Mental Health Concerns of Patients and Their Caregivers in the Food Allergy Research & Education (FARE) Patient Registry Add to the Burden of Food Allergy

309

Sachin Gupta,[1] Christopher Warren,[2] Arpamas Seetasith,[1] Robert Schuldt,[1] Ruchi Gupta,[2] Thomas B. Casale[3]

[1]Genentech, Inc., South San Francisco, CA, USA; [2]Northwestern University, Chicago, IL, USA; [3]University of South Florida, Tampa, FL, USA

Background

- IgE-mediated food allergies[1] carry a risk of severe reactions,[2] including anaphylaxis, and can impart psychosocial burden on patients and caregivers.[3]
- Although anxiety is associated with food allergies,[4] little is known about the mental health status of patients or their caregivers.
 - Additionally, the clinical phenotypes and experience of patients with single versus multiple food allergies have not been well characterized.

Objectives

- To describe the self-reported mental health characteristics of patients with food allergies, and their caregivers, using the Food Allergy Research & Education (FARE) Patient Registry database.
- To compare mental health characteristics between patients with single versus multiple food allergies.

Methods

FARE Patient Registry

- The FARE Patient Registry[5] (NCT04653324) conducts online surveys of patients with food allergies, or caregivers acting on behalf of patients, and collects cross-sectional data regarding their experiences.
- Using data from the FARE Patient Registry from May 2017 to January 2021, we selected patients with the following characteristics: completed survey, reside in the United States, have reported ≥1 food allergy without discordant data for single food allergy, and reported age at diagnosis.
 - A total of 5588 patients met the inclusion criteria and 1680 (30%) patients and/or their caregivers completed a survey assessing their mental health concerns related to food allergies, and levels of mental health screening and care.

Results

Patient Characteristics

- Demographic characteristics of FARE Patient Registry participants who completed the mental health survey were similar between patients with single and multiple food allergies (**Table**).

Table. Patient Characteristics

Characteristic, n (%)	Patients With Single Food Allergy n=276	Patients With Multiple Food Allergies n=1404
Female	177 (64)	848 (60)
White	228 (83)	1093 (78)
Age <18 years	158 (57)	765 (55)
Diagnosis at <6 years of age	178 (64)	969 (69)
FARE survey respondent		
Patient	105 (38)	595 (42)
Caregivers	171 (62)	809 (58)
Comorbidities		
Atopic dermatitis	94 (34)	751 (53)
Asthma	99 (36)	742 (53)
Allergic rhinitis	82 (30)	652 (46)
Mental health disorders*	67 (24)	445 (32)

FARE, Food Allergy Research & Education. *In response to the survey question, "Has the participant ever been diagnosed with a mental health disorder?" The types of mental health disorders were not specified in the questionnaire.

- The most common allergens were peanuts (64%; 1082/1680), tree nuts (62%; 1046/1680), egg (41%; 697/1680), and milk (37%; 620/1680).
- Most patients (62%; 1041/1680) experienced an average of ≥1 food-allergic reaction, of which 53% (889/1680) occurred away from home and 51% (717/1680) within 5 minutes of exposure, and 31% (518/1680) of patients reported a severe/very severe recent reaction.
- Most patients were exposed accidentally (78%; 1055/1680), by ingestion (86%; 1164/1680), and 9% (123/1680) of patients experienced a reaction when they were alone.
- Anxiety was the most common emotion that participants self-reported as a result of eating the food that produced an allergic reaction (**Figure 1**).

