Cost-Effectiveness Analysis of Baloxavir Marboxil as Seasonal Influenza Treatment Compared to Oseltamivir in China

Yuekun Tang[1] | Hongli Jiang[1*] | Yan Xia2 | Mi Tang[2]

[1] School of Publich Health, Fudan University, Shanghai, China

[2] Shanghai Roche Pharmaceuticals Ltd, Shanghai, China

Corresponding to: Hongli Jiang. Email: jianghl@fudan.edu.cn

Acknowledgements: This analysis was funded by Shanghai Roche Pharmaceuticals Ltd.

Background

- During 2006-2019, annual influenza-associated excess influenza-like illness(ILI) outpatient consultations, severe acute respiratory infections(SARI) hospitalizations and excess respiratory deaths were 3 million, 2.34 million, 0.09 million, respectively, in China[1].
- Baloxavir acts on the cap-dependent endonuclease specific to influenza A and B viruses, inhibiting the cap-snatching step in viral mRNA transcription[2]

Objective

- To estimate the cost-effectiveness of baloxavir marboxil versus oseltamivir to treat seasonal influenza patients in China from the healthcare system perspective.

Methods

- **Target population:** Chinese patients with ILI. According to health status, they were divided into otherwise healthy (OWH) and high risk adults.
- **Model structure:** A one-year decision tree model was constructed to estimate the incremental cost-effectiveness ratio (ICER) from the healthcare system perspective. The following events were considered: whether the patient is antiviral treated, utilization of medical services, development of complications, death in ICU (Figure 1).
- **Effect inputs:** A transmission dynamics model assuming that 10% of patients who receive antiviral therapy was leveraged to estimate the spread of the disease in the Chinese population and the protection rate of baloxavir to avoid influenza transmission. Influenza complication rates are quoted from phase III clinical trials of baloxavir and other epidemiological parameters from available literature [3-5], detailed constituent ratio in Table 1. According to the clinical trials and life expectancy in China, baloxavir used in high risk patients averting one case of premature death were calculated by 13.2 QALYs.
- **Costs inputs:** Direct medical costs were collected from hospitals, including 1,496 outpatients with influenza like illness and 308 influenza related inpatients, the detailed per capita costs in Table 2.
- **Sensitivity analysis:** One-way sensitivity analysis and probabilistic sensitivity analysis were adopted to verify the robustness of the results.

Table 1 Effect inputs

Items	OWH	High Risk
Outpatient complications constituent ratio (with oseltamivir)		
Pneumonia	0.1250	0.1429
Bronchitis	0.7500	0.6429
Otitis media	0.1250	0.0714
Nasosinusitis	0.0000	0.1429
Outpatient complications constituent ratio (with baloxavir)		
Pneumonia	0.1250	0.000
Bronchitis	0.5625	0.8750
Otitis media	0.1250	0.0000
Nasosinusitis	0.1875	0.1250
Inpatient	0.0102	0.0514
ICU	0.2167	0.2353

Table 2 Costs

Items	OWH	High Risk
Outpatient without antiviral therapy (per capita, CNY¥)		
Influenza	80.2	76.4
Pneumonia	243.5	245.7
Bronchitis	266.0	136.9
Otitis media	113.8	71.2
Nasosinusitis	40.6	40.6
Inpatient without antiviral therapy (per capita, CNY¥)		
Ward	2,843.4	5363.9
ICU	16,096.6	34,286.1
Oseltamivir	219.56	
Baloxavir	498	

