Assessing health system capacity for delivery of a disease-modifying therapy for Alzheimer's disease: a multi-country analysis

Nathalie Budd[1], Valerie Crowell, PhD[2], Ilke Mirik Danaci[2], Haakon B. Nygaard, MD, PhD[3]

[1]F. Hoffmann-La Roche Ltd, Basel, Canada [2]F. Hoffmann-La Roche Ltd, Basel, Switzerland
[3]Division of Neurology and Djavad Mowafaghian Centre for Brain Health, University of British Columbia, Vancouver, Canada

1

What does this mean for the AD community?

Given current health system capacity, patients may face challenges to access AD disease modifying therapies (DMTs) in the near-term. These challenges, which occur at the confirmatory diagnosis step and beyond, could lead to delays in treatment for early AD, during which time many patients may progress to more severe disease. A better understanding of the largest bottlenecks can help to plan and prioritize strategies to ensure that eligible patients can benefit from DMTs as soon as possible.

Conclusions

Given current health system resources in France, Canada,Germany, Italy, Spain, the UK and the US, only an estimated 1-5% of patient demand for AD DMTs could be met. Neurologists, MRIs, and PET scans represent the largest bottlenecks. Improved detection and diagnostic methods, alternative formulations, and health system strengthening are critical to help alleviate bottlenecks.

Background

- Recent estimates suggest 100 million people globally are afflicted with Alzheimer's disease (AD), many in the early stages[1]. Disease modifying therapies (DMTs) for early AD have now received regulatory approval, however, health systems face enormous challenges to detect, diagnose and treat all patients who could benefit.
- With the availability of DMTs, patient demand is expected to increase along the AD care pathway. Consequently, substantial additional resources, including health care provider time, infrastructure and equipment, will be required. Insufficient capacity at any point along the pathway will result in bottlenecks that prevent or delay patients from moving to the next step of care.
- Understanding the nature and magnitude of health system bottlenecks is essential for health system planning, resource allocation and selection of optimal solutions to ensure broad and equitable patient access to AD DMTs.

Methodology

- A model was developed to estimate the difference between required and available resources across the AD care pathway in the near term (1-3 years) following DMT introduction. The pathway is comprised of four main steps: screening, diagnosis, eligibility testing (with positron emission tomography (PET) scanners or cerebrospinal fluid (CSF) Amyloid-β (Aβ) testing), and treatment/follow-up (Fig. 1).
- Resources considered were health care personnel (AD specialists, AD nurses, pharmacists, radiologists and technologists), magnetic resonance imaging (MRI) and PET scanners. Resources associated with chairs for intravenous (IV) infusions and CSF procedures were assumed to have unrestricted capacity.
- Patient demand and available resources were informed by a pragmatic review of literature on AD care-seeking, provider behavior, epidemiology, and health system variables in France, Canada, Germany, Italy, Spain, the United Kingdom (UK) and the United States (US).
- Assumptions about required resources were sourced from clinical trial protocols, literature and expert opinion.
- Annual total and satisfied demand at each patient journey step, additional resources required to meet total patient demand, and, in Canada, the US and the UK, the additional patients that could be served by a shift away from PET to CSF Aβ testing were estimated.
- Although they were not explicitly considered as a bottleneck, the additional time required of primary care providers (PCP) to detect possible cases of early AD was estimated.

