Matching-adjusted indirect comparison (MAIC) of entrectinib▼ versus crizotinib in patients with *ROS1* non-small cell lung cancer (NSCLC): an updated analysis

Paula Chu[1], Catherine Mitchell[2], Sarah Batson[2], Amine Aziez[1], Miranta Antoniou[1]

[1]F. Hoffman-La Roche, Basel, Switzerland ; [2]Mtech Access, Bicester, United Kingdom

INTRODUCTION

- C-ros oncogene 1 (*ROS1*) fusion-positive advanced NSCLC accounts for approximately 1–2% of non-squamous NSCLC[1-3]
- Targeted therapies are the standard of care for patients with *ROS1 fusion*-positive advanced NSCLC, with crizotinib being the first treatment licensed as a first-line option specifically for patients with *ROS1*-positive NSCLC
- Entrectinib was approved by the US Food and Drug Administration (FDA) in August 2019 and by the European Commission (EMA) in July 2020 for the treatment of adults with *ROS1* fusion-positive, metastatic NSCLC
- The aim of the current study is to perform an Indirect Treatment Comparison (ITC) to estimate relative treatment effects of entrectinib compared with crizotinib in patients with *ROS1* fusion-positive NSCLC
- This is an update of a previously published systematic literature review (SLR) and MAIC analysis which used earlier data cuts for both entrectinib and crizotinib. The previous analysis suggested improved outcomes (OS, ORR, AE Disc) for patients treated with entrectinib versus crizotinib[4]

METHODS

Systematic literature review

- To inform the ITCs, a SLR was required to identify all trial evidence for entrectinib and crizotinib in *ROS1* fusion-positive advanced or metastatic NSCLC
- The electronic searches were performed using defined keywords in the databases Medline®, Medline® Epub Ahead of Print (In-Process & Other Non-Indexed Citations), Embase, and EBM Reviews on 31st March 2020. Additional searches of congress proceedings from the past 3 years, reference lists of included publications, Health Technology Assessment (HTA) bodies, and clinical trial registries were conducted to identify relevant evidence. Inclusion criteria are presented in **Table 1**
- A study was performed assessing the feasibility of performing an ITC comparing entrectinib and crizotinib in patients with *ROS1* fusion-positive NSCLC, and considered both clinical and methodological homogeneity

Indirect Treatment Comparison

- Unanchored matching adjusted indirect treatment comparison (MAIC) was used to perform the indirect treatment comparison, as data came from single-arm clinical trials
- Matching was based on known prognostic and predictive factors: age, sex, smoking status, line of treatment, ECOG performance status, histology, and CNS metastases
- As the percentage of patients with CNS metastases was unknown in the PROFILE 1001 trial, three scenario analyses were performed in which the assumed percentage of patients with CNS in PROFILE 1001 was varied (scenarios as defined in Table 3). The choice of CNS percentage in each scenario was described in Chu et al. 2020[4]
- Outcomes included objective response rate (ORR), overall survival (OS), progression free survival (PFS), and discontinuation due to adverse events
- Hazard ratios (HRs) comparing entrectinib cohort(s) and the comparative evidence source were estimated using weighted Cox proportional hazards models. Confidence intervals were estimated using bootstrap sampling

Table 1. Inclusion criteria for the SLR

Criteria	Include	
Population	Adult patients with *ROS1* fusion-positive NSCLC	
Interventions	Entrectinib	
Comparators	Crizotinib	
Outcomes	Studies reporting at least one outcome of interest, as a primary or secondary outcome, including:	
	<u>Efficacy</u>: ORR; DOR; TTR; TTP; CBR; DCR; OS; PFS; Response rates (CR, PR, SD); Duration of treatment and duration of treatment beyond progression.	<u>Safety</u>: All-grade treatment related AEs; Treatment related Grade 3 or 4 AEs; Treatment related SAEs; Tolerability: Dose reductions and interruptions, discontinuation (any reason), discontinuation (due to AEs)
	All HRQL and PROs measures captured in trials	
Setting/study design	Prospective randomized control trials (Phase 2–4), non-randomized clinical studies, observational studies (retrospective/prospective)	
Language of publication	No restriction	
Date of publication	No restriction	

AE, adverse event; CBR, clinical benefit rate; CR, complete response; DCR, DOR, duration of response; HRQL, health-related quality of life; NSCLC, non-small cell lung cancer; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; PR, partial response; PRO, patient reported outcome; SAE, serious adverse event; SD, stable disease; TTP, time to progression; TTR, time to response.

