessed May 20, 2020.
4. Teng E, et al. Oral presentation OC32 at the 13th Clinical Trials in Alzheimer's Disease conference (CTAD). November 4-7, 2020. Digital event.
5. Quartino AL, et al. Oral presentation at Population Approach Group Europe (PAGE). June 10-13, 2014. Alicante, Spain. [abstract 3187].
6. Rogers JA, et al. Oral presentation at the 40th Midwest Biopharmaceutical Statistics Workshop. May 22-24, 2017. Muncie, IN, USA.
7. Ostrowitzki, S et al. Alz Res Therapy. 2017;9:95.
8. Klein, G et al. Alzheimers Res Ther. 2019;11(1):101.
9. Salloway, S et al. Alzheimers Res Ther. 2018 Sep 19;10(1):96.
10. Cummings, JL et al. Neurology. 2018;90(21):e1889-e1897.
11. Delor, I et al. CPT: Pharmacometrics Syst Pharmacol. 2013;2(10):e78.
12. Jamalian S, et al. Oral Presentation LB04 at the 13th Clinical Trials in Alzheimer's Disease (CTAD). November 4-7, 2020. Digital event.
13. Blaettler, T et al. Alzheimers Dement. 12:7S(12):P609.

Acknowledgments:

We would like to thank Christina Rabe, Tobias Bittner, and the Roche/Genentech LeAD (Leveraging exploratory Analysis and Data in AD) team for preparation of the dataset and Amita Joshi, Christina Rabe, and the semorinemab team for providing valuable feedback

Semorinemab is being developed by Genentech and was identified in collaboration with AC Immune SA, Lausanne, Switzerland.

This study was funded by Genentech Inc., South San Francisco, CA. ADNI is funded by the National Institute on Aging, the National Institute of Biomedical Imaging and Bioengineering, and through generous contributions from other external parties.

Disclosures:

Editorial support for the development of this poster was provided by Sarah Engelberth, PhD, of Medical Expressions, funded by F. Hoffmann-La Roche

S Jamalian, P Chanu, V Ramakrishnan, K Wildsmith, B Toth, P Manser, E Teng, J Jin, A Quartino, and JC Hsu are full time employees of Genentech, Inc and may own company stock/stock options

M Dolton is a full time employees of Roche Products Australia Pty Ltd and may own company stock/stock options

Presented at the American Academy of Neurology (AAN) 2021 Annual Meeting, April 17-22, 2021 (Virtual)