Figure 1. AD capacity model flow

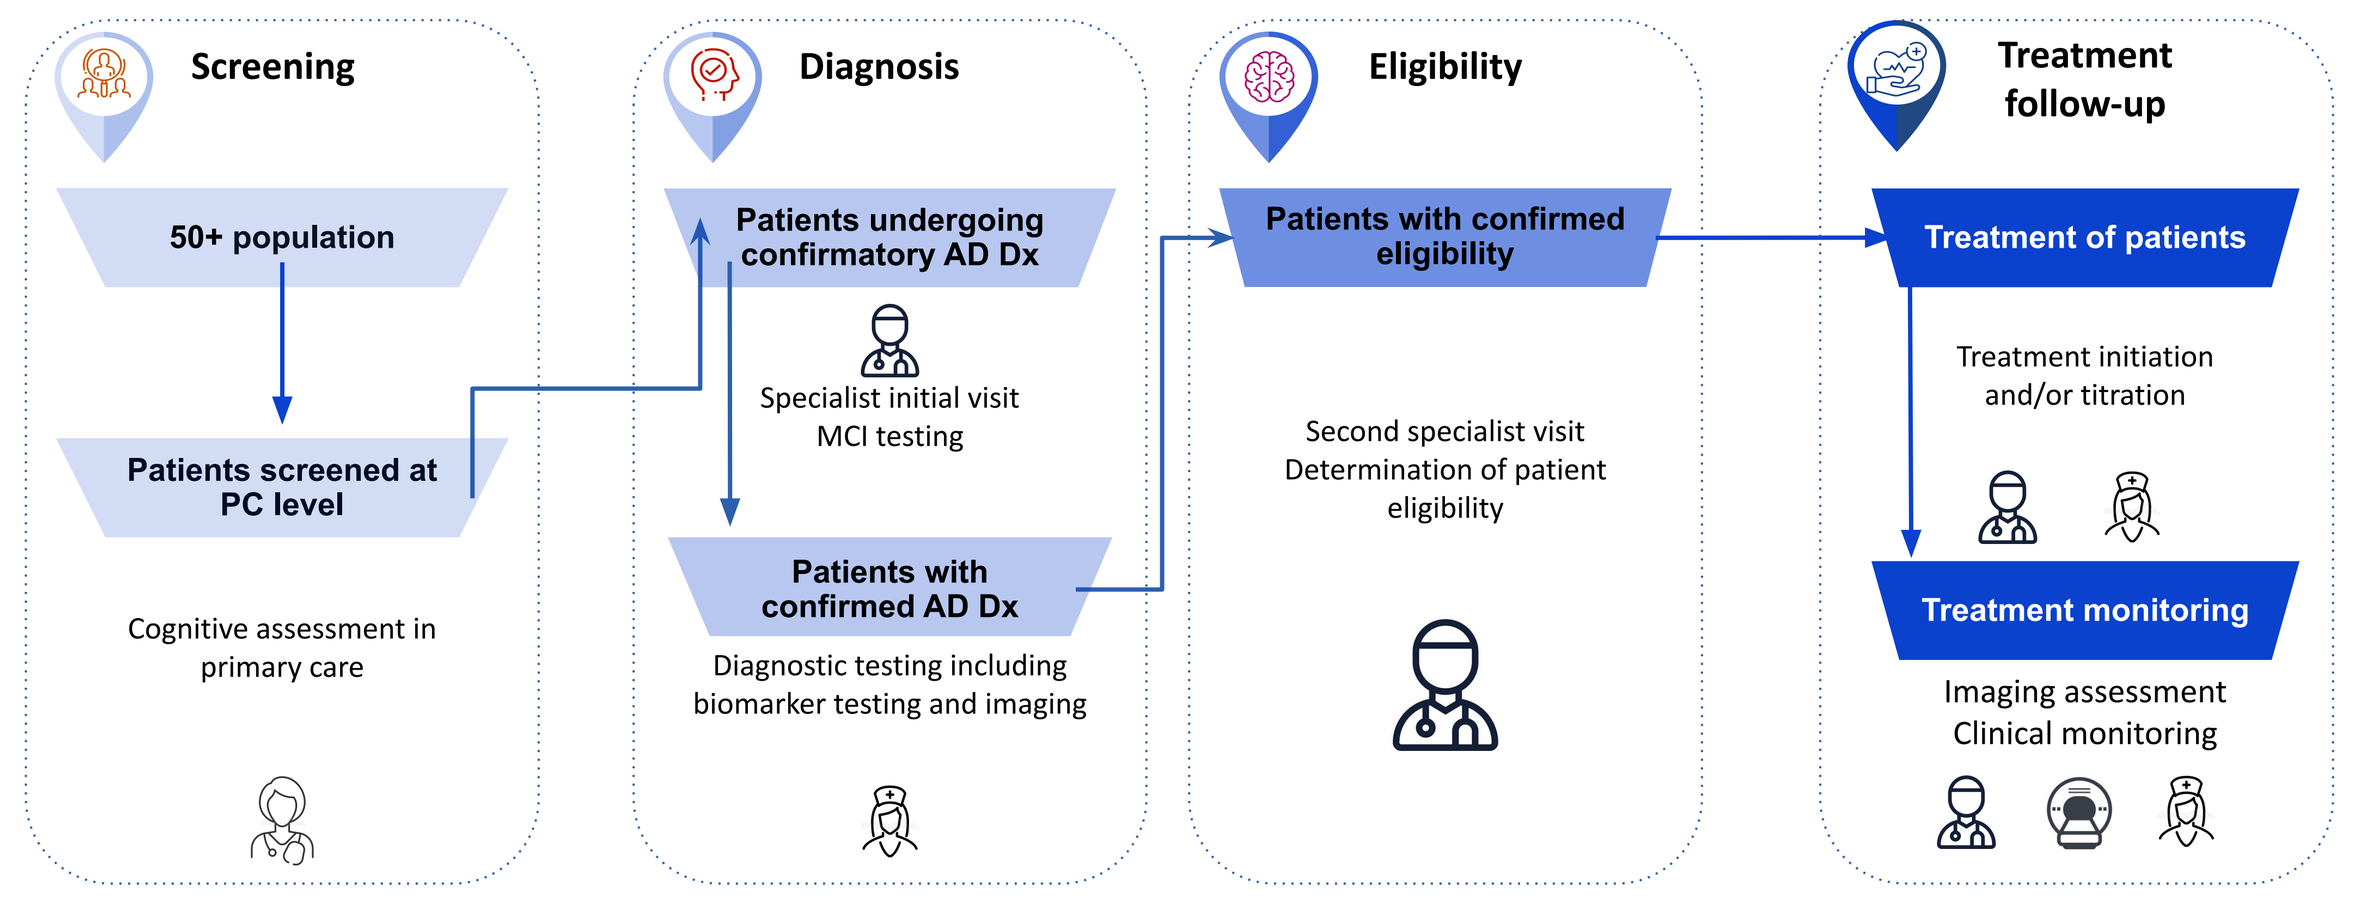
Screening
50+ population
Patients screened at PC level
Cognitive assessment in primary care
Diagnosis
Patients undergoing confirmatory AD Dx
Specialist initial visit MCI testing
Patients with confirmed AD Dx
Diagnostic testing including biomarker testing and imaging
Eligibility
Patients with confirmed eligibility
Second specialist visit Determination of patient eligibility
Treatment follow-up
Treatment of patients
Treatment initiation and/or titration
Treatment monitoring
Imaging assessment Clinical monitoring