RESULTS

Systematic literature review

- The screening procedure resulted in a final evidence base for the SLR of 54 publications related to 41 unique studies (Figure 1)
- Following meta-analysis feasibility assessment, data suitable for evidence synthesis included pooled data from three entrectinib single arm trials (ALKA-372-001, STARTRK-1 and STARTRK-2) and a single study investigating crizotinib (PROFILE 1001)
- These studies reported overlapping patient population characteristics and comparable endpoints, permitting a MAIC for entrectinib with crizotinib (PROFILE 1001)[5] . The previously published MAIC was based on PROFILE1001 data with shorter follow up[6]

Figure 1. PRISMA diagram for *ROS1* fusion-positive NSCLC SLR

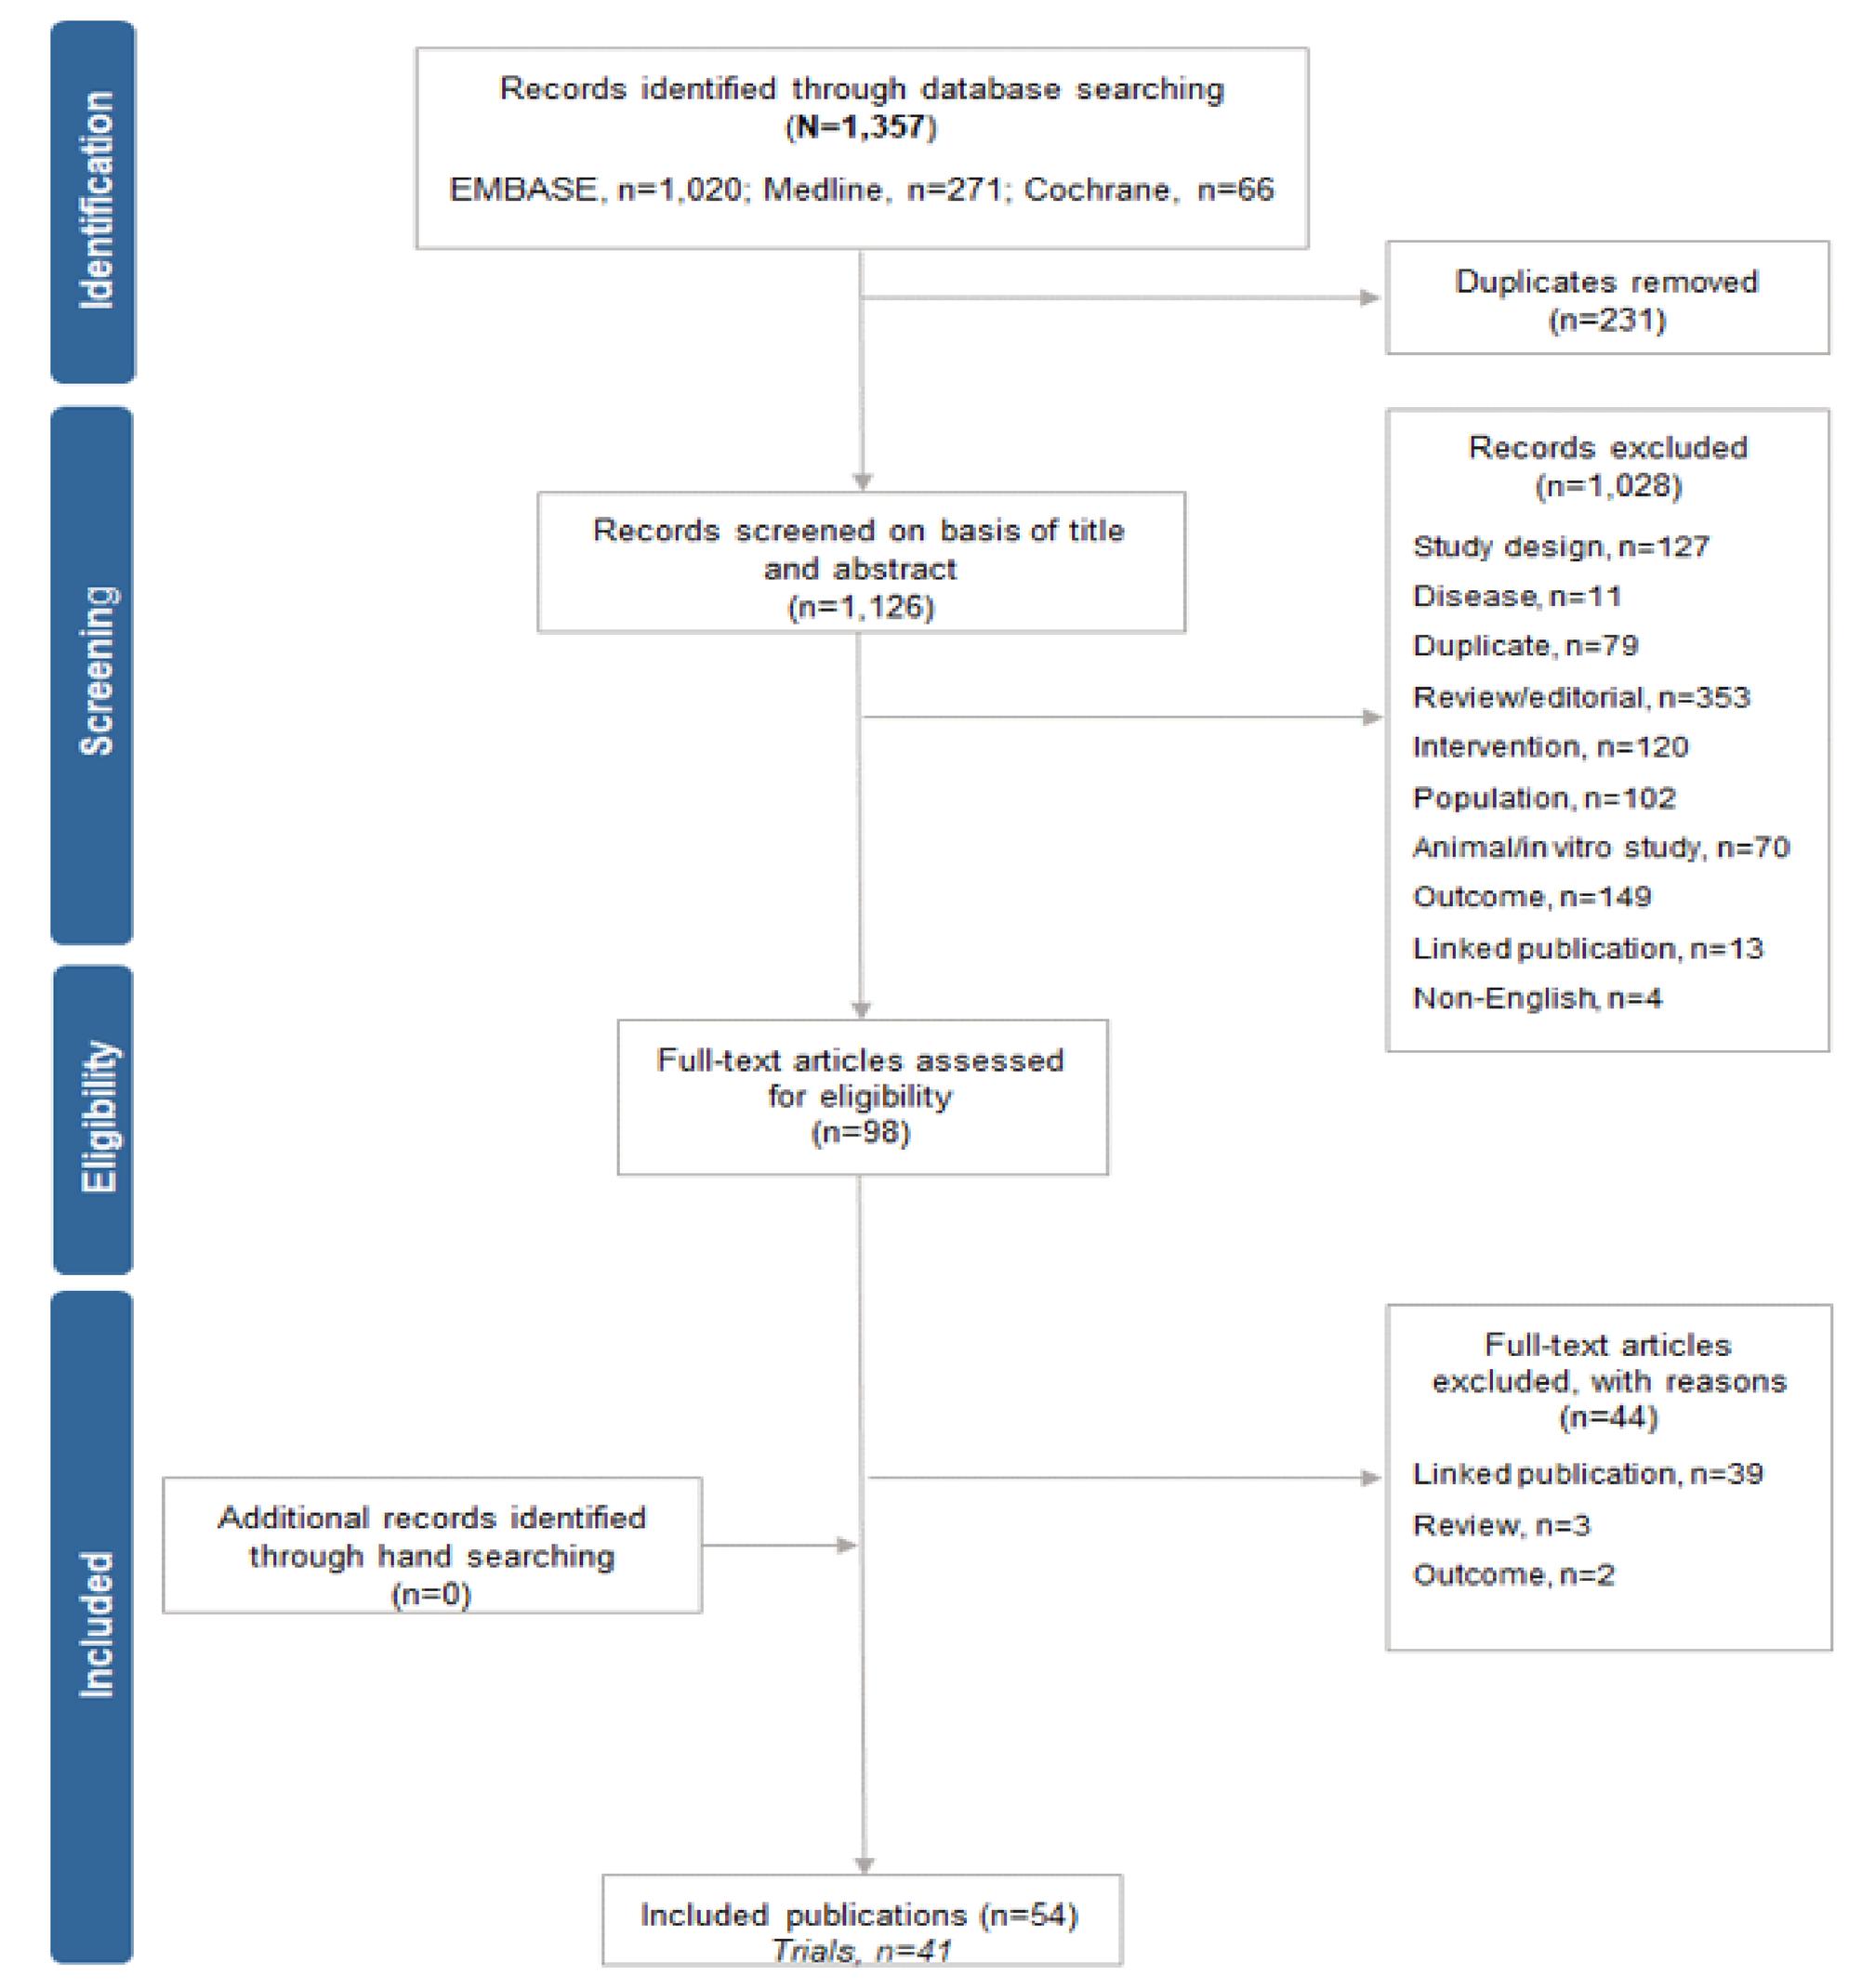

Identification
Screening
Eligibility
Included
Records identified through database searching (N=1,357)