Figure 1. Commonly Reported Emotional Food-Allergic Reaction Symptoms

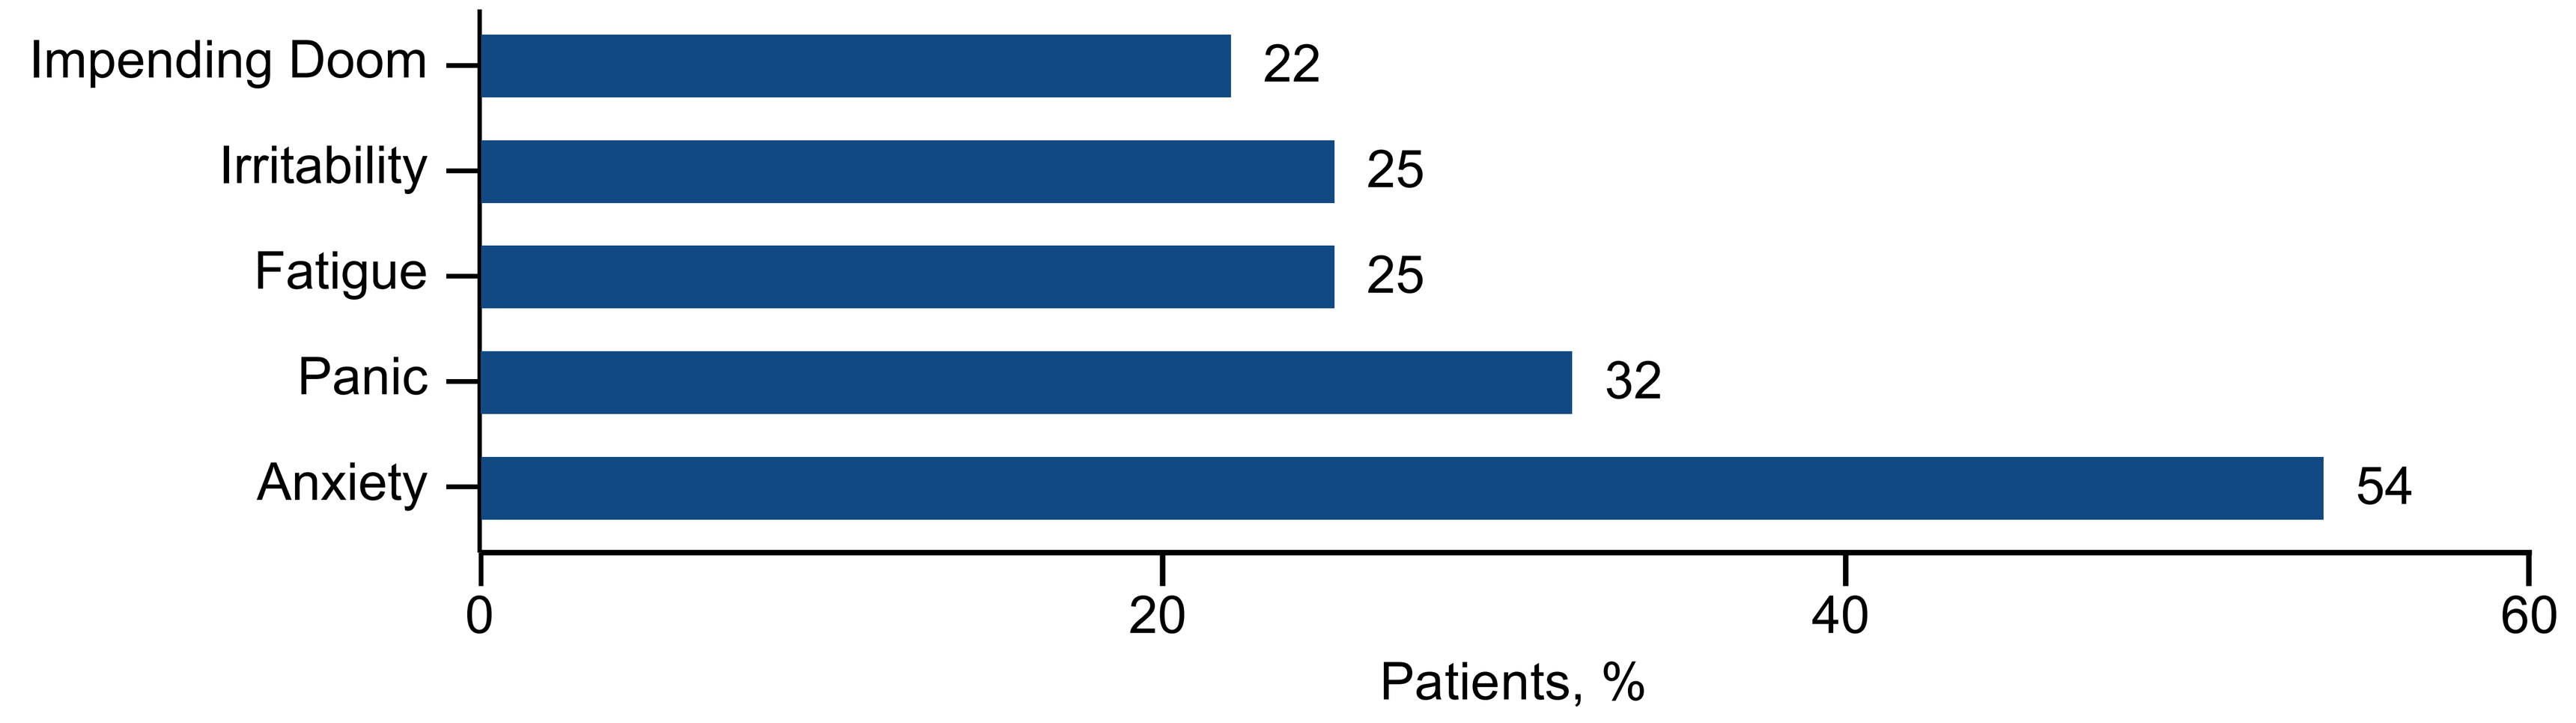
Impending Doom
22
Irritability
25
Fatigue
25
Panic
32
Anxiety
54
0
20
40
60
Patients, %