The model calculated the capacity gap as the difference between the total required resources and the currently available resources, for each step of the patient journey (diagnosis, eligibility, and treatment /monitoring).
There were three essential elements of the model:

[Number of patients (Capacqity demand) x Resource required per patient] - Available health care capacity = Capacity Gap

I. Health care capacity demand (i.e. prevalence, diagnostic rate, Aβ positivity rate, treatment rate, market shares)
II. Health care resources required (i.e. assignment of resource allocations or patient slots to various personnel or infrastructure)
III. Available health care capacity (i.e. number of health system personnel and infrastructure resources available, estimate of time available)

Results

- Given current health system resources in these countries, only 1-5% of patient demand for DMTs could be met (Fig. 2).
- The largest bottlenecks are at the diagnosis and eligibility steps.
- Satisfied demand for diagnosis was highest in Germany, where AD specialists are more numerous, and lowest in France; however, in both countries only 2- 3% of demand for treatment could be met.
- In Canada, UK and the US, a greater reliance on PET scans vs CSF resulted in only 1-2% of demand for eligibility testing being satisfied.

y.

Figure 2. Satisfied demand for diagnosis, eligibility testing, and treatment as a percentage of total demand, in the first year of DMT availability

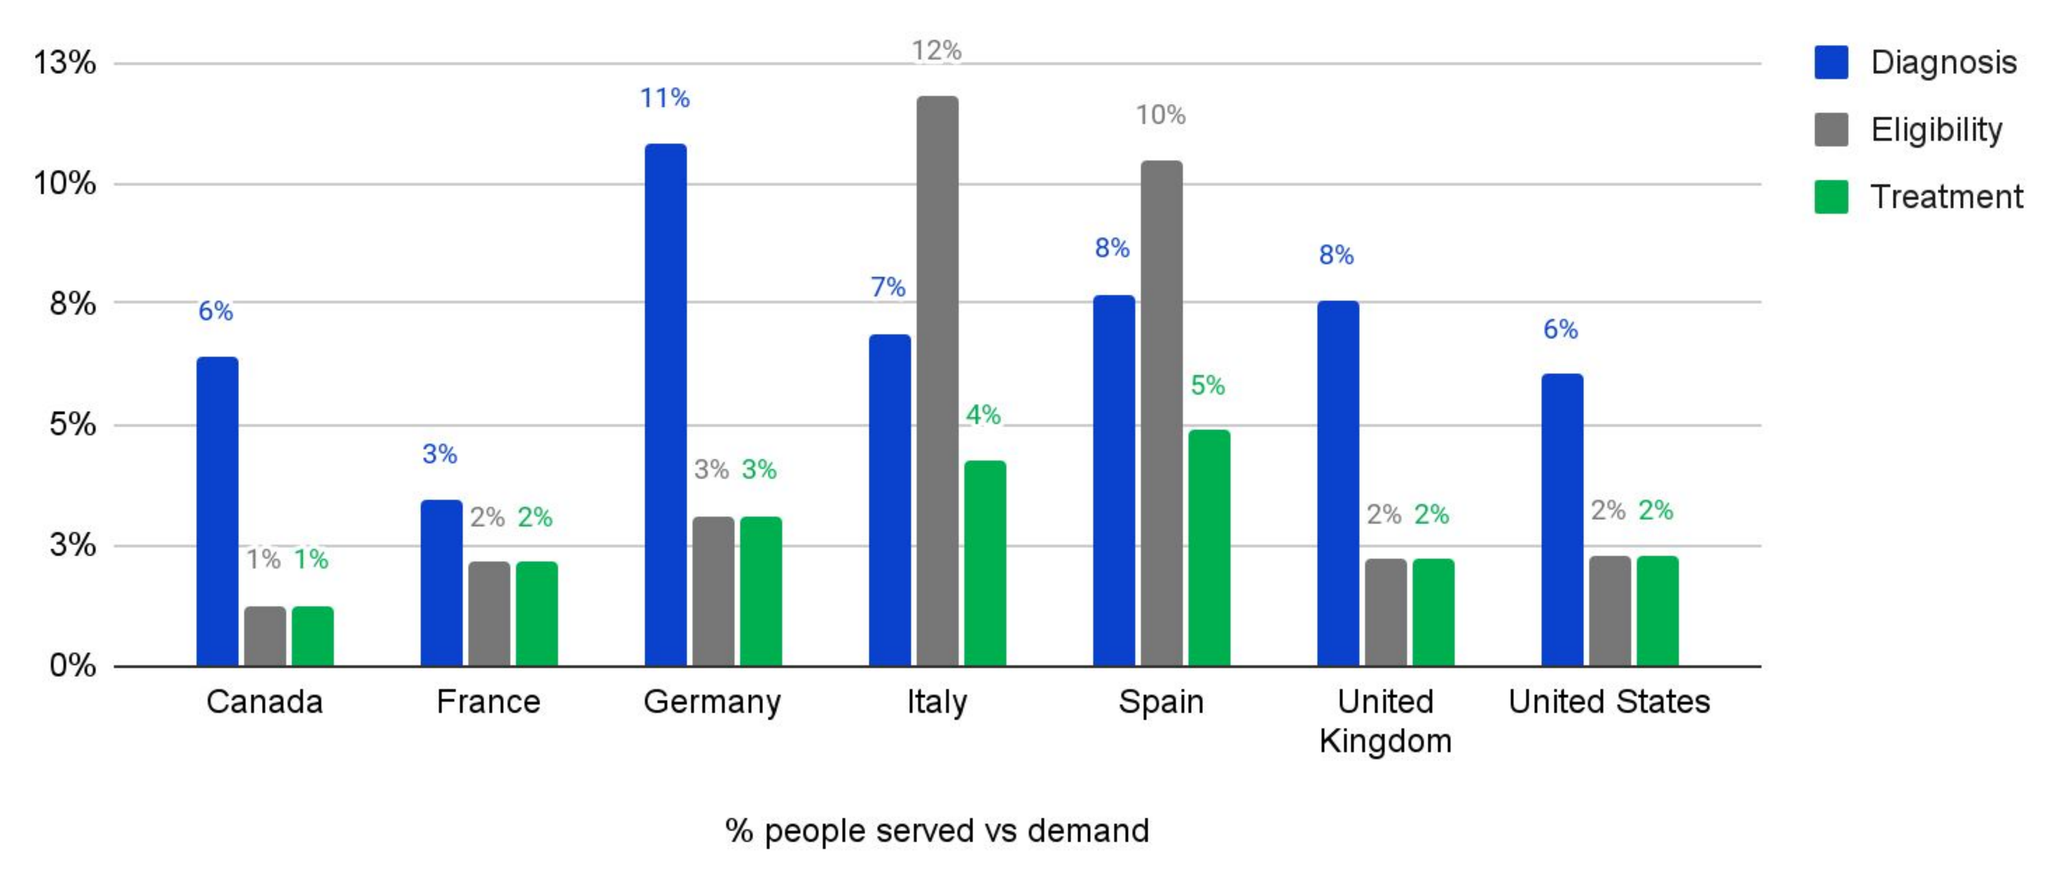
13%
10%
8%
5%
3%
0%
Canada 6% 1% 1%
France 3% 2% 2%
Germany 11% 3% 3%
Italy 7% 12% 4%
Spain 8% 10% 5%
United Kingdom 8% 2% 2%
United States 6% 2% 2%
% people served vs demand
Diagnosis
Eligibility
Treatment

- Neurologists, MRIs and PET scans were the largest bottlenecks across countries. Neurologist capacity was particularly limited in France, the UK and the US, where their numbers would need to increase by 41, 18 and 17 fold, respectively, to meet demand. MRI capacity would need to increase by 10-19 fold, while Canada, Germany, the UK and the US would require 31-79 fold existing PET scan capacity (Fig. 3).