EMBASE, n=1,020; Medline, n=271; Cochrane, n=66
Duplicates removed (n=231)
Records screened on basis of title and abstract (n=1,126)
Records excluded (n=1,028)
Study design, n=127
Disease, n=11
Duplicate, n=79
Review/editorial, n=353
Intervention, n=120
Population, n=102
Animal/in vitro study, n=70
Outcome, n=149
Linked publication, n=13
Non-English, n=4
Full-text articles assessed for eligibility (n=98)
Full-text articles excluded, with reasons (n=44)
Linked publication, n=39
Review, n=3
Outcome, n=2
Additional records identified through hand searching (n=0)
Included publications (n=54)
Trials, n=41

NSCLC, non-small cells lung cancer; *ROS1*, c-ros oncogene 1;SLR, systematic literature review

Indirect Treatment Comparison

- Matching successfully led to an entrectinib patient population that is equivalent to the PROFILE 1001 population based on the matching characteristics, as seen in **Table 2**

Table 2. Baseline Characteristics included in estimation of MAIC weights

	Intervention	Sample Size (ESS)	Age, years	Never smoked, %	ECOG 2, %	Adenocarcinoma, %	Treatment naïve, %	Female, %	Stage IV CNS, %
Scenario 1	Crizotinib	53	55.0	75.5	1.9	96.2	13.2	56.6	18.1
	Entrectinib	161	54.7	62.7	9.9	96.9	37.3	64.6	32.9
	Entrectinib re-weighted	**(96.4)**	**55.0**	**75.5**	**1.9**	**96.2**	**13.2**	**56.6**	**18.1**
Scenario 2	Crizotinib	53	55.0	75.5	1.9	96.2	13.2	56.6	24.6
	Entrectinib	161	54.7	62.7	9.9	96.9	37.3	64.6	32.9
	Entrectinib re-weighted	**(101)**	**55.0**	**75.5**	**1.9**	**96.2**	**13.2**	**56.6**	**24.6**
Scenario 3	Crizotinib	53	55.0	75.5	1.9	96.2	13.2	56.6	32.9
	Entrectinib	161	54.7	62.7	9.9	96.9	37.3	64.6	32.9
	Entrectinib re-weighted	**(101)**	**55.0**	**75.5**	**1.9**	**96.2**	**13.2**	**56.6**	**32.9**