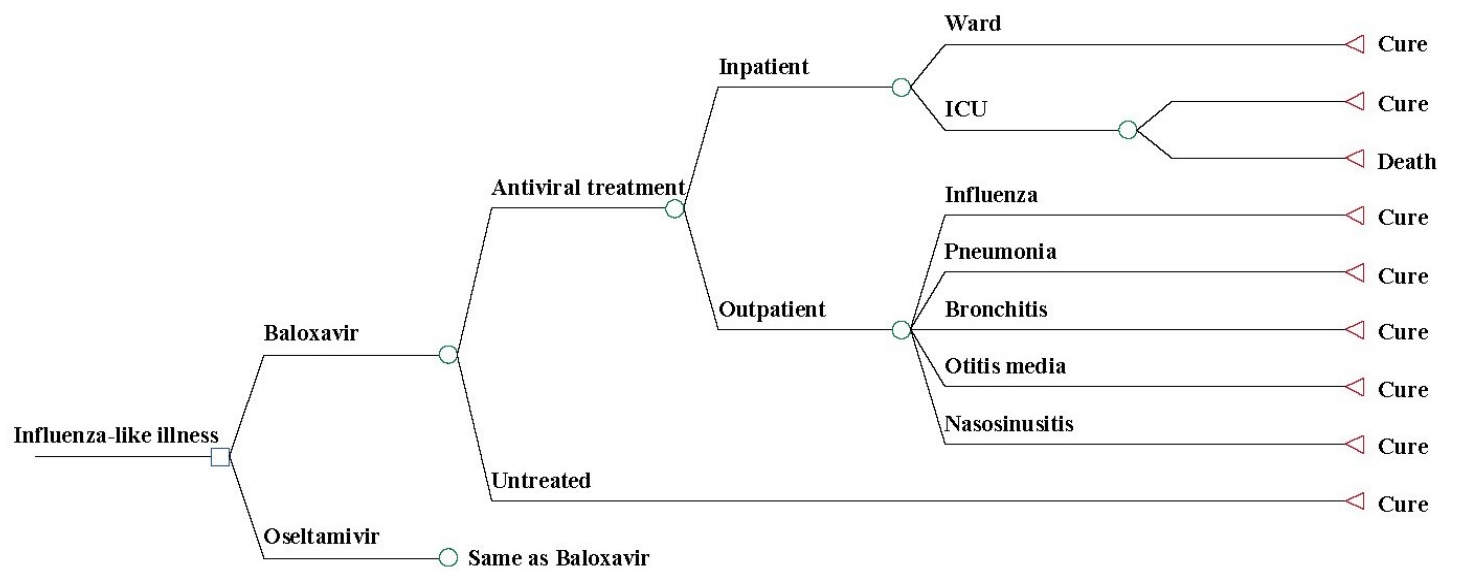
Influenza-like illness
Baloxavir
Oseltamivir
Same as Baloxavir
Antiviral treatment
Untreated
Inpatient
Outpatient
Ward
ICU
Influenza
Pneumonia
Bronchitis
Otitis media
Nasosinusitis
Cure
Death

Figure 1 Model Structure

Results

Base case Results

- Baloxavir provided 0.000246 and 0.000186 more QALYs per capita in otherwise healthy and high risk populations with fewer costs, respectively.
- In otherwise healthy and high risk populations, the baloxavir was dominant (Table 3)

Sensitivity Analysis Results

- One-way sensitivity analysis showed the results were generally robust (Figure 3, Figure4).
- In OWH populations ICER were largely influenced by the number of infections reduced by baloxavir and the number of hospitalizations avoided.
- In High Risk populations ICER were largely influenced by QALY of patients with influenza alone and the number of hospitalizations avoided by baloxavir.
- The cost-effectiveness acceptability curve (CEAC) shows that baloxavir to be the cost-effective strategy in 100% of cases at the WTP threshold of 10,000 CNY per QALY gained in otherwise healthy populations, and 100% of cases at the WTP threshold of 0 CNY per QALY gained in high risk populations. (Figure 5 and Figure 6)

Table 3 Base case Results (per capita)

	Costs (CNY¥)	Inc. Costs	QALY	Inc. QALY	ICER (CNY¥/QALY)
Otherwise Healthy					
Ose	31.899800	-	0.958733	-	
Bal	28.891263	-3.008537	0.958979	0.000246	dominant
High Risk					
Ose	67.513149	-	0.958610	-	-
Bal	55.431299	-12.081850	0.958796	0.000186	dominant

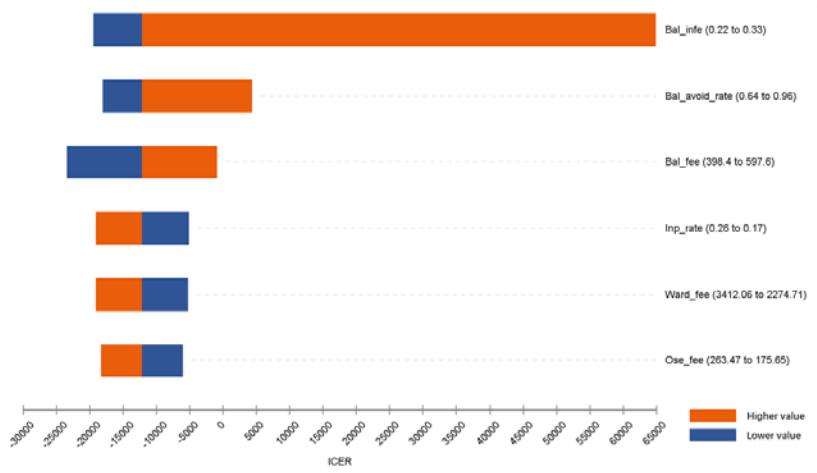
Bal_infe (0.22 to 0.33)
Bal_avoid_rate (0.64 to 0.96)
Bal_fee (398.4 to 597.6)
Inp_rate (0.26 to 0.17)
Ward_fee (3412.06 to 2274.71)
Ose_fee (263.47 to 175.65)
Higher value
Lower value
ICER