Figure 3. Fold increase in health care resources required

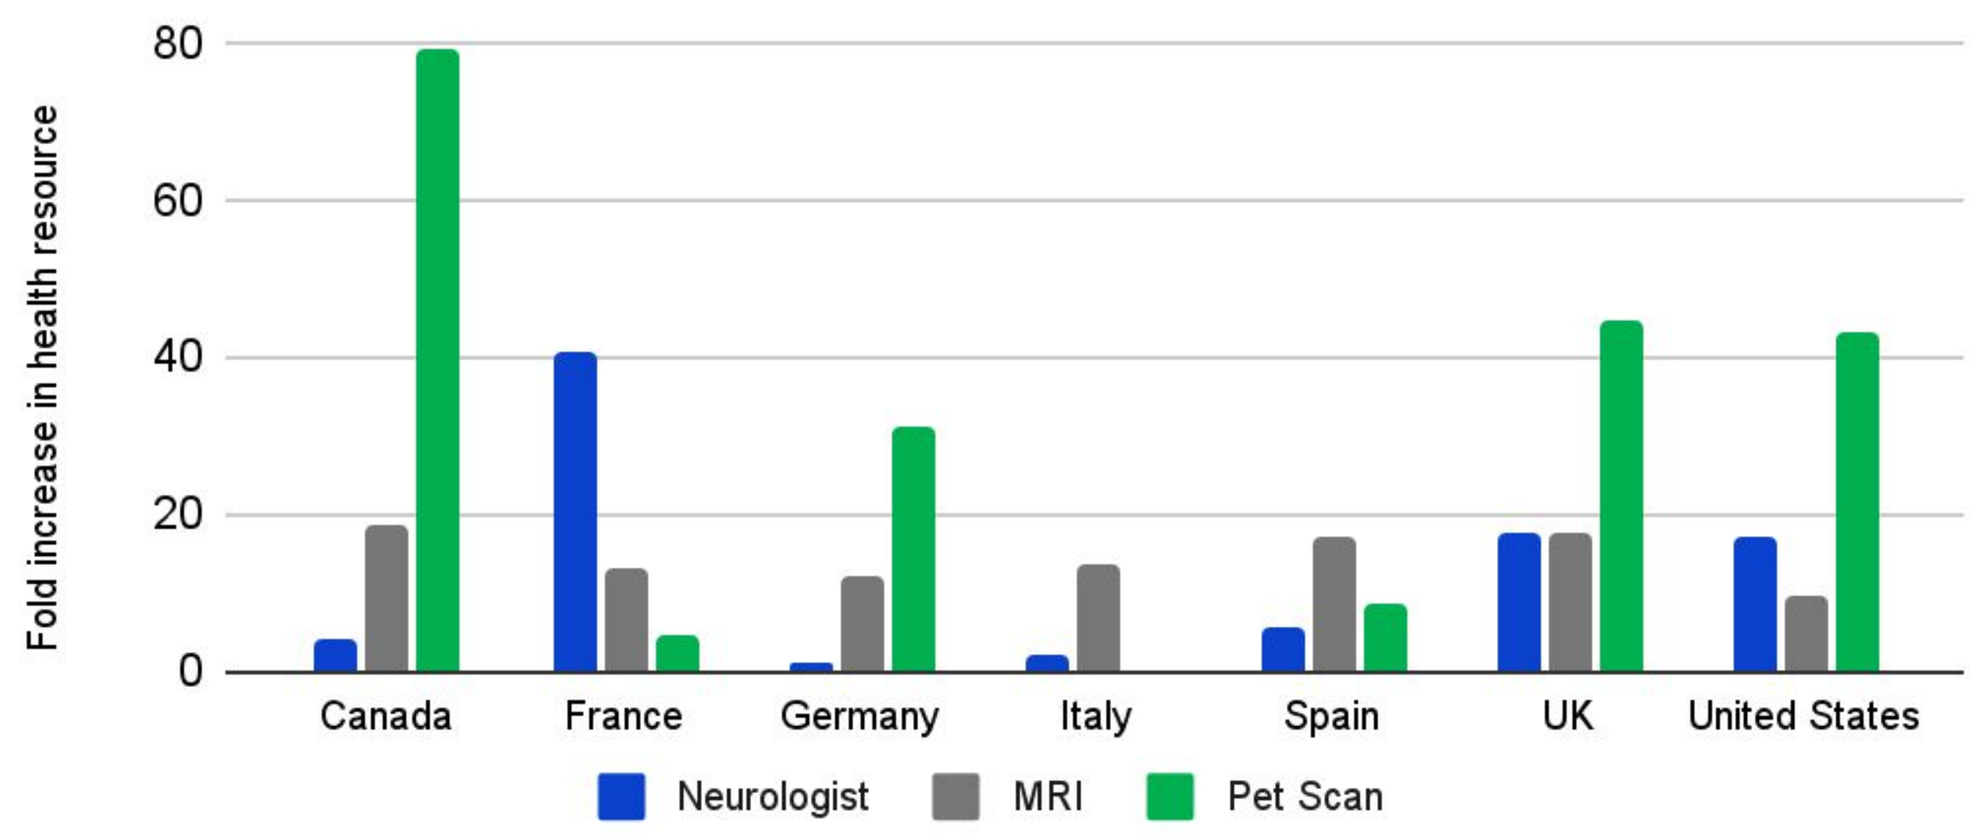
Fold increase in health resource
80
60
40
20
0
Canada
France
Germany
Italy
Spain
UK
United States
Neurologist
MRI
Pet Scan

- Through a shift to 90% CSF testing, two to five times more patients in Canada, the UK and the US could be tested for DMT eligibility (Fig. 4).