Notes: Scenario 1 assumes 18.1% CNS metastasis, Scenario 2 assumes 24.64% CNS metastasis and Scenario 3 assumes 32.9% for efficacy analyses and 41.1% for safety analyses. Age is mean for Entrectinib, median for Crizotinib. CNS, central nervous system; ECOG, eastern cooperative oncology group; ESS, effective sample size.

▼This medical product is subject to additional monitoring. This will allow quick identification of new safety information. Healthcare professionals are asked to report any suspected adverse reactions. See section 4.8 of the SmPC for details on how to report adverse reactions.

Table 3. Summary of the results of entrectinib vs crizotinib in *ROS1* NSCLC patients

Scenario	Endpoint*	Effect Size¥	95% CI
Scenario 1 (18.1% CNS metastasis - as reported by Wu et al 2018)	OS – HR	0.75	(0.46, 1.11)
	PFS BICR – HR	1.23	(0.89, 1.60)
	ORR IA – OR	1.10	(0.70, 1.60)
	AE Disc – OR	0.70	(0.30, 1.30)
Scenario 2 (24.6% CNS metastasis - as per Flatiron analyses by RWD team)	OS – HR	0.80	(0.49, 1.17)
	PFS BICR – HR	1.26	(0.92, 1.64)
	ORR IA – OR	1.00	(0.70, 1.50)
	AE Disc – OR	0.70	(0.30, 1.30)
Scenario 3 (32.9% CNS metastasis for efficacy analyses and 41.1% CNS metastasis for safety analyses - same percentages as in the entrectinib studies)	OS – HR	0.87	(0.54, 1.27)
	PFS BICR – HR	1.29	(0.94, 1.68)
	ORR IA– OR	0.90	(0.60, 1.50)
	AE Disc – OR	0.70	(0.30, 1.10)

Notes: * OS, PFS BICR and ORR IA were assessed based on efficacy set (n=161) and AE Disc was assessed based on safety set (n= 209); ¥ OS, PFS and AE effect size <1, and ORR effect size >1 favour entrectinib vs crizotinib. AE, adverse events; BICR, blinded independent central review; CI, confidence interval; CNS, central nervous system; Disc, discontinuation; HR, hazard ratio; IA, investigator assessed; ORR, objective response rate; OR, odds ratio; OS, overall survival; PFS, progression-free survival. ORR IA was reported for the PROFILE 1001 study[5]. It is unclear if PFS reported in the PROFILE 1001 study[5] is IA or BICR.