Patient Mental Health Concerns

- About two-thirds (62%; 1035/1680) of patients reported mental health concerns related to food allergies, and concerns were more prevalent in patients with multiple food allergies (**Figure 2**).
- Over one-third (38%; 635/1680) of patients with food allergies reported mental health concerns that carried over to non–food allergy–related problems.
- Only 8.7% (146/1680) of patients were screened for mental health disorders during their food allergy appointment.

Figure 2. Patient-Reported Mental Health Concerns

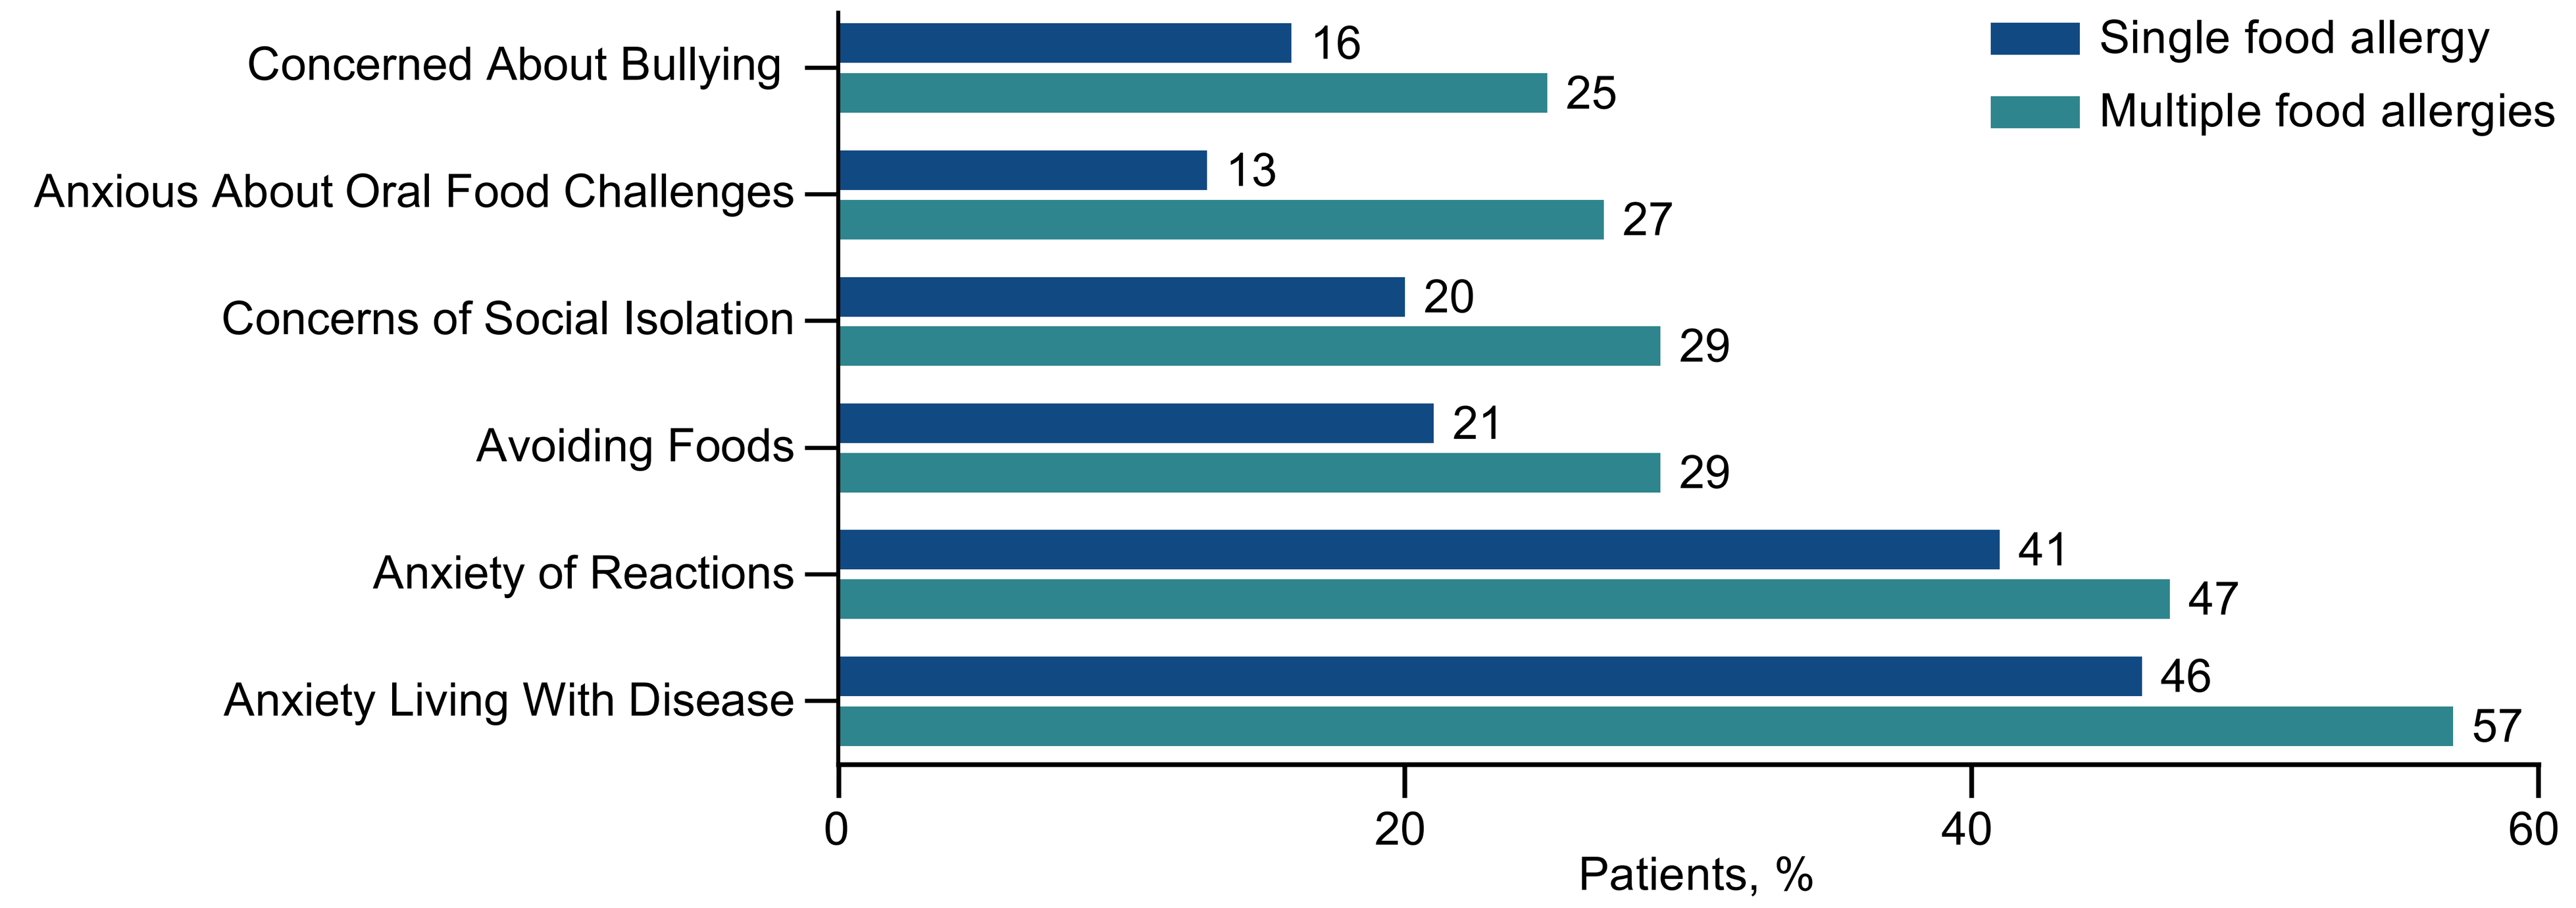
Single food allergy
Multiple food allergies
Concerned About Bullying
16
25
Anxious About Oral Food Challenges
13
27
Concerns of Social Isolation
20
29
Avoiding Foods
21
29
Anxiety of Reactions
41
47
Anxiety Living With Disease
46
57
0
20
40
60
Patients, %