Figure 4. Number of people getting access to eligibility testing when shifted to 90% CSF vs Baseline in the first year of DMT availability

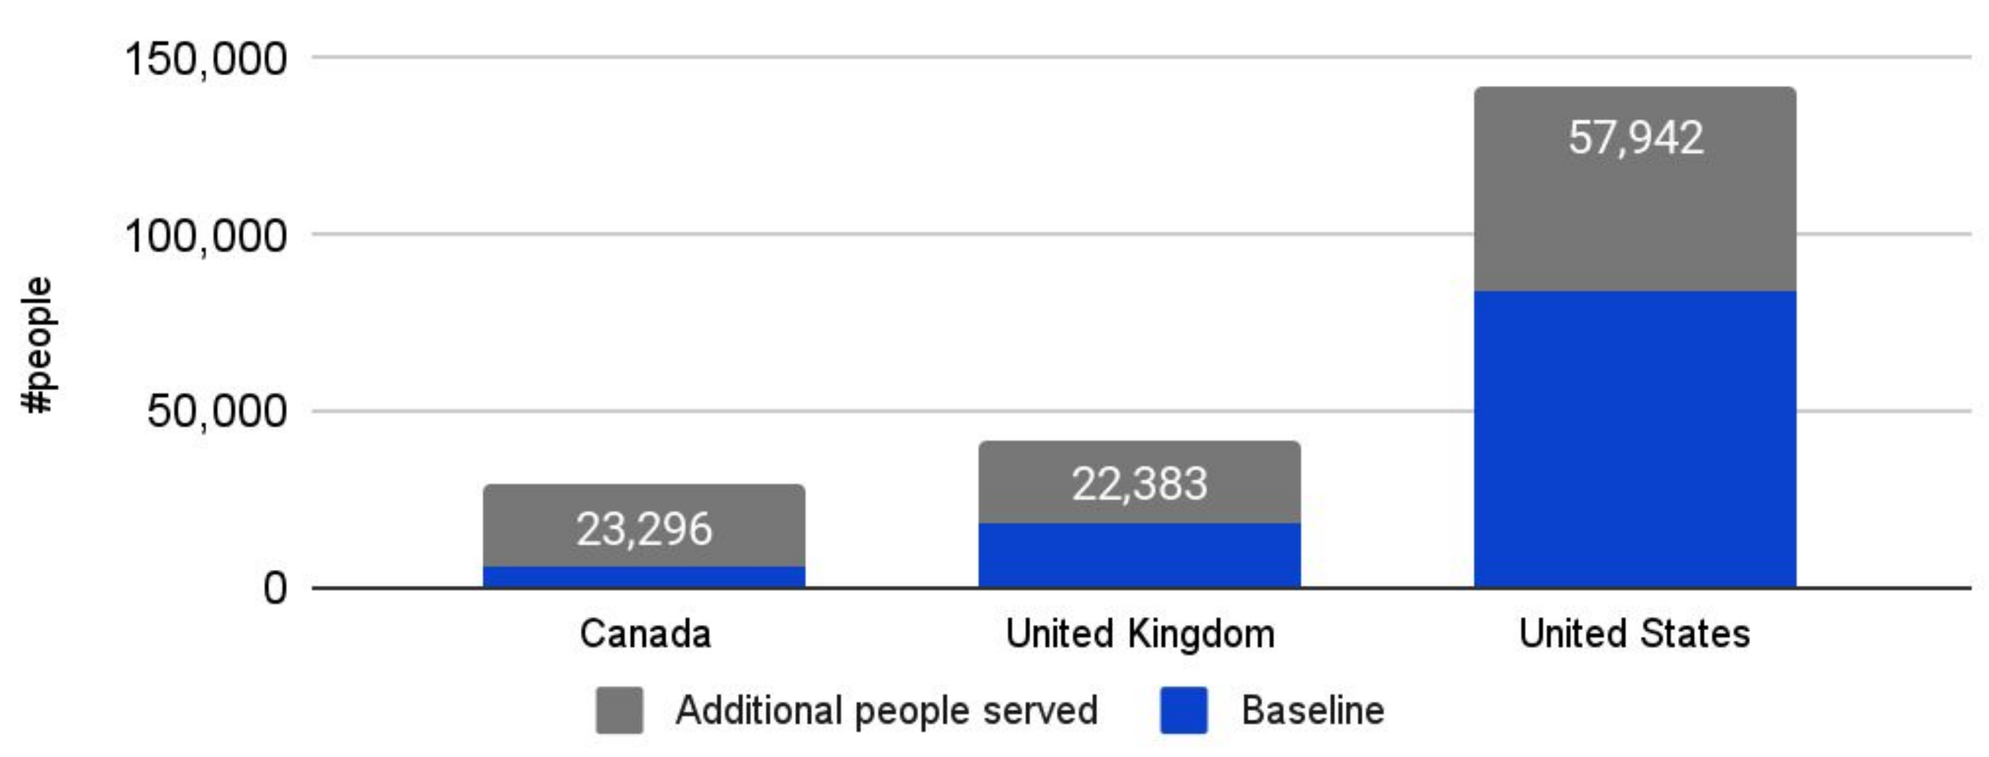
#people
150,000
100,000
50,000
0
23,296
22,383
57,942
Canada
United Kingdom
United States
Additional people served
Baseline