Figure 2. Kaplan-Meier plots for Scenario 1

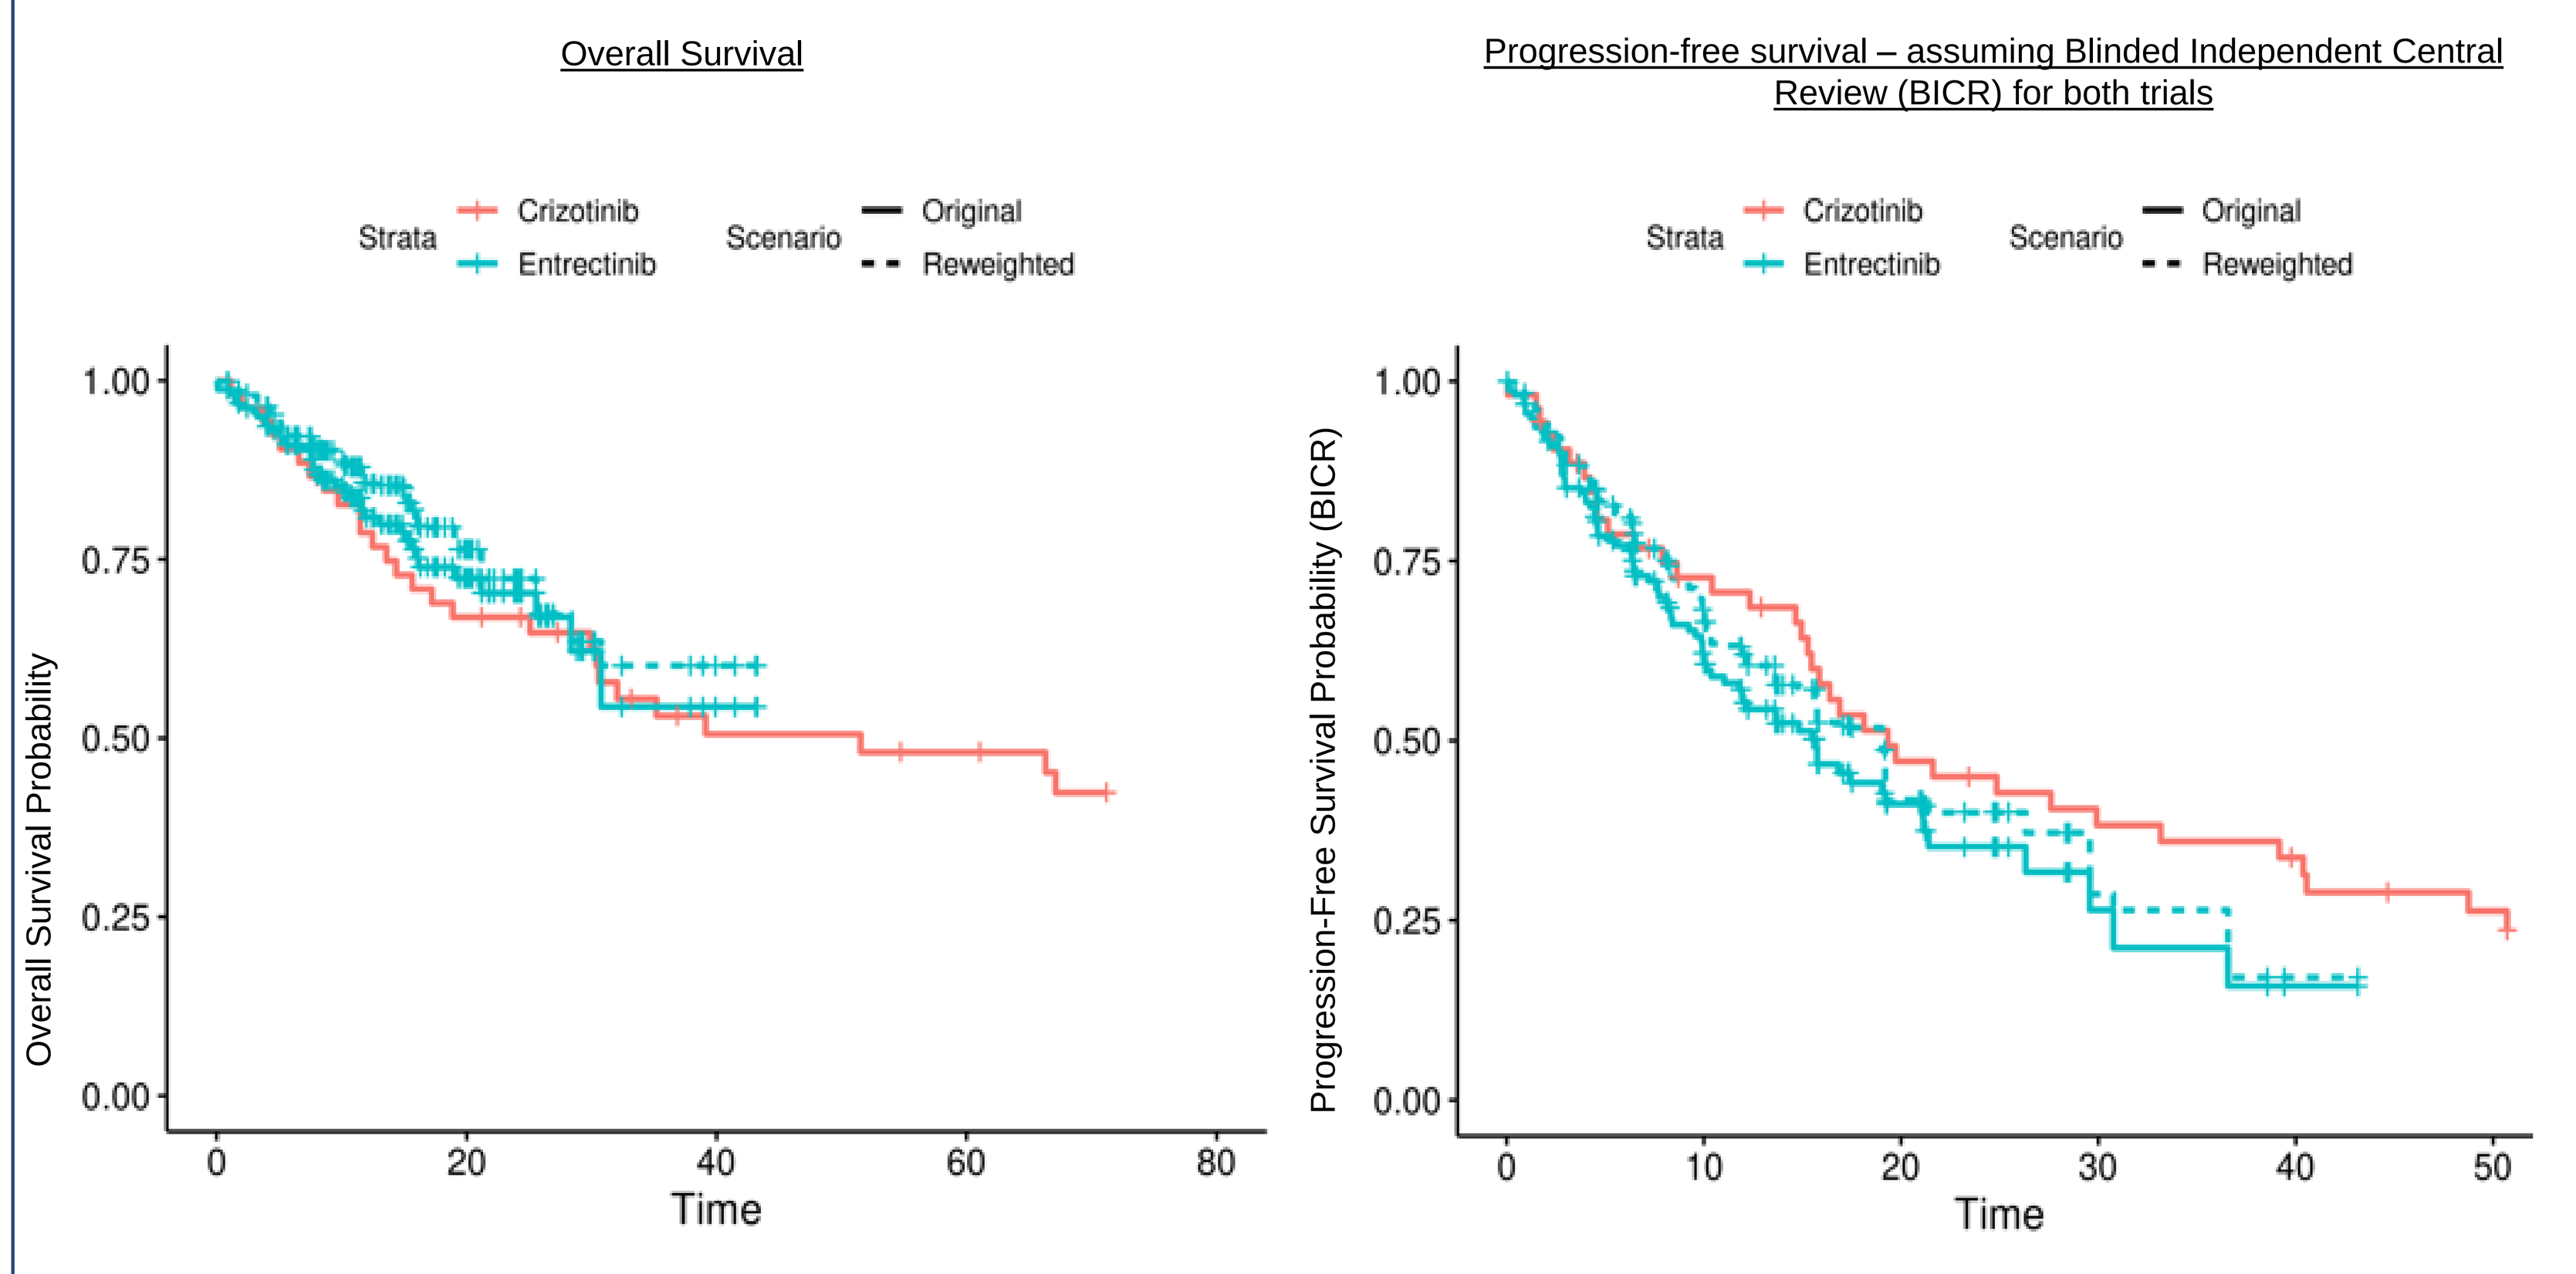

Overall Survival
Progression-free survival – assuming Blinded Independent Central Review (BICR) for both trials
Strata
Crizotinib
Entrectinib
Scenario
Original
Reweighted
Overall Survival Probability
Progression-Free Survival Probability (BICR)
1.00
0.75
0.50
0.25
0.00
0
20
40
60
80
10
30
50
Time

Scenario 1 - assumes 18.1% CNS metastases in PROFILE 1001 -as reported by Wu et al 2018[7]

Overall survival

- In the indirect comparison of entrectinib with crizotinib (PROFILE 1001), a trend for improved survival can be seen in favor of entrectinib (HR<1 indicates reduced risk of death with entrectinib versus comparator), although this difference is not statistically significant (CI includes 1)
- The HR for entrectinib versus crizotinib based on the MAIC suggests that treatment with entrectinib may reduce the risk of death compared with crizotinib in all three scenarios for CNS metastases in PROFILE 1001, as shown in **Table 3**