Caregiver Mental Health Concerns

- The most common concerns reported by caregivers of patients with food allergies were fear of trusting others with their child (33%; 551/1680), fear for safety of their child (32%; 544/1680), and struggling with stress (23%; 391/1680; **Figure 3**).

Figure 3. Caregiver-Reported Mental Health Concerns

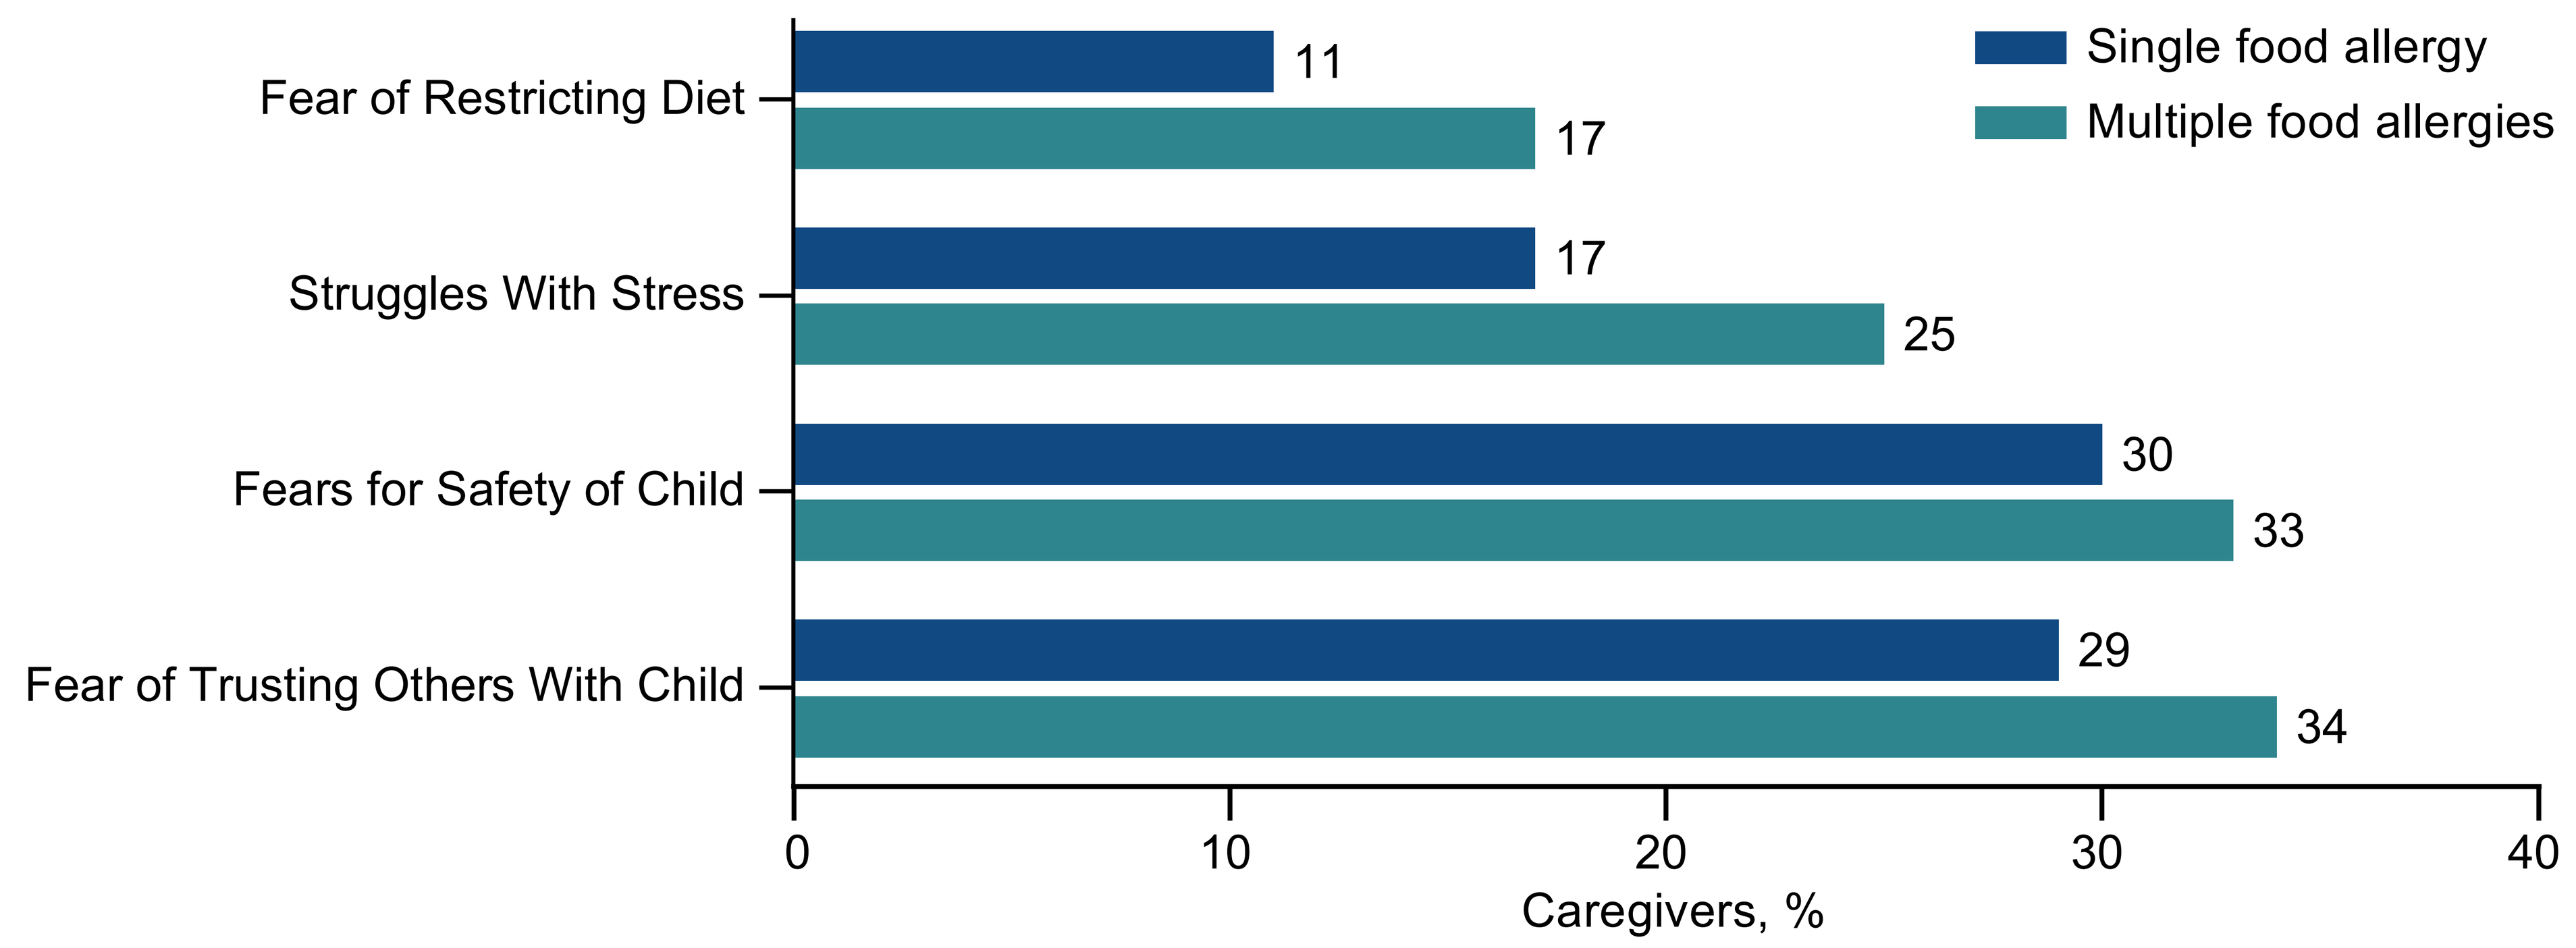
Single food allergy
Multiple food allergies
Fear of Restricting Diet
11
17
Struggles With Stress
17
25
Fears for Safety of Child
30
33
Fear of Trusting Others With Child
29
34
0
10
20
30
40
Caregivers, %