- Detection of early AD would require an estimated 4.0 to 11.3 hours of PCP time per month (Fig. 5).
- The US would require nearly double the number of monthly hours of PCP time per PCP (11 hours per month) as compared to Canada, France, Germany, Italy, Spain and the UK.

Figure 5. Required hours of PCP time per month per PCP

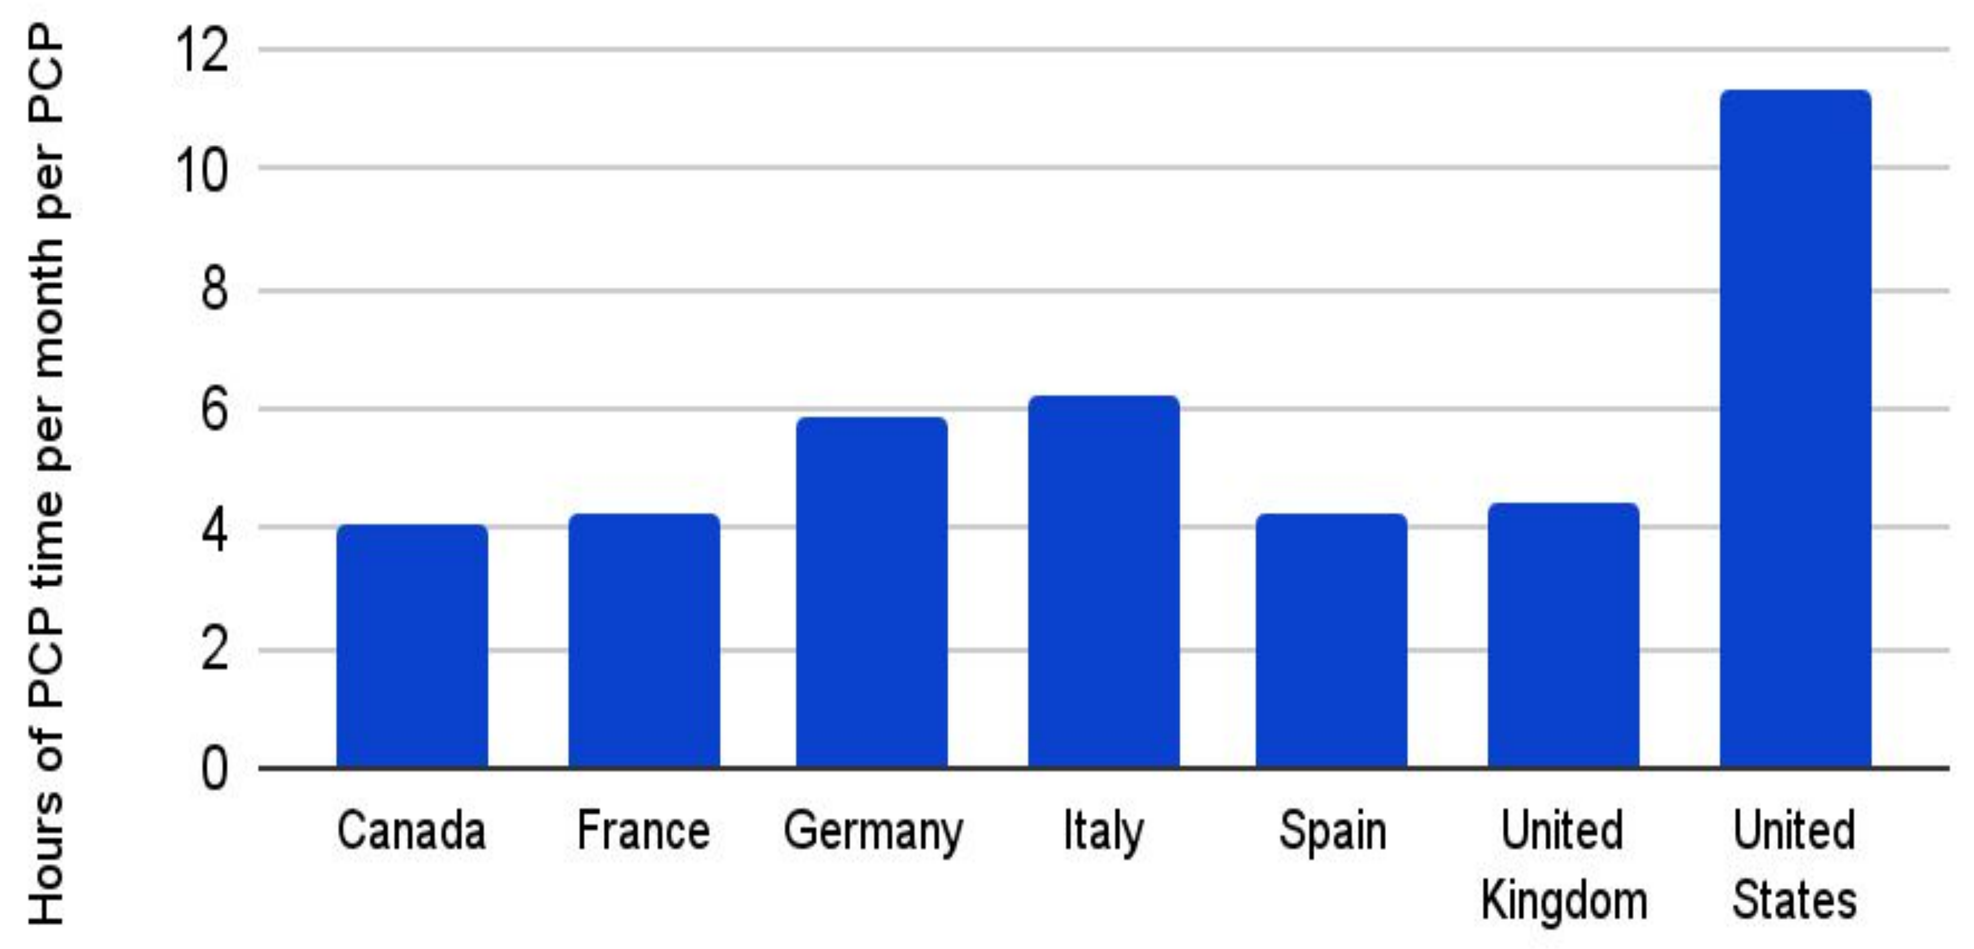
Hours of PCP time per month per PCP
12
10
8
6
4
2
0
Canada
France
Germany
Italy
Spain
United Kingdom
United States