Progression-free survival

- It was unclear whether the evidence available for crizotinib was reported as Investigator Assessed (IA) or Blinded Independent Central Review (BICR). PFS was assessed by BICR in entrectinib studies and is used here.
- The HR for entrectinib versus crizotinib based on MAIC suggests that treatment with entrectinib may be associated with a trend towards higher risk of disease progression relative to crizotinib with the results for all scenarios of CNS metastases not being statistically significant, as shown in **Table 3**

Objective response rate (ORR)

- MAIC analysis results suggest that the adjusted entrectinib population may be associated with similar ORR compared with crizotinib in all three scenarios of CNS metastases. None of the results is statistically significant for any of the scenarios, as shown in **Table 3**

Discontinuation due to adverse events

- This outcome was assessed in the safety population (n=209)
- MAIC analyses suggest that entrectinib is associated with lower odds of discontinuation due to AE compared with crizotinib, although none of the estimates were statistically significant, as shown in **Table 3**

DISCUSSION

- Effective treatments are needed for patients with *ROS1* fusion-positive tumors with both systemic and CNS activity. CNS metastases are associated with a high disease burden and reduced quality of life
- Entrectinib has been designed to effectively penetrate and remain in the CNS as opposed to crizotinib. Preclinical and clinical data for entrectinib demonstrate that it has good CNS exposure and activity.
- Due to the rarity of *ROS1* fusions and single-arm trial design, the MAIC methodology was used to provide indirect comparative efficacy and safety estimates
- Analyses were limited by the small sample size of the available clinical evidence and the relative immaturity of data. The proportion of patients with CNS metastases at baseline was not reported in PROFILE 1001 and it was unclear whether PFS was assessed by BICR or IA. Finally, patients in the entrectinib cohort had a median survival follow-up of 15.8 months compared to 62.6 months in PROFILE 1001. Given these limitations, the results should be interpreted with caution.

CONCLUSIONS

- Consistently with the previously published analysis, results from the updated MAIC analysis suggest a trend towards improved OS and safety with entrectinib versus PROFILE1001 in *ROS1* NSCLC patients
- Comparing different trials without patient-level data requires assumptions to be made around characteristics and their weighting; this therefore limits the inferences that can be drawn from the results. Additionally, given the immaturity of the data, the small sample size and the unknown percentage of CNS metastasis in PROFILE 1001, results should be interpreted with caution
- While these results support the value of entrectinib as an efficacious new treatment for *ROS1* fusion-positive patients, further evidence comparing entrectinib directly with crizotinib will help to better understand the value of entrectinib in clinical practice. A head-to-head randomized clinical trial of entrectinib versus crizotinib will be initiated in mid-2021 to provide direct comparative evidence

ACKNOWLEDGMENTS / CONFLICTS OF INTEREST

We would like to thank Amine Aziez, formerly employed by F. Hoffmann-La Roche, for his contribution to the abstract which preceded this poster.

PC, AA, AM are employees of Roche; CM and SB are employees of Mtech Access who received funding from Roche for the update of the SLR.

REFERENCES

1. Bergethon, et al. J Clin Oncol 2012
2. Dugay, et al. Oncotarget 2017
3. Davies and Doebele. Clin Cancer Res 2013
4. Chu P, Antoniou M, Bhutani M, Aziez A, Daigl M. Matching-adjusted indirect comparison: entrectinib versus crizotinib in *ROS1* fusion-positive non-small cell lung cancer. Journal of Comparative Effectiveness Research 2020. DOI: 10.2217/cer-2020-0063.
5. Shaw A, Riely GJ, Bang YJ, Kim DW, Camidge DR, Shapiro GI, et al. Crizotinib in ROS1-rearranged advanced non-small-cell lung cancer (NSCLC): updated results, including overall survival, from PROFILE 1001. Annals of Oncology. 2019;30(7):1121-6.
6. National Institute for Health and Care Excellence (NICE). TA 529: Crizotinib for treating ROS1-positive advanced non-small-cell lung cancer, https://www.nice.org.uk/guidance/ta529(2018, accessed 10 September 2018)
7. Wu YL, Yang JCH, Kim DW, et al. Phase II study of crizotinib in east Asian patients with ROS1-positive advanced non–small-cell lung cancer. Journal of Clinical Oncology2018; 36: 1405-1411.

Find a copy of this e-poster at *Medically* using the short link:
https://bit.ly/361ZLMP

Copies of this e-poster obtained through QR, AR and/or text key codes are for personal use only and may not be reproduced without written permission of the authors

Visit *Medically.Roche.com* for more information.