Figure 3 Tornado Diagram in OWH

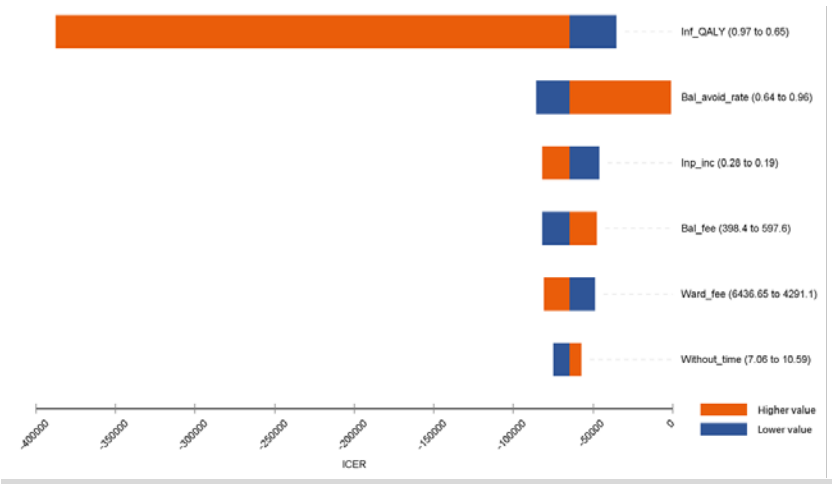
Inf_QALY (0.97 to 0.65)
Bal_avoid_rate (0.64 to 0.96)
Inp_inc (0.28 to 0.19)
Bal_fee (398.4 to 597.6)
Ward_fee (6436.65 to 4291.1)
Without_time (7.06 to 10.59)
Higher value
Lower value
ICER

Figure 4 Tornado Diagram in High Risk

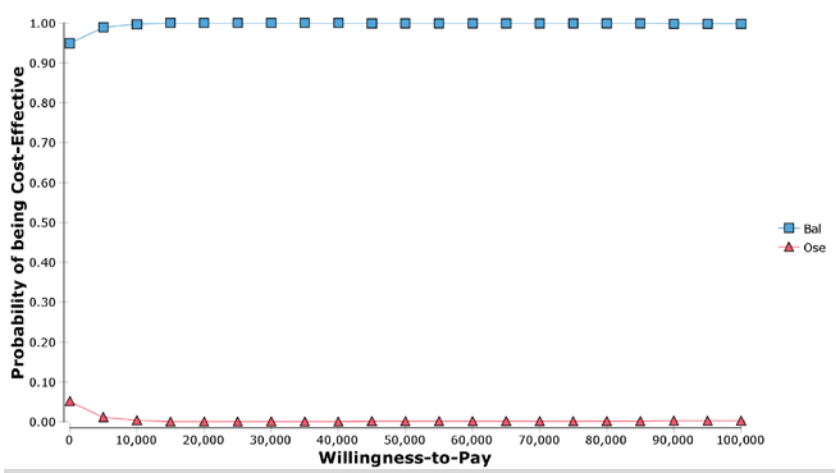
Probability of being Cost-Effective
Willingness-to-Pay
Bal
Ose

Figure 5 CEAC (OWH)

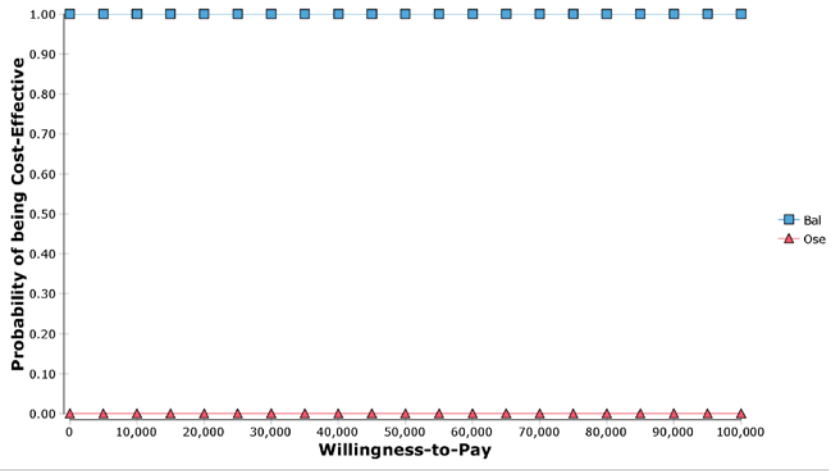
Probability of being Cost-Effective
Willingness-to-Pay
Bal
Ose

Figure 6 CEAC (High Risk)

Conclusion

Compared to oseltamivir, baloxavir is a very cost-effective intervention treating seasonal influenza in China. Both in otherwise healthy and high risk populations, baloxavir is a dominant alternative.

References

[1] Gong H., et al. Natl Med J China, 2021, 101(8), 560-567. (In Chinese)
[2] Omoto S., el al. Sci Rep 2018;8:9633.
[3] SHIONOGI CO. L. Clinical Study Report: 1601T0831[R]. 2018.
[4] SHIONOGI CO. L. Clinical Study Report: 1601T0832[R]. 2018.
[5] Kamal M A., et al. Br J Clin Pharmacol, 2017,83(7): 1580-1594.