Discussion

- Current health system capacity is radically insufficient to deliver access to AD DMTs.
- The confirmation of the presence of Aβ to determine treatment eligibility using CSF can increase capacity at this step.
- Additionally, early detection solutions to facilitate patient triage in primary care, such as blood-based biomarkers, and alternative DMT formulations to reduce administration burden, will be essential. Investments in human resources and infrastructure will also be critical to alleviate health system bottlenecks.
- Modelling health care capacity has inherent limitations. Key limitations include the focus on a single prominent model of care, the scope of the resources considered, and the flexibility of shifting resources from other disease areas.

Further information

- Additional details can be accessed in a related publication, *A model predicting healthcare capacity gaps for Alzheimer's disease modifying treatment in Canada* Black et al., Can J of Neur Sci, 2023

References

- [1]Gustavsson A, Norton N, Fast T, Frölich L, Georges J, Holzapfel D, Kirabali T, Krolak-Salmon P, Rossini PM, Ferretti MT, Lanman L, Chadha AS, van der Flier WM. Global estimates on the number of persons across the Alzheimer's disease continuum. Alzheimer's Dement. 2023 Feb;19(2):658-670.

Abbreviations

- DMT, disease modifying therapy; AD, Alzheimer's disease; PET, positron emission tomography, CSF, cerebrospinal fluid, PCP, primary care providers IV, intravenous, United States US, United Kingdom UK, Amyloid beta peptide (Aβ)

Acknowledgments

- We would like to thank all participants, study partners, investigators, and site staff for past and future engagement on the gantenerumab program
- This study was funded by F. Hoffmann-La Roche Ltd, Basel, Switzerland.

Disclosures

Nathalie Budd is an employee F. Hoffmann-La Roche Ltd. and owns stock in F. Hoffmann-La Roche Ltd., Valerie Crowell is an employee F. Hoffmann-La Roche Ltd. and owns stock in F. Hoffmann-La Roche Ltd., Ilke Mirik Danaci is an employee F. Hoffmann-La Roche Ltd. and owns stock in F. Hoffmann-La Roche Ltd., Dr. Haakon Nygaard has received consultancy fees from Hoffmann-La Roche Ltd. and support for this study, as noted above.

- NB: There may be associated costs for downloading data. These costs may vary depending on your service provider and may be high if you are using your smartphone abroad. Please check your phone tariff or contact your service provider for more details.
- Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.
- In order to use the QR code, please use or download an app called "QR code reader" from the Apple App Store™ or android Play Store™.

Presented at the 16th Clinical Trials on Alzheimer's Disease (CTAD) Conference; 24–27 October, 2023; Boston, MA, USA, and online