- Many caregivers sought mental health care to cope with worry related to caring for patients with food allergies (13%; 226/1680).
 - A higher proportion of caregivers of patients with multiple food allergies sought mental health care (14%; 203/1404), compared with single food allergy (8%; 23/276).

Limitations

- Given that the FARE Patient Registry is voluntary, the data may be reflective of the type of patient responding to a registry request and may not be representative of all patients with food allergies.
- Data in FARE are self-reported or reported by a parent or caretaker on behalf of a child and, therefore, could be subject to recall bias.
- The FARE "Mental Health Concerns" survey asks questions on typical concerns, such as worry, stress, anxiety, or sadness about food allergy, or an impact on quality of life, but should not be used for diagnostic purposes.

Conclusions

- Mental health concerns are prominent among patients with food allergies and their caregivers, especially for patients with multiple food allergies, which adds to the burden of food allergies.
 - Concerns also spilled over to non–food allergy–related problems.
- In addition, many patients living with food allergies who have mental health concerns are not screened during their food allergy appointment.
 - About one-third of patients reported a mental health disorder as a comorbidity.
 - About 1 in 10 patients are screened for mental health disorders during their appointment.
- Clinicians should screen patients and their caregivers for mental health diagnoses and provide information and effective treatment options.

References

1. Tanno LK, et al. *World Allergy Organ J*. 2016;9:24.
2. Gupta RS, et al. *JAMA Netw Open*. 2019;2:e185630.
3. Golding MA, et al. *Ann Allergy Asthma Immunol*. 2021;127:536–47.e3.
4. Moen ØL, et al. *J Pediatr Nurs*. 2019;46:e52–63.
5. FARE Patient Registry. FARE website. Accessed December 7, 2022. https://www.foodallergypatientregistry.org/.

Disclosures

SG, AS, RS: employees of Genentech, Inc.; stockholders in Roche. **CW:** institutional research funding from FARE, the National Institutes of Health, and the Sunshine Charitable Foundation. **RG:** support from ρ Inc, Aimmune, Allergy and Asthma Network, FARE, Mylan, National Confectioners Association, National Institutes of Health, Northwestern University Clinical and Translational Sciences Institute, Stanford Sean N. Parker Center for Allergy Research, Thermo Fisher Scientific, and UnitedHealth Group; has served as a medical consultant/advisor for Aimmune, Before Brands, DBV Technologies, Kaléo, Mylan, and Pfizer. **TBC:** consultant and speaker bureau member for Genentech, Inc.; consultant for Novartis Pharmaceuticals Corporation; chief medical advisor for FARE.

Acknowledgments

This analysis was funded by Genentech, Inc., a member of the Roche Group. Medical writing assistance was provided by Nilisha Fernando, PhD, of Envision Pharma Group, and funded by Genentech, Inc., a member of the Roche Group.

Scan code for ePoster/PDF or visit https://bit.ly/3W8IUQE

Presented at the American Academy of Allergy, Asthma & Immunology (AAAAI) 2023 Annual Scientific Meeting, February 24–27, 2023, San Antonio, TX, USA

Copyright © 2023. All rights reserved. Contact Arpamas Seetasith